Poster 22

Design of the non-interventional, prospective study CLEVER (CLadribine Tablets – EValuation of thERapy satisfaction)

Tjalf Ziemssen[1], Torsten Wagner[2], Anita Posevitz-Fejfár[2]

[1]Neurologische Klinik und Poliklinik, Universitätsklinikum Carl Gustav Carus an der Technischen Universität Dresden, MS-Zentrum, Germany;
[2]Merck Serono GmbH, Darmstadt, Germany

INTRODUCTION

- Receiving marketing authorization in Germany on August 22nd, 2017, Cladribine tablets represent the first oral short-course treatment indicated for adult patients with relapsing multiple sclerosis (RMS)[1].
- To date, there are no data on patients' treatment satisfaction with Cladribine tablets.

OBJECTIVES

- This study aims to prospectively evaluate treatment satisfaction in the initial treatment phase with Cladribine tablets

METHODS

Study Design

- Non-Interventional Study (NIS) in RMS patients treated with Cladribine tablets (3,5 mg/kg body weight, administered as per label requirements) (Fig. 1 and 2).
- The primary endpoint was defined as overall treatment satisfaction 24 weeks after treatment initiation by the means of TSQM-1.4.
- Secondary endpoints include management, efficacy and tolerability (TSQM-1.4, week 4 vs week 24), patients' characteristics and profile prior to Cladribine treatment, such as prior MS treatments, disease severity (number of relapses and lesions) and evaluation of predictors of treatment satisfaction.
- Furthermore, the impact of participation in a Patient Support Program (PSP) is evaluated.
- Participants of this NIS have the possibility to take part in PASS as a continuation for long term observation of safety aspects of Cladribine tablets.

Population

- Recruited patients (at treatment initiation or within 24 weeks after treatment initiation) are first-time users of Cladribine tablets.
- A signed informed consent is required.
- Patients with per label contraindications are excluded.

Variables

- Demographic data, MS and medication history, course of disease (level of disability), laboratory values and safety data.
- Questionnaires: TSQM (total and subscores).
- Usage and service related experience and PSP participation

Data Sources

- Data will be collected by means of an e-CRF using the data documentation system MSDS[3D].
- Paper questionnaires will be completed by the patient and transferred into the eCRF by the Study nurse.

Figure 1: Study design

CLEVER

- Start of study: Nov 2017
- End of recruitment: Dec 2019
- Documentation period per patient: 6 months
- Interim analysis after complete documentation of 350 (50%) patients
- Final Report: Dec 2020

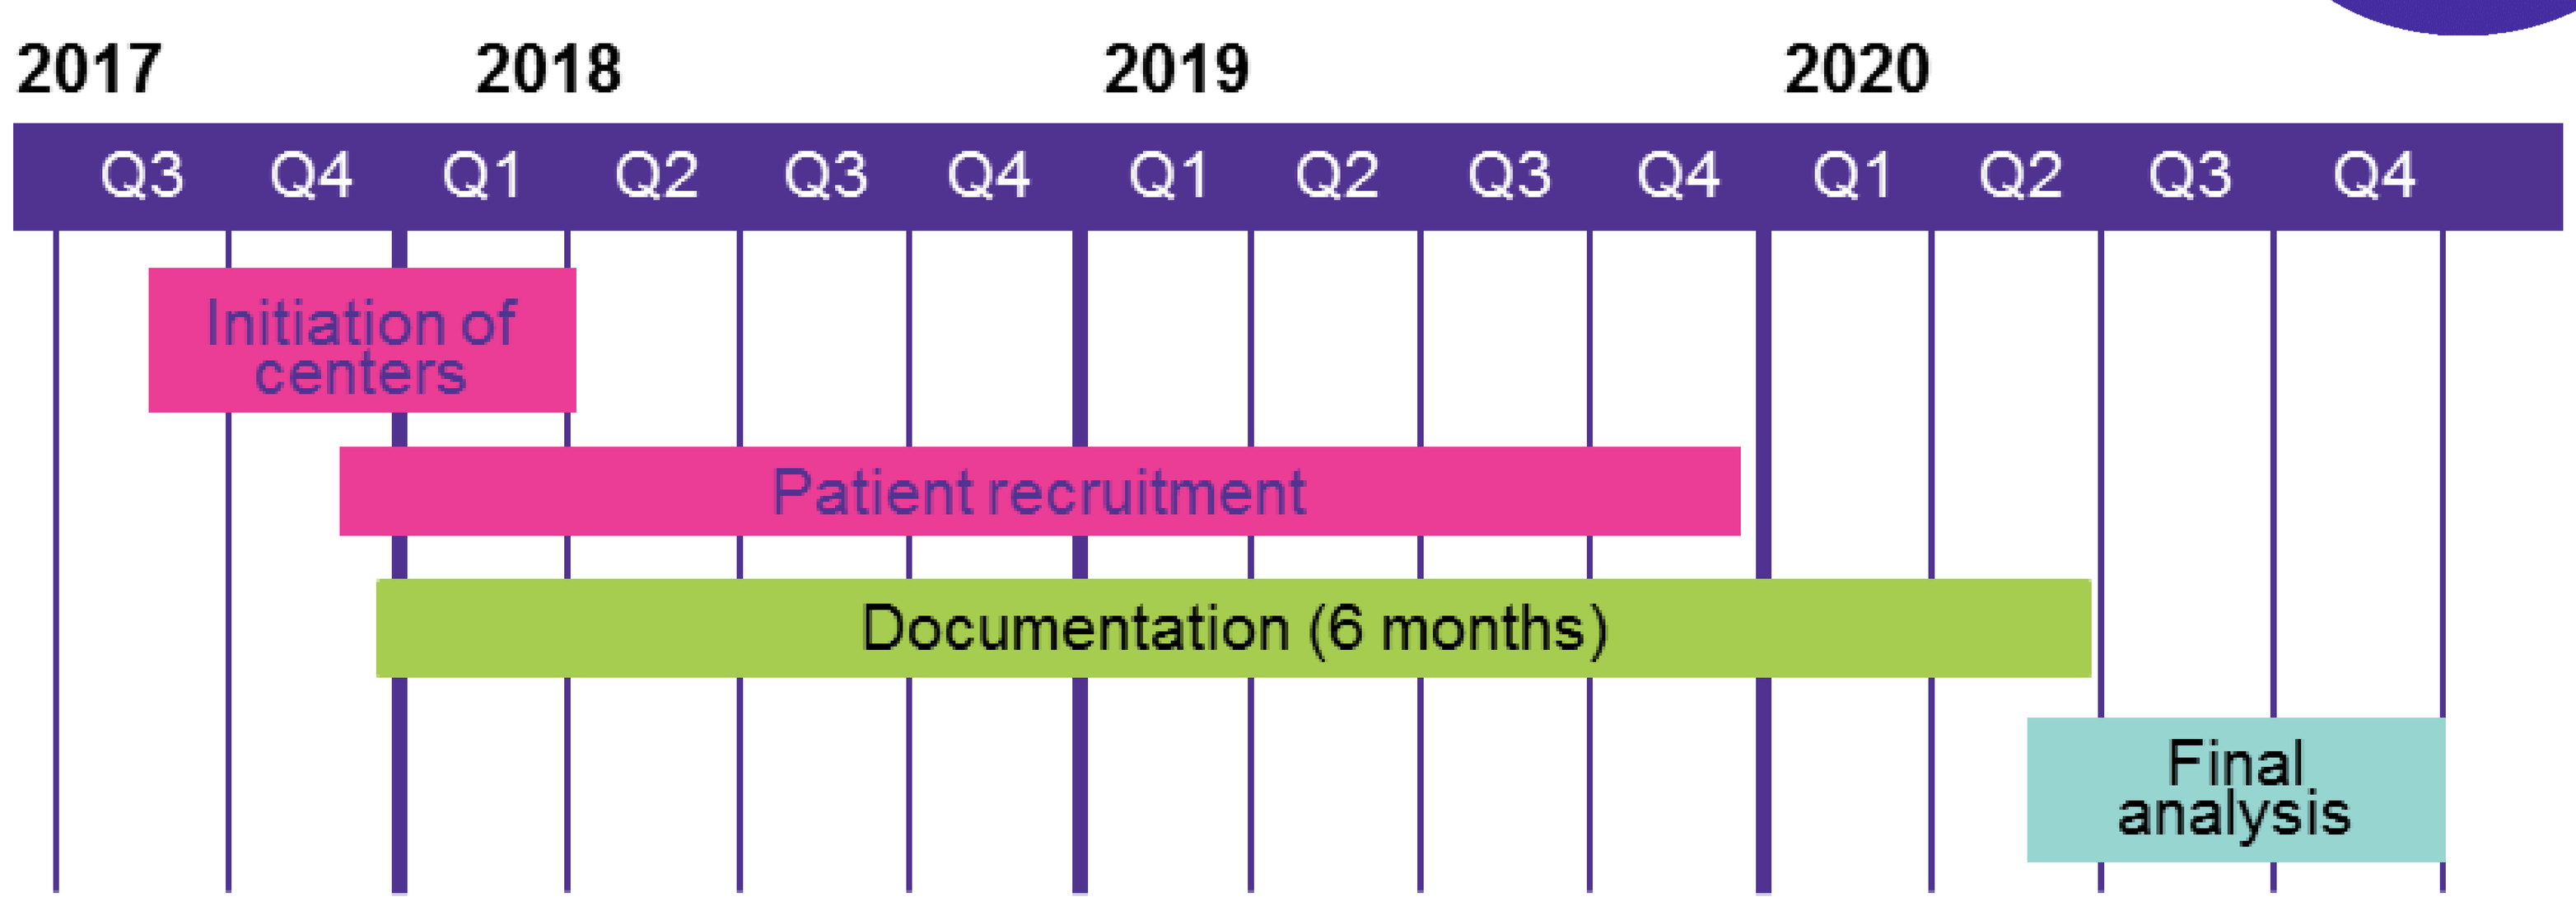
2017
2018
2019
2020
Q3 Q4 Q1 Q2 Q3 Q4 Q1 Q2 Q3 Q4 Q1 Q2 Q3 Q4
Initiation of centers
Patient recruitment
Documentation (6 months)
Final analysis

Figure 2: Study size, timeline and cladribine treatment

- Study size:
 - 700 patients with RRMS who have been prescribed Cladribine tablets according to the German SPC.
 - 100 study centers in Germany
- Inclusion of 1st patient: January 16th, 2018

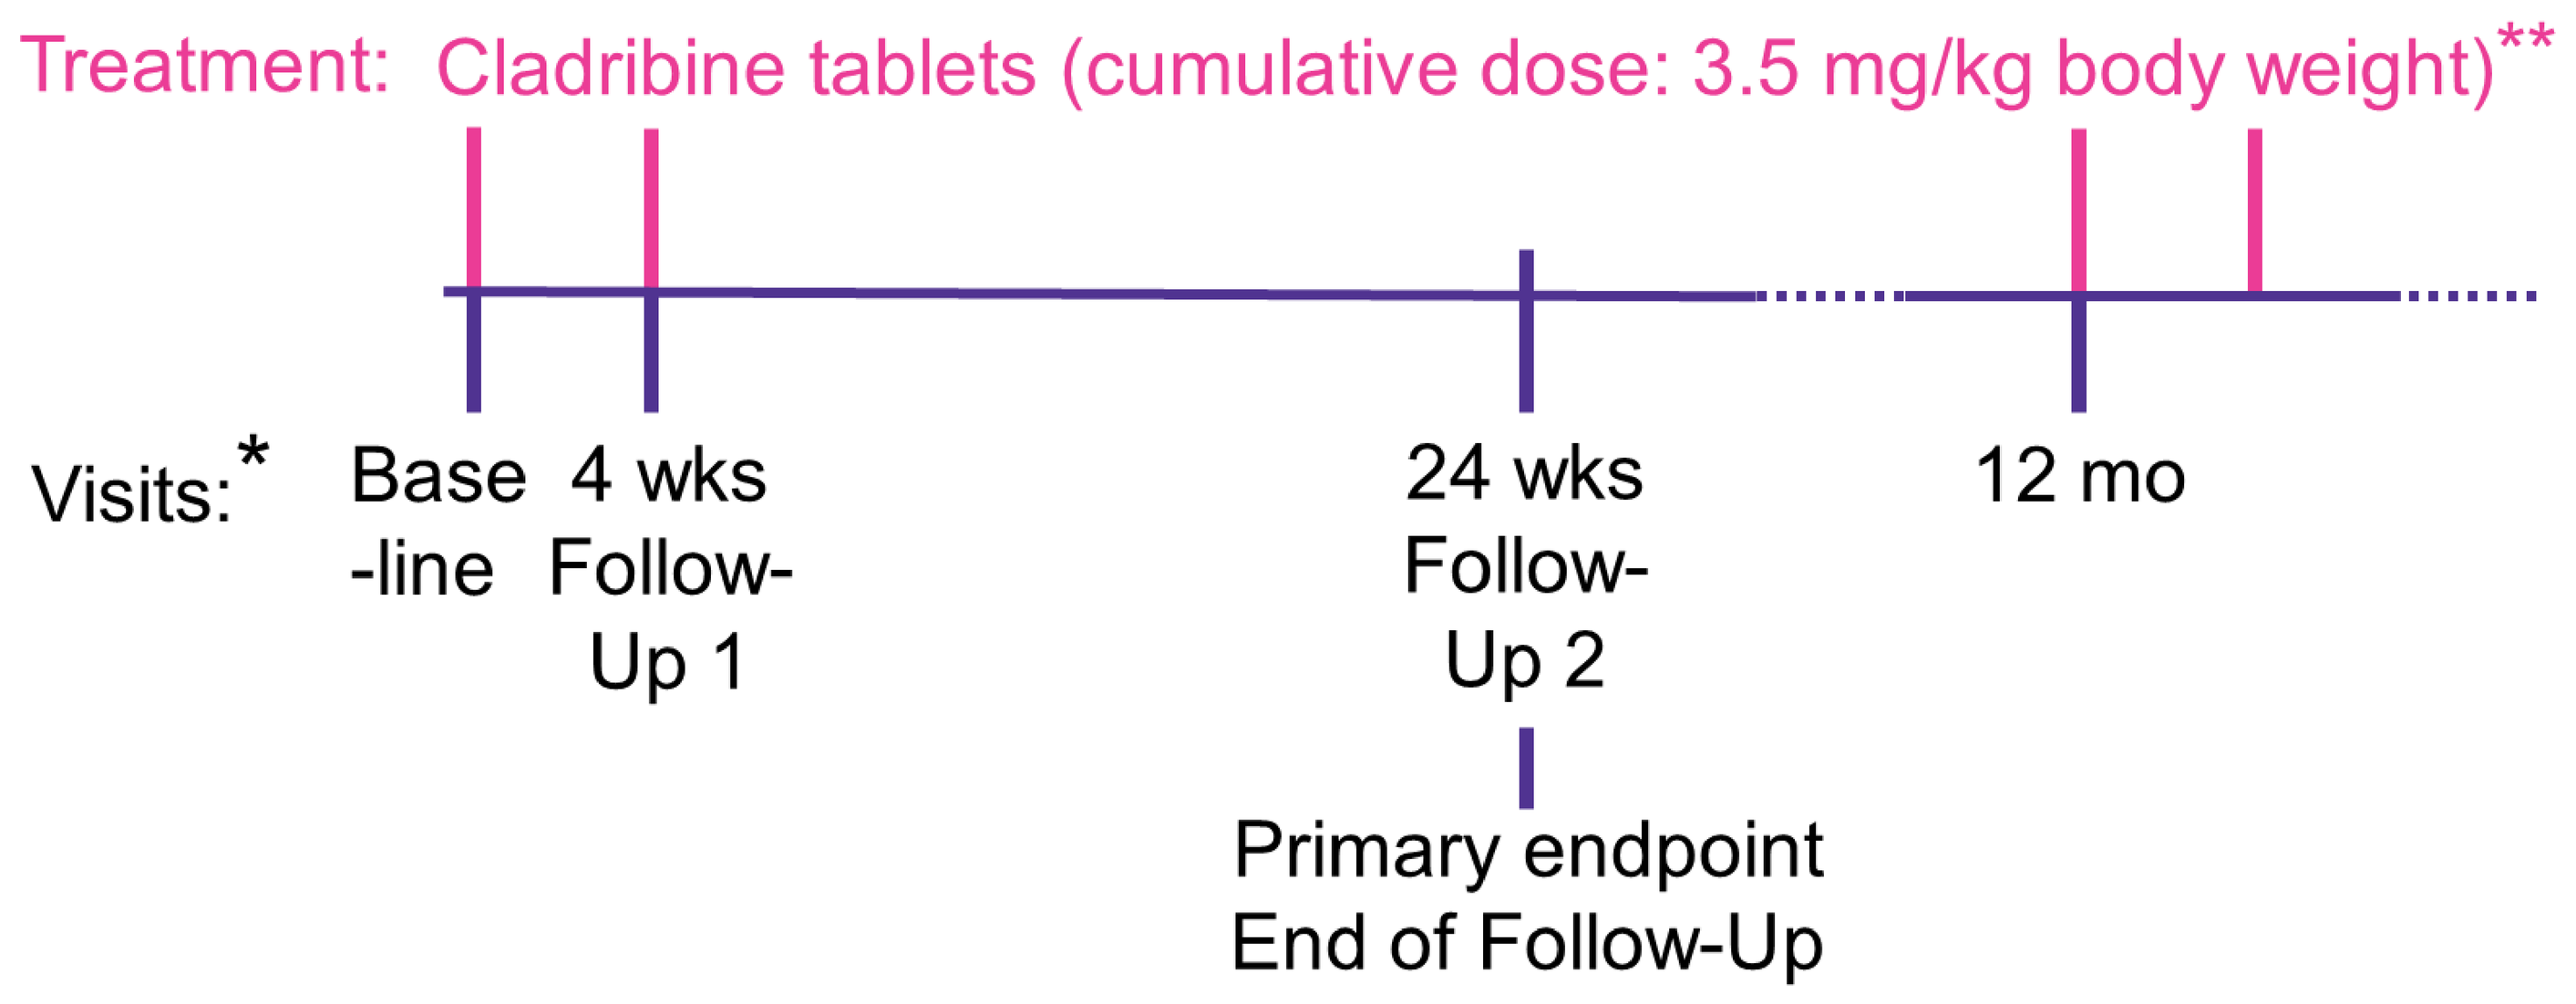
Treatment: Cladribine tablets (cumulative dose: 3.5 mg/kg body weight)**
Visits:*
Base-line
4 wks Follow-Up 1
24 wks Follow-Up 2
12 mo
Primary endpoint
End of Follow-Up

* Records produced at baseline, at week 4 (prescription of the tablets for the second treatment phase) and at week 24 (expected monitoring visit).
** Cladribine tablets: 3.5 mg/kg body weight over 2 years, 1 treatment course (2 treatment weeks, beginning of the 1st and 2nd month of the respective treatment year. Each treatment week consists of 4 or 5 days on which a patient receives 10 mg or 20 mg (one or two tablets) as a single daily dose, depending on body weight.

Table 1: Observation plan

Assessment	Screening	Baseline	Documentation of visit (weeks)		Study termination (loss to FU, exclusion, IC withdrawal)
			4*	24*	
Demography	-	✓	-	-	-
Medical History	-	✓	-	-	-
Treatment Satisfaction (TSQM)	-	✓	✓	✓	✓
Usage related questionnaire	-	-	✓	✓	✓
Service related questionnaire	-	-	✓	✓	✓
PSP participation	-	✓	✓	✓	✓
Reason for study termination	-	-	-	-	✓

Documented if recorded: EDSS score, 9-HPT score, T25-FW score, MRT (number of new lesions), relapse rate, laboratory values
FU: follow-up; IC: Informed Consent; * Documentation during routine visits according to clinical practice and drug approval
Medical History: MS, malignancies, infections; History of therapy: DMD therapies, immunosuppressants; MS history: number of relapses, number of new MRT lesions (active T1 and new T2 lesions) within 12 months, degree of disability

CONCLUSION

The study investigates treatment satisfaction of RMS patients in the initial treatment phase with Cladribine tablets. This important patient-reported outcome will provide valuable additional information on secondary benefits of Cladribine tablets.

REFERENCES

1. SmPC MAVENCLAD 10 mg tablets (May 2018)

DISCLOSURES

TZ has received reimbursements for participation in scientific advisory boards from Bayer, Biogen, Novartis, Merck, Teva and Genzyme. He has also received speaker honorarium from Bayer, Biogen, Genzyme, Merck, Novartis, Teva, Sanofi and Almirall. TW and APF are employees of Merck Serono GmbH, Darmstadt, Germany.

ACKNOWLEDGEMENTS

This trial was sponsored by Merck KGaA, Darmstadt, Germany. The authors would like to thank the patients, investigators, co-investigators, and the study teams at each of the participating centers and at Merck KGaA, Darmstadt, Germany, and Merck Serono*, Darmstadt.
Medical writing assistance was provided by med:unit and funded by Merck Serono*, Darmstadt.
*An affiliate of Merck KGaA, Darmstadt, Germany.

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors.

GET POSTER PDF

Presented at 26th Annual Meeting of the European Charcot Foundation; 15 – 17 November, 2018; Baveno, Italy.