Efficacy of a Third Course of Alemtuzumab in Patients With Active Relapsing-Remitting Multiple Sclerosis Who Experienced Disease Activity After the Initial Two Courses: Pooled Analysis of CARE-MS I and II

Patrick Vermersch[1], Anthony Traboulsee[2], Aaron Boster[3], Ann D Bass[4], Regina Berkovich[5], Giancarlo Comi[6], Óscar Fernández[7], Ho Jin Kim[8], Volker Limmroth[9], Jan Lycke[10], Richard AL Macdonell[11], Basil Sharrack[12], Heinz Wiendl[13], Tjalf Ziemssen[14], Maria Melanson[15], Nadia Daizadeh[15], Barry A Singer[16];
on behalf of the CARE-MS I, CARE-MS II, and CAMMS03409 Investigators

[1]University of Lille, Lille, France; [2]University of British Columbia, Vancouver, BC, Canada; [3]OhioHealth Neurological Physicians, Columbus, OH, USA; [4]Neurology Center of San Antonio, San Antonio, TX, USA; [5]University of Southern California, Keck School of Medicine, Los Angeles, CA, USA; [6]University Vita-Salute San Raffaele, Milan, Italy; [7]Fundación IMABIS, Hospital Universitario Carlos Haya, Málaga, Spain; [8]Research Institute and Hospital of National Cancer Center, Goyang, South Korea; [9]Klinik für Neurologie und Palliativmedizin, Cologne, Germany; [10]University of Gothenburg, Gothenburg, Sweden; [11]Austin Health and Florey Institute of Neuroscience and Mental Health, Melbourne, VIC, Australia; [12]Sheffield Teaching Hospitals NHS Foundation Trust, Sheffield, UK; [13]University of Münster, Münster, Germany; [14]Center of Clinical Neuroscience, Carl Gustav Carus University Hospital, Dresden, Germany; [15]Sanofi, Cambridge, MA, USA; [16]MS Center for Innovations in Care, Missouri Baptist Medical Center, St Louis, MO, USA

OBJECTIVE

- **To evaluate efficacy of one alemtuzumab retreatment in pooled CARE-MS I and II patients who received only 1 additional alemtuzumab treatment (Course 3) due to relapse and/or MRI activity**

INTRODUCTION

- In CARE-MS I (NCT00530348) and II (NCT00548405), 2 courses of alemtuzumab resulted in significantly greater improvements on clinical and MRI outcomes versus SC IFNB-1a over 2 years[1,2]
- The most frequent adverse events (AEs) with alemtuzumab were infusion-associated reactions (IARs); other AEs of interest included autoimmune AEs[1,2]
- Alemtuzumab-treated patients who were followed up for an additional 4 years in an extension study (NCT00930553) experienced durable efficacy in the absence of continuous treatment; 63% of CARE-MS I and 50% of CARE-MS II patients did not receive additional alemtuzumab or other disease-modifying therapy (DMT)[3-7]
- The durable effects of alemtuzumab may be due to its selective depletion and distinct pattern of repopulation of circulating CD52-expressing T and B lymphocytes[8,9]
 - Following depletion, a relative increase in regulatory T cells and a decrease in proinflammatory cytokines occurs, potentially leading to a rebalancing of the immune system[10,11]
 - The exact mechanism of action of alemtuzumab is not fully elucidated

METHODS

Patients

- Patients in CARE-MS I and II had active RRMS and were either treatment-naive (CARE-MS I) or had an inadequate response to prior therapy (CARE-MS II) at core study baseline[1,2]

Treatment

- In CARE-MS I and II, patients received 2 courses of alemtuzumab 12 mg/day IV (on 5 consecutive days at baseline and on 3 consecutive days 12 months later)
- In the extension study, patients could receive additional treatment with alemtuzumab (on 3 consecutive days ≥12 months after the most recent course) as needed for relapse or MRI activity, or receive other licensed DMTs at the investigator's discretion
 - As-needed alemtuzumab retreatment criteria included: ≥1 protocol-defined relapse, or ≥2 new/enlarging T2 hyperintense and/or new gadolinium (Gd)-enhancing T1 brain or spinal cord lesions on MRI
 - Retreatment disqualification criteria included (but were not limited to): pregnancy, diagnosis of immune thrombocytopenia (ITP) or other immune cytopenia, anti-glomerular basement membrane disease, and history of malignancy (except basal cell carcinoma)

Statistical Analyses

- Post hoc analyses were performed on data through Year 6 (end of the fourth year of the extension study) to assess outcomes in patients who received only 1 alemtuzumab retreatment (ie, Course 3)
 - Patients were included in efficacy and safety analyses based on the following criteria:
 - Course 3 was received prior to Month 61 to allow for ≥1 year of follow-up after retreatment
 - No other DMTs were received through Year 6
- Results were compared with the cohort of patients who received no alemtuzumab retreatment or other DMT after the initial 2 courses of alemtuzumab
- Annualized relapse rate (ARR) was estimated using a negative binomial model[a]
- Kaplan-Meier estimates were used to assess proportions of patients with 6-month confirmed disability improvement (CDI) and 6-month confirmed disability worsening (CDW)

RESULTS

Patients

- 811 patients received alemtuzumab 12 mg in the CARE-MS I and II studies, with 790 (97%) completing the core studies; 742/790 (94%) entered the extension study and 669/742 (90%) remained on study through Month 72 (Year 6)
- 302/742 (41%) patients who entered the extension received alemtuzumab retreatment through Year 6
- Of these 302 patients, the majority received only 1 alemtuzumab retreatment (ie, Course 3) through Year 6 (**Figure 1A**)
 - Course 3 was most frequently given in Year 3 (**Figure 1B**)
 - Mean time from Course 2 to Course 3 was 2.6 years (median [range]: 2.6 years [1.0–5.0 years])

CONCLUSIONS

- **In CARE-MS patients who received only 1 alemtuzumab retreatment (Course 3) due to relapse and/or MRI activity, alemtuzumab effectively reduced relapses and improved disability without further treatment**
 - **Efficacy outcomes, including disability improvement, were favorable and clinically meaningful in the years following Course 3**
- **There were no apparent changes in the safety profile of alemtuzumab after 1 retreatment (Course 3), including autoimmune AEs, infections, or malignancies**
- **These data support the favorable benefit-risk of administering additional alemtuzumab treatment in patients with disease activity following Course 2 to achieve durable disease control**

Figure 1. Patients Receiving Alemtuzumab Retreatment Through Year 6

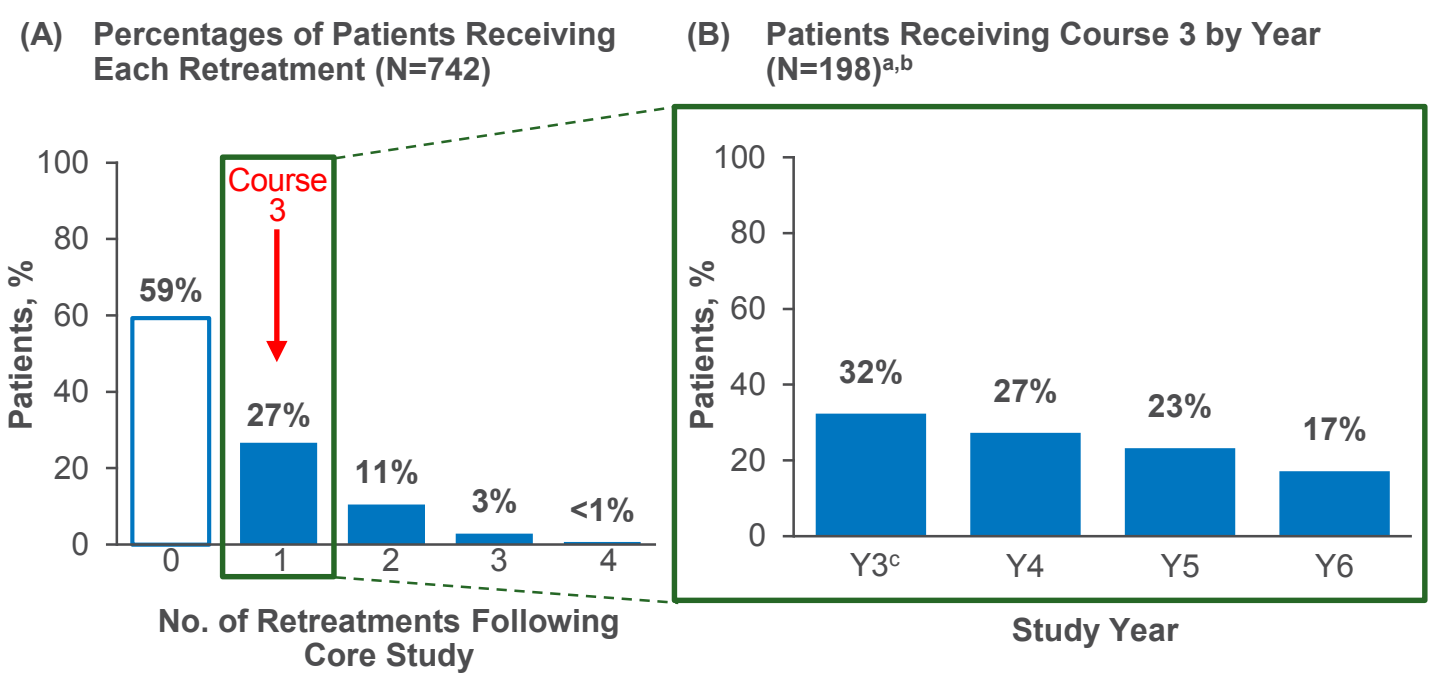
(A) Percentages of Patients Receiving Each Retreatment (N=742)
(B) Patients Receiving Course 3 by Year (N=198)a,b
Patients, %
100
80
60
40
20
0
Course 3
59%
27%
11%
3%
<1%
0
1
2
3
4
No. of Retreatments Following Core Study
32%
27%
23%
17%
Y3c
Y4
Y5
Y6
Study Year

Y=year; percentages may not equal 100% due to rounding
[a]All patients received a third course of alemtuzumab within 60 months of Course 2; [b]All patients received only 1 retreatment through Year 6; [c]3 patients received Course 3 <12 months after Course 2, on Days 353, 358, and 359 after Course 2

- Of the 198 patients who received only 1 retreatment through Year 6, 143 fulfilled criteria of receiving Course 3 before Month 61 (to allow for 1-year follow-up after Course 3) and receiving no DMT to be included in efficacy and safety analyses
- Baseline characteristics were comparable among patients who received only 1 retreatment, no retreatment, and the overall study population (**Table 1**)
 - Disease duration was slightly longer in the only 1 retreatment group, compared with both the no retreatment group and the overall study population

Table 1. Baseline Characteristics in Alemtuzumab-Treated Patients With and Without Retreatment Through Year 6

Parameter	All CARE-MS I/II (N=811)	No Retreatment[a,b] (N=409)	Only One Retreatment[b,c] (N=143)
Age, years	34.0 (8.2)	34.0 (8.2)	34.0 (8.1)
Female, n (%)	530 (65.4)	258 (63.1)	99 (69.2)
White, n (%)	744 (91.7)	383 (93.6)	134 (93.7)
EDSS score	2.4 (1.1)	2.3 (1.1)	2.3 (1.2)
Years since initial relapse	3.4 (2.5)	3.1 (2.5)	3.7 (2.5)
No. of relapses in prior 1 year	1.7 (0.8)	1.8 (0.8)	1.7 (0.8)
No. of relapses in prior 2 years	2.7 (1.1)	2.6 (1.0)	2.6 (1.1)
Gd-enhancing lesion count	2.3 (5.6)	2.1 (4.7)	2.3 (4.9)
Brain parenchymal fraction	0.82 (0.02)	0.82 (0.02)	0.82 (0.02)

[a]All patients received only the initial 2 courses of alemtuzumab in the core studies and no retreatment in extension; [b]All patients received no other DMT through 6 years; [c]Course 3 had to occur between core study baseline and Month 61 to allow for 1-year post-Course 3 data
All values are mean (SD) unless indicated otherwise

Clinical Efficacy

- In patients who received only 1 retreatment:
 - ARR significantly declined after retreatment and remained low over the following 3 years (**Figure 2A**); ARR was similar to relapse rates in patients who received no retreatment (**Figure 2B**)
 - Change in mean EDSS 12 months after Course 3 was -0.12, and the percentage with stable/improved EDSS scores increased to 71% after retreatment (**Figure 3A**); 88%–90% of those who were not re-treated had stable/improved EDSS scores over Years 3–6
 - Despite the need for retreatment, the percentage free from CDW was 97% both 12 months before and 12 months after Course 3; 82%–89% of patients who were not re-treated were free from CDW over Years 3–6 (CDW: ≥1-point EDSS increase [or ≥1.5 points if baseline EDSS=0] confirmed over 6 months)
 - The percentage with CDI significantly increased to 17.5% after retreatment (**Figure 3B**); 35%–45% of patients who were not re-treated achieved CDI over Years 3–6

Figure 2. Relapse Rates in Patients Who Received (A) Only One Alemtuzumab Retreatment and (B) No Retreatment

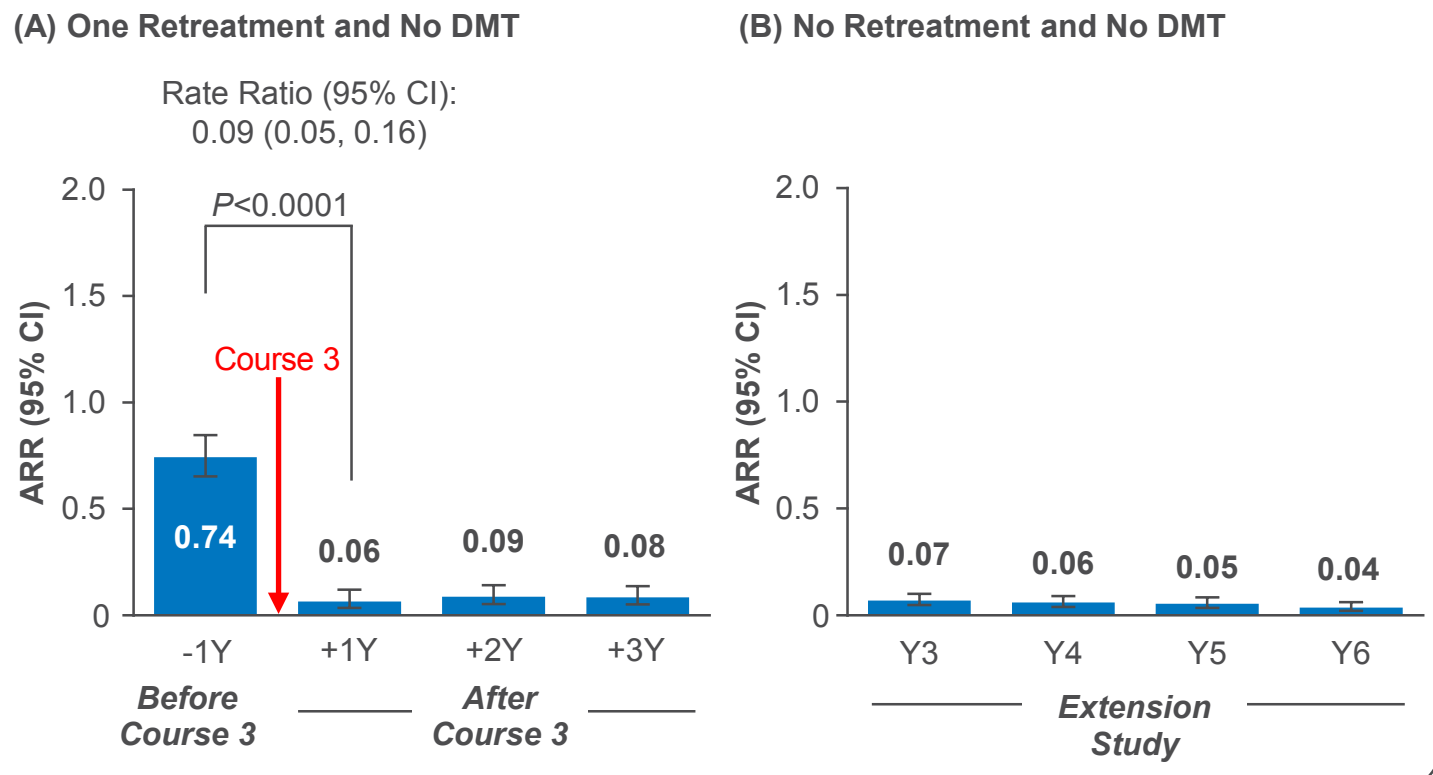
(A) One Retreatment and No DMT
Rate Ratio (95% CI): 0.09 (0.05, 0.16)
P<0.0001
Course 3
ARR (95% CI)
2.0
1.5
1.0
0.5
0
0.74
0.06
0.09
0.08
-1Y
+1Y
+2Y
+3Y
Before Course 3
After Course 3
(B) No Retreatment and No DMT
0.07
0.06
0.05
0.04
Y3
Y4
Y5
Y6
Extension Study

Figure 3. Percentage of Patients (A) With Stable/Improved EDSS and (B) With 6-Month CDI Increased After Only One Alemtuzumab Retreatment

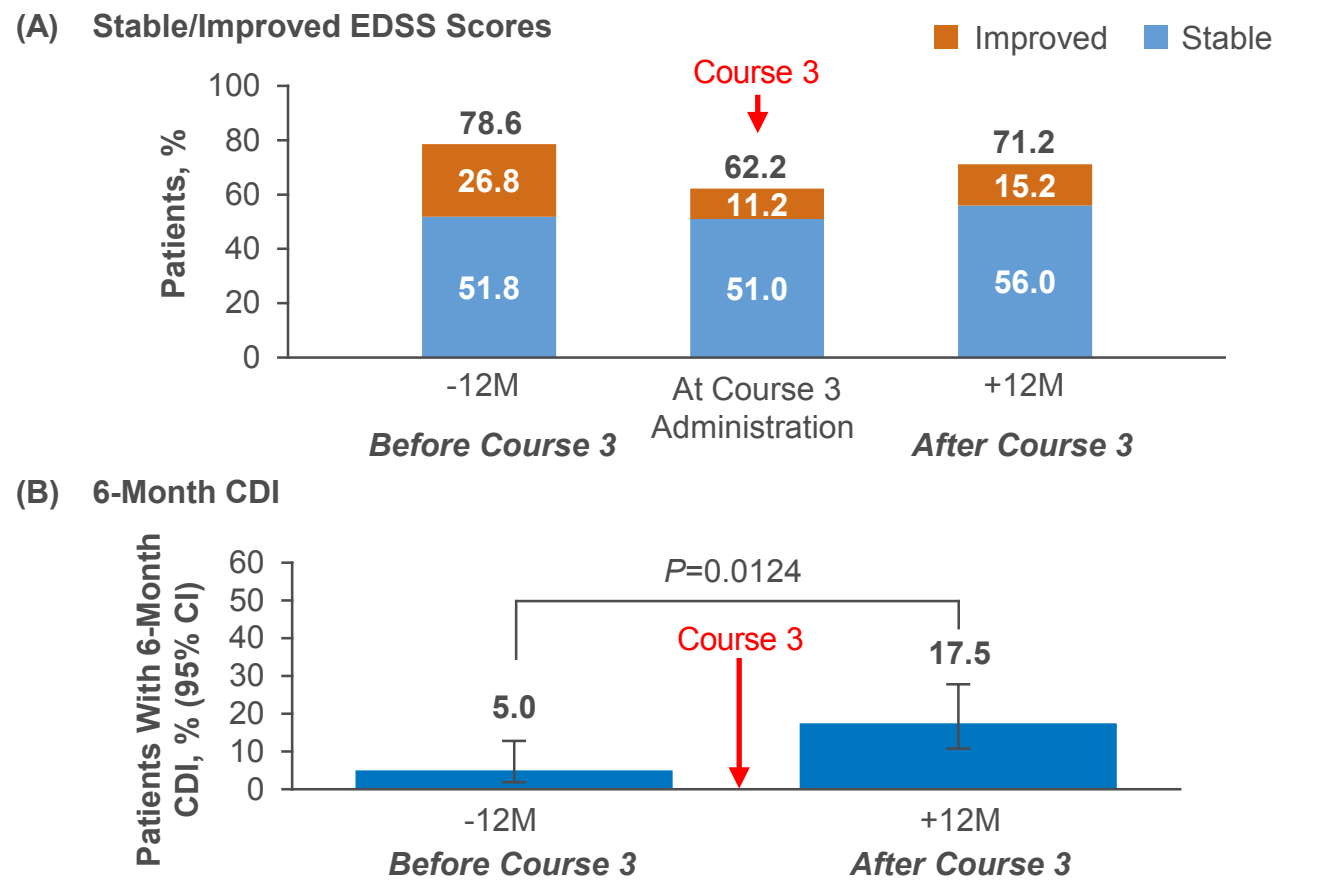
(A) Stable/Improved EDSS Scores
Improved
Stable
Course 3
Patients, %
100
80
60
40
20
0
78.6
26.8
51.8
62.2
11.2
51.0
71.2
15.2
56.0
-12M
At Course 3 Administration
+12M
Before Course 3
After Course 3
(B) 6-Month CDI
Patients With 6-Month CDI, % (95% CI)
60
50
40
30
20
10
0
P=0.0124
Course 3
5.0
17.5
-12M
+12M
Before Course 3
After Course 3

M=month; improved EDSS score: ≥1.0-point decrease from core study baseline; stable EDSS score: ≤0.5-point change in either direction from core study baseline
CDI: ≥1-point EDSS decrease from baseline confirmed over 6 months; CDI is assessed only in patients with baseline EDSS score ≥2.0; CDI baseline for -12M is defined as the most recent measurement prior to the 12-month period before Course 3; CDI baseline for +12M is defined as the measurement just prior to Course 3; *P*-value for comparison between -12M CDI and +12M CDI is based on McNemar's test for statistical significance

Safety

- No patients withdrew from the extension study due to AEs
- Incidences of AEs, including serious AEs, infections and serious infections, were similar between patients who received only 1 alemtuzumab retreatment and those who received no retreatment (**Table 2**)
- IAR and serious IAR incidences decreased by course, and were similar to that of the overall study population
- Incidences of thyroid AEs were similar between patients who received only 1 alemtuzumab retreatment, and both those who received no retreatment and the overall study population, with low reported incidences of serious thyroid AEs
- Incidences of ITP and nephropathies were low in patients who received only 1 retreatment (ITP: 1.4%; nephropathies: 0.7%) and patients who received no retreatment (ITP: 2.2%; nephropathies: 0%)
- Incidences of malignancies were low and similar between patients who received only 1 alemtuzumab retreatment (2.1%), and both those who received no retreatment (2.2%) and the overall study population (2.0%)
- No deaths were reported in patients receiving alemtuzumab retreatment

Table 2. AE Incidences in Alemtuzumab-Treated Patients With and Without Retreatment Through Year 6

	Alemtuzumab 12 mg		
	Incidence, n (%)		
	All CARE-MS I/II (N=811)	No Retreatment[a,b] (N=409)	Only One Retreatment[b,c] (N=143)
Any AE	800 (98.6)	404 (98.8)	143 (100)
Serious AEs	289 (35.6)	135 (33.0)	42 (29.4)
Infections	677 (83.5)	341 (83.4)	125 (87.4)
Serious infections	53 (6.5)	27 (6.6)	6 (4.2)
Autoimmune AEs[d]			
Thyroid AEs	343 (42.3)	197 (48.2)	59 (41.3)
Serious thyroid AEs	41 (5.1)	27 (6.6)	5 (3.5)
ITP	21 (2.6)	9 (2.2)	2 (1.4)
Nephropathies	2 (0.2)	0	1 (0.7)
Malignancies	16 (2.0)	9 (2.2)	3 (2.1)
IARs	739 (91.1)	369 (90.2)	134 (93.7)
Serious IARs	26 (3.2)	11 (2.7)	4 (2.8)

[a]All patients received only the initial 2 courses of alemtuzumab in the core studies and no retreatment in extension; [b]All patients received no other DMT through 6 years; [c]Course 3 had to occur between core study baseline and Month 61 to allow for 1-year post-Course 3 data; [d]First occurrence of AE

Sensitivity Analysis

- Efficacy and safety were also analyzed for the cohort of patients who received ≥1 alemtuzumab retreatment before Month 61 and no DMT through Year 6; including those who received only 1 retreatment (Course 3) and those who received >1 retreatment; data were censored from the time of the second retreatment (Course 4) onward to assess the efficacy of the first retreatment (Course 3)
- 253/742 (34%) patients fulfilled criteria for the ≥1 retreatment group
 - Of these, 156 (62%) and 71 (28%) received 1 and 2 retreatments, respectively
- Efficacy outcomes (ie, ARR, percentage with stable/improved EDSS, percentage free from 6-month CDW, and percentage with 6-month CDI) were similar to those in patients who received only 1 retreatment
- Incidences of AEs, IARs, and autoimmune AEs were similar to those in patients who received only 1 retreatment

References

1. Cohen JA, et al. *Lancet* 2012;380:1819-28.
2. Coles AJ, et al. *Lancet* 2012;380:1829-39.
3. Havrdova E, et al. *Neurology* 2017;89:1107-16.
4. Coles AJ, et al. *Neurology* 2017;89:1117-26.
5. Ziemssen T, et al. *Ther Adv Neurol Disord* 2017;10:343-59. 6. Coles AJ, et al. *Mult Scler* 2016;22:213. 7. Fox EJ, et al. *Mult Scler* 2016;22:P1150. 8. Cox AL, et al. *Eur J Immunol* 2005;35:3332-42. 9. Hu Y, et al. *Immunology* 2009;128:260-70. 10. Zhang X, et al. *J Immunol* 2013;191:5867-74. 11. De Mercanti S, et al. *Neurol Neuroimmunol Neuroinflamm* 2016;3:e194.

Acknowledgments and Disclosures

CARE-MS Steering Committees and CAMMS03409 Investigators. This poster was reviewed by Jordan Messer, PharmD, and Colin Mitchell, PhD, of Sanofi. Editorial support for the poster was provided by Rebecca L Orndorff, PhD, and Panos Xenopoulos, PhD, of Envision Scientific Solutions, and was funded by Sanofi. CARE-MS I, CARE-MS II, and the extension study were funded by Sanofi and Bayer HealthCare Pharmaceuticals. **PV:** Consulting and/or speaking fees, and research support (Almirall, Bayer, Biogen, Celgene, Merck Serono, Novartis, Sanofi, Servier, and Teva). **AT:** Consulting and/or speaking fees, and grant/research support (Biogen, Chugai, Roche, Sanofi, and Teva). **AB:** Consulting fees and/or fees for non-CME services (Biogen, Mallinckrodt, Medtronic, Novartis, Sanofi, and Teva). **ADB:** Consulting fees/fees for non-CME services from commercial interest or their agents/grant and research support (Biogen, Mallinckrodt, Novartis, Roche-Genentech, Sanofi, and Teva Neuroscience). **RB:** Advisory boards and consulting (Acorda, Avanir, Bayer, Biogen, Novartis, Questcor, Sanofi, and Teva). **GC:** Consulting fees (Actelion, Bayer, Merck Serono, Novartis, Sanofi, and Teva); lecture fees (Bayer, Biogen Dompé, Merck Serono, Novartis, Sanofi, Serono Symposia International Foundation, and Teva). **OF:** Speaking and/or consulting (Allergan, Almirall, Bayer-Schering, Biogen, Merck Serono, Novartis, Sanofi, and Teva); compensation for serving as a journal editor, associate editor, or member of an editorial advisory board (*Revista Española de Esclerosis Múltiple*); and research support (Hospital Foundation FIMABIS). **HJK:** Consulting and/or speaking fees (Bayer, Biogen, Genzyme, HanAll BioPharma, MedImmune, Merck Serono, Novartis, Teva-Handok, and UCB); research support (Genzyme, Kael-GemVax, Merck Serono, MSIP [Ministry of Science, ICT & Future Planning], Teva-Handok, and UCB); steering committee member (MedImmune); co-editor (*Multiple Sclerosis Journal – Experimental, Translational, and Clinical*); and associate editor (*Journal of Clinical Neurology*). **VL:** Honoraria for consulting and speaking at symposia (Bayer, Biogen, Merck Serono, Novartis, Roche, Sanofi, and Teva, with approval by the HR Department, Cologne General Hospital, and University of Cologne). **JL:** Lecture honoraria (Novartis) and unconditional research support (Biogen, Novartis, and Teva). **RALM:** Compensation for advisory board and/or speaking fees (Bayer, Biogen, Merck, Novartis, Roche, Sanofi, and Teva); research support (Biogen, Merck, Novartis, Sanofi, and Teva). **BSh:** Nothing to disclose. **HW:** Consulting and/or speaking fees (Bayer, Biogen, Behring, EMD Serono, Fresenius Medical Care, Merck Serono, Novartis, Sanofi, Roche, and Teva); license fee payments (Huber-Verlag); and grant/research support (Neotope Bioscience, Novartis, and PML Consortium). **TZ:** Consulting and/or speaking fees (Almirall, Bayer, Biogen, Merck, Novartis, Roche, Sanofi, and Teva); grant/research support (Biogen, Novartis, Sanofi, and Teva). **MM and ND:** Employees of Sanofi. **BAS:** Speaking and/or consulting (Acorda, Bayer, Biogen, EMD Serono, Genentech, Novartis, Sanofi, and Teva), and research support (Acorda, Alkermes, Biogen, MedImmune, Novartis, Roche, and Sanofi).

CARE-MS=Comparison of Alemtuzumab and Rebif® Efficacy in Multiple Sclerosis

Rebif® is a registered trademark of Merck Serono Europe Ltd.

Previously presented at the 7th Joint European Committee for Treatment and Research in Multiple Sclerosis (ECTRIMS) – Americas Committee for Treatment and Research in Multiple Sclerosis (ACTRIMS) Meeting, 25–28 October 2017, Paris, France.

Alemtuzumab is approved in >65 countries around the world for treatment of adults with relapsing forms of multiple sclerosis (MS). In the EU, it is approved to treat patients with relapsing-remitting MS with active disease defined by clinical or imaging features. In the US, the indication provides that, because of its safety profile, the use of alemtuzumab should be reserved for patients who generally have had an inadequate response to 2 or more drugs indicated for the treatment of MS. This material may contain information that is outside of the approved labeling in some countries.

[a]Relapses were confirmed per the protocol by an independent, blinded relapse adjudication panel in the core study and by the investigator in the extension study.

Presented at the 25th Annual Meeting of the European Charcot Foundation, 30 November – 2 December 2017, Baveno, Italy
Funding provided by Sanofi