55

Interferon β-1a s.c. tiw treatment response and disability progression

E. Järvinen[1], A. Murtonen[2], M.L. Sumelahti[2]

[1]Merck Finland, Espoo, Finland and University of Helsinki, Department of Medicine, Helsinki, Finland; [2]University of Tampere, Faculty of Medicine and Life Sciences, Tampere, Finland

INTRODUCTION

- Immunomodulatory treatments in multiple sclerosis (MS) aim to reduce disease activity and prevent progression.[1]
- Platform-injectable therapies are usually well tolerated, and serious adverse effects are rare.[2]
- Due to efficacy and tolerability issues the optimal treatment success in MS remains challenging.[3]
- Information on the long-term disability progression and adherence to treatment in real-life is needed to help the management of MS.

OBJECTIVES

- To assess factors that relate to treatment adherence and disability progression among relapsing MS patients with interferon β-1a s.c tiw as the first treatment, in a retrospective long-term real-life cohort.

METHODS

Study Design

- Confirmed relapsing MS patients diagnosed during 1981-2010 in Western Finland were scrutinized to fulfill inclusion criteria for the first treatment with interferon β-1a s.c tiw 22µg or 44µg, information on EDSS from treatment start and causes of discontinuation from 1996 to 2010.

Statistical Analysis

- Kaplan-Meier (KM) was applied to study median time to discontinuation by causes categorized in groups: side effects, disease activation, pregnancy plan and antibody formation (MXA titer).
- Longitudinal linear regression models were applied to assess risk of disability progression during the treatment period.
- Odd's ratios (OR) with 95% confidence intervals (95% CI) were calculated for gender, age at diagnosis, treatment delay (time from diagnosis to treatment start), dose of interferon β-1a s.c tiw, baseline EDSS and EDSS change in one year.

RESULTS

Table 1. Characteristics of the study population

		n	%
Total		293	100
Men		86	29.4
Women		207	70.6
Interferon β-1a s.c tiw 22µg		252	86.0
Interferon β-1a s.c tiw 44µg		41	14.0
	Mean	**Median**	**SD**
Age at diagnosis, years	33.6	32.0	9.5
Age at treatment start, years	36.4	36.0	10.1
Time from diagnosis to treatment, years	2.8	1.0	4.6
EDSS at treatment start	1.7	1.0	1.6
Duration of treatment period, years	2.9	1.9	2.89

- A total of 293 cases were included. At treatment start EDSS was 3.0 or less in 253 cases (86%). Mean duration of treatment episode during the 14-year follow-up was 2.9 years.
- Disease activation in 29.0% (n=59) and side effects 26.4% (n=54) were the main causes of discontinuation in 204 cases (70.0 %).
- Median survival time to discontinuation was one year in side-effect group (flu like reactions, laboratory values or skin problems) and two years in group where disease activation and pregnancy plans were cause of discontinuation (**Figure 1.**)

Figure 1. Time from interferon β-1a s.c. tiw start to treatment discontinuation by cause

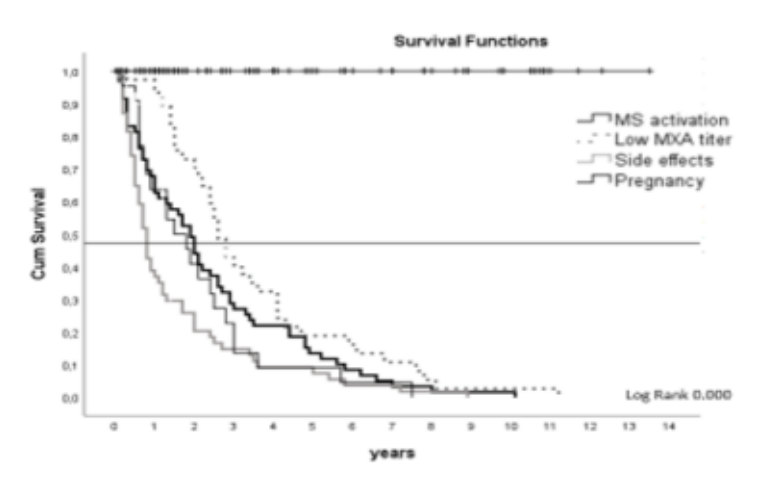
Survival Functions
Cum Survival
MS activation
Low MXA titer
Side effects
Pregnancy
Log Rank 0.000
years

Risk of disability progression: yearly EDSS changes

- Yearly EDSS changes by treatment duration during the follow up are given in **Figures 2 and 3.** The mean EDSS during the first 10 years of treatment points at mild disability (3.0 or less).

Figure 2. Observed absolute EDSS values in yearly intervals from treatment start

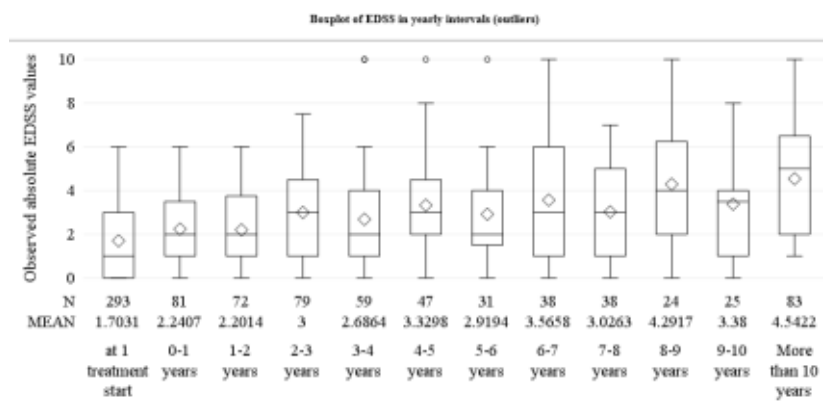
Boxplot of EDSS in yearly intervals (outliers)
Observed absolute EDSS values
N 293 81 72 79 59 47 31 38 38 24 25 83
MEAN 1.7031 2.2407 2.2014 3 2.6864 3.3298 2.9194 3.5658 3.0263 4.2917 3.38 4.5422
at 1 treatment start
0-1 years
1-2 years
2-3 years
3-4 years
4-5 years
5-6 years
6-7 years
7-8 years
8-9 years
9-10 years
More than 10 years

Figure 3. Observed absolute EDSS values from treatment start in groups of 0-1, >1.0 - 3.0 and > 3.0

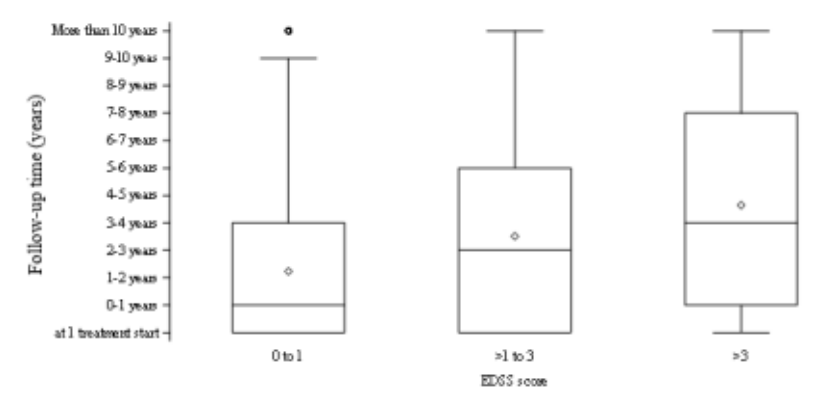
Follow-up time (years)
More than 10 years
9-10 years
8-9 years
7-8 years
6-7 years
5-6 years
4-5 years
3-4 years
2-3 years
1-2 years
0-1 years
at 1 treatment start
0 to 1
>1 to 3
>3
EDSS score

Risk of disability progression: longitudinal EDSS analysis

- In longitudinal analysis a low baseline EDSS (0-1) as compared to EDSS 3.0 or higher showed a statistically significant lower risk (OR 0.25, 95% CI 0.1-0.6) for disease progression during the treatment period. An increased risk related to one-year increase vs. no increase in EDSS during the treatment phase (1.20, 1.1-1.3). A 10-year difference in age at diagnosis was a significant risk factor (1.40, 1.0-1.9).
- No risk was shown for gender (OR 0.67, 0.4-1.2), for yearly delay to treatment start from diagnosis (1.05, 1.0-1.1), nor for initial dose of interferon β-1a s.c tiw (1.0, 0.4-2.3) (**Figure 4**.).

Figure 4. Longitudinal linear regression model. Prognostic clinical factors for risk of disability progression.

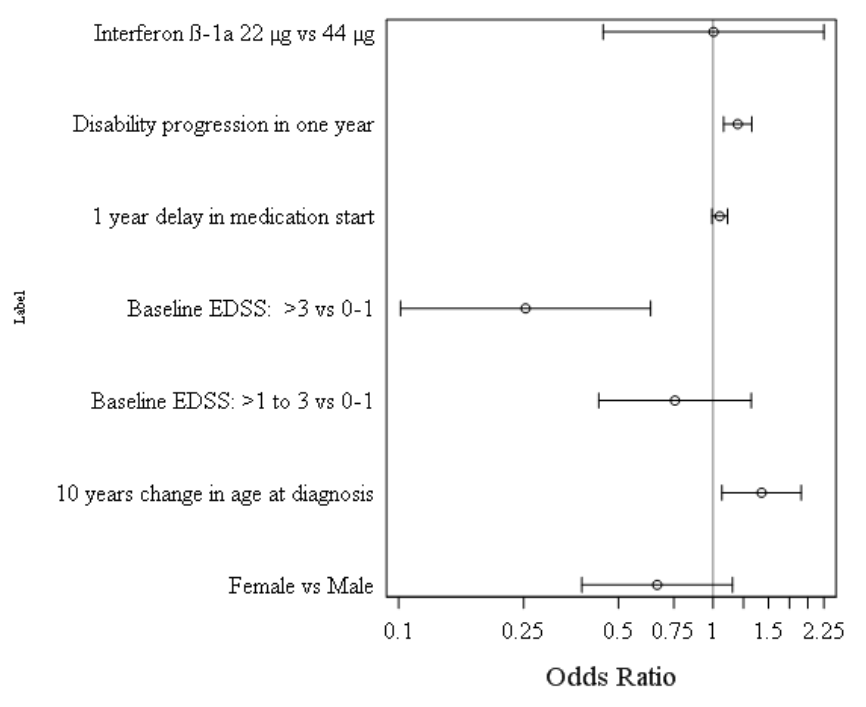
Interferon ß-1a 22 µg vs 44 µg
Disability progression in one year
1 year delay in medication start
Baseline EDSS: >3 vs 0-1
Baseline EDSS: >1 to 3 vs 0-1
10 years change in age at diagnosis
Female vs Male
Label
0.1
0.25
0.5
0.75
1
1.5
2.25
Odds Ratio

CONCLUSIONS

- **No disability at treatment start, (EDSS 0-1.0) and young age at diagnosis were related to a lower disability progression risk**
- **Side effects, pregnancy plans, and disease activation affected treatment adherence**
- **The results point at the importance of active monitoring for disability status**
- **Regular follow-up at treatment phase and actions to diminish side-effects are needed to reach the goal of efficacious and tolerated long-term treatment with interferon β-1a s.c tiw**

REFERENCES

1. Giovannoni G. *Curr Opin Neurol*. 2018; 31: 1-11
2. Bayas A, et al. *Expert Opin. Drug Deliv*. 2015;
3. Ziemssen T, et al. J Neurol. 2015;

ACKNOWLEDGEMENTS

This study was sponsored by Merck Finland, Helsinki. Finland*. The authors would like to thank Pasi Korhonen (StatFinn) and Melina Tervomaa (StatFinn) for help in statistical analysis and Pedro Testi (Merck Research Clinical Operations) for coordination of the study.
*An affiliate of Merck KGaA, Darmstadt, Germany.

DISCLOSURES

E.J. Is an employee of Merck Finland, an affiliate of Merck KGaA, Darmstadt, Germany.

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors.