ENSEMBLE PLUS Study Design: An Investigation of Shortened Ocrelizumab Infusion Time on Infusion-Related Reactions in Patients With Relapsing Multiple Sclerosis From the Phase IIIb ENSEMBLE PLUS Study

B Brochet,[1] C Nos,[2] MS Freedman,[3] J Killestein,[4] J Overell,[5] R Buffels,[6] K Fitovski,[6] L Mehta,[7] Q Wang,[6] H-P Hartung[8]

[1]University of Bordeaux, Bordeaux, France; [2]Multiple Sclerosis Center of Catalonia, Hospital Vall d'Hebron, Barcelona, Spain; [3]University of Ottawa and the Ottawa Hospital Research Institute, Ottawa, ON, Canada; [4]Department of Neurology, VU University Medical Center, Amsterdam, The Netherlands; [5]Institute of Neurological Sciences, Glasgow, UK; [6]F. Hoffmann-La Roche Ltd, Basel, Switzerland; [7]Genentech, Inc., South San Francisco, CA, USA; [8]Department of Neurology, UKD, Center of Neurology and Neuropsychiatry and LVR-Klinikum, Heinrich-Heine University Düsseldorf, Düsseldorf, Germany

BACKGROUND AND OBJECTIVE

- Ocrelizumab is an intravenously administered, humanised anti-CD20+ monoclonal antibody approved for the treatment of relapsing forms of multiple sclerosis (MS) and primary progressive MS
- The current approved total infusion time of ocrelizumab (600 mg dose) is 5.5–6 hours, and includes:
 - — Premedication/infusion preparation: 1 hour
 - — Infusion: 3.5–4 hours
 - — Post-infusion observation: 1 hour
- A reduction in infusion time may reduce the burden of administration on the patient, nursing staff and hospital facilities
- Infusion-related reactions (IRRs) are the most common ocrelizumab treatment-related adverse events (AEs)[1,2]
- IRR frequency is greatest during the first infusion and severity is generally mild-to-moderate[3]
 - — The frequency and grade of IRRs in patients with relapsing MS who received ocrelizumab in the pivotal Phase III study is presented in **Figure 1**
- IRR frequency/severity can be minimised by following the guidelines for the management of specific AEs (**Table 1**)

Figure 1. IRRs by infusion and grade[a] in relapsing multiple sclerosis patients receiving ocrelizumab 600 mg IV in pivotal Phase III OPERA I and OPERA II (pooled data) studies[3]

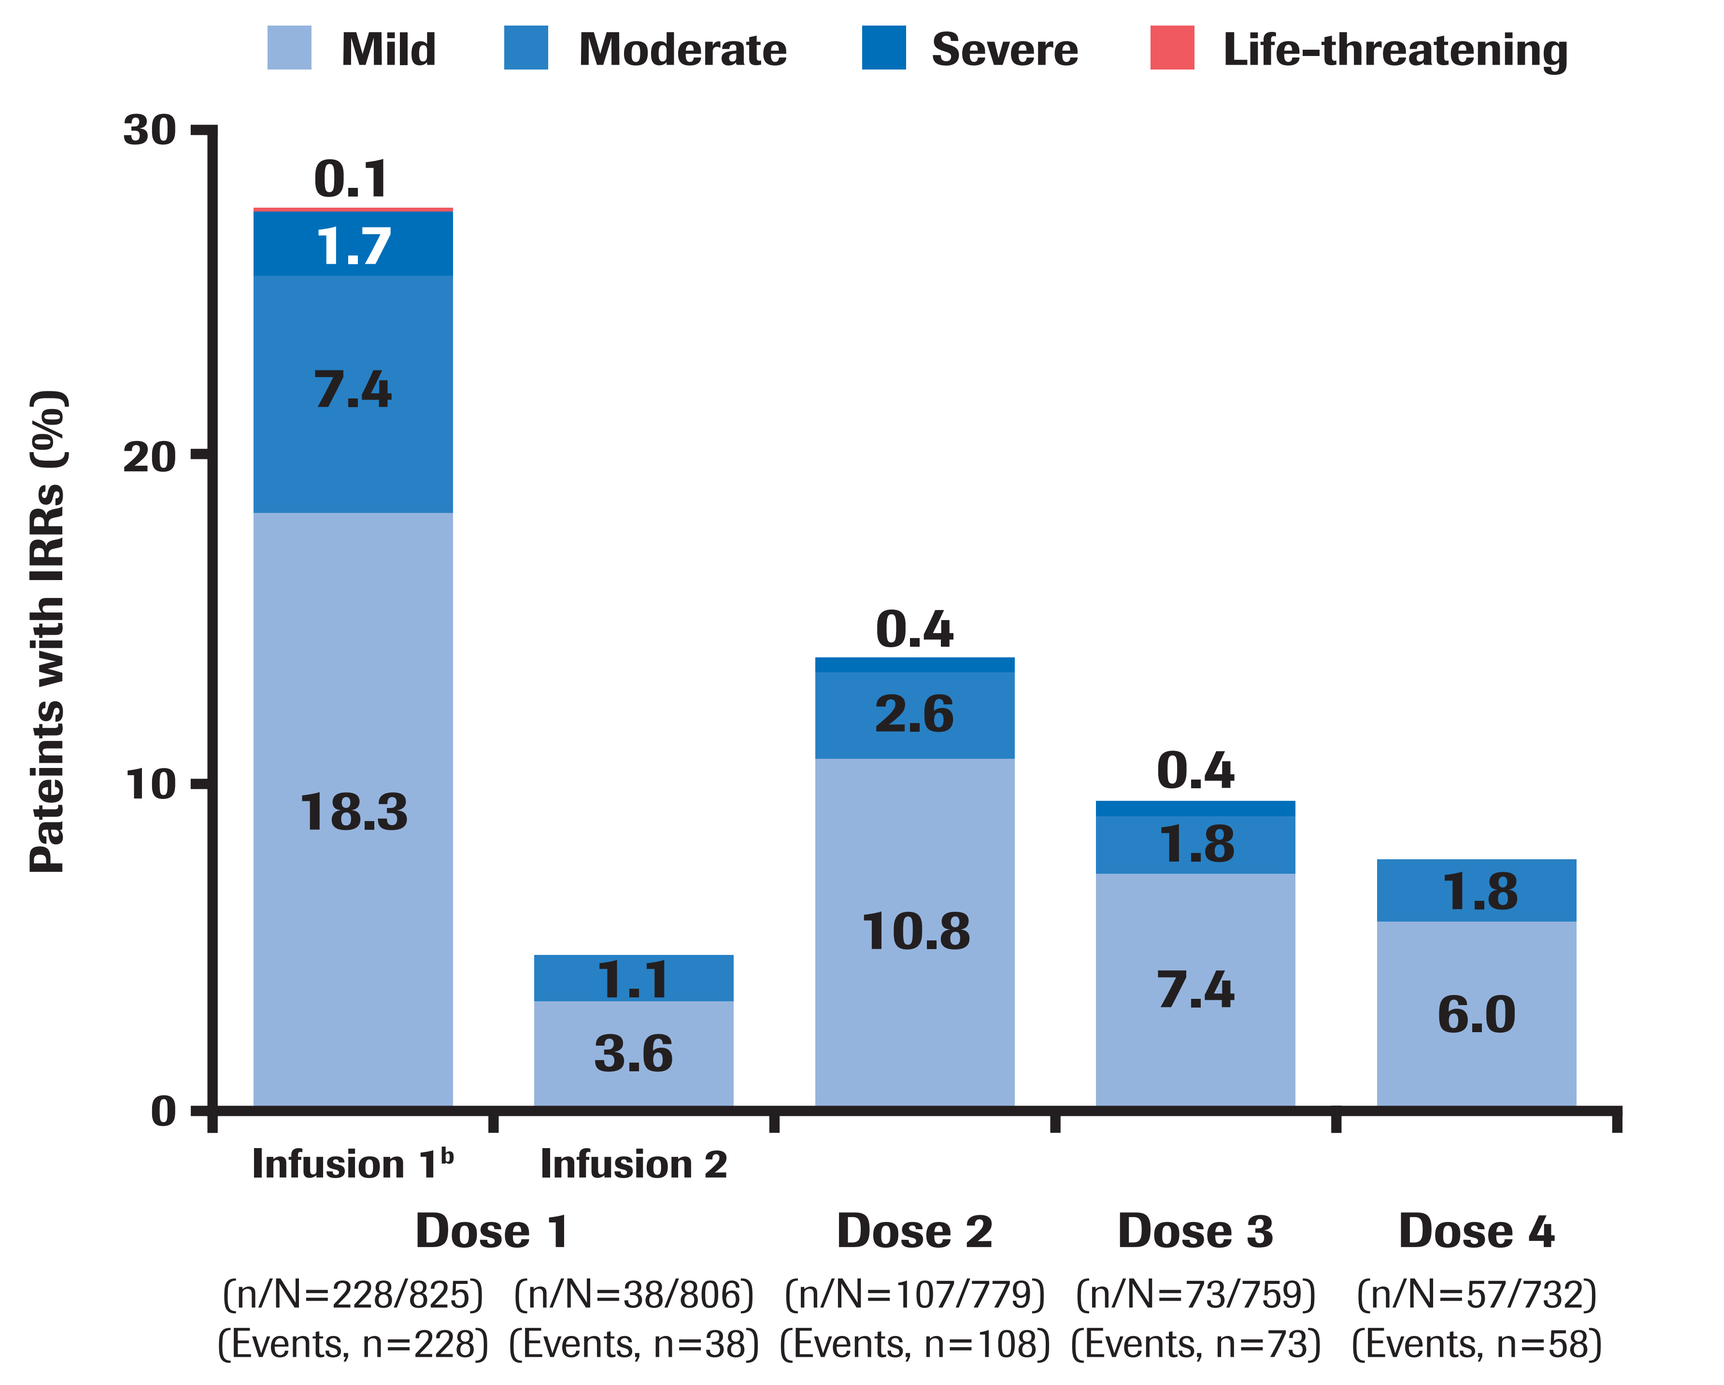
Mild
Moderate
Severe
Life-threatening
Pateints with IRRs (%)
30
20
10
0
0.1
1.7
7.4
18.3
1.1
3.6
0.4
2.6
10.8
0.4
1.8
7.4
1.8
6.0
Infusion 1[b]
Infusion 2
Dose 1
Dose 2
Dose 3
Dose 4
(n/N=228/825) (Events, n=228)
(n/N=38/806) (Events, n=38)
(n/N=107/779) (Events, n=108)
(n/N=73/759) (Events, n=73)
(n/N=57/732) (Events, n=58)

Premedication	Timing
Methylprednisolone 100 mg IV[c]	Slow infusion completed approximately 30 minutes prior to infusion
Antihistamine[d]	30–60 minutes prior to infusion
Antipyretic[e]	May also be considered

[a]Grading per Common Terminology Criteria for Adverse Events (CTCAE); [b]Eleven patients (1.3%) withdrew from ocrelizumab treatment due to an IRR during the first infusion.
IRR, infusion-related reaction; IV, intravenous. [c]Or equivalent; [d]e.g. diphenhydramine to reduce IRR frequency and severity; [e]e.g. paracetamol.

Table 1. Infusion pretreatment and guidelines for management of specific adverse events

Event	Action to be taken
Mild-to-moderate IRR	If the event that a patient experiences is a mild-to-moderate IRR (e.g. headache), the infusion rate should be reduced to half the rate at the time of the event. This reduced rate should be maintained for at least 30 minutes. If tolerated, the infusion rate may then be increased according to the patient's initial infusion schedule
Severe IRR (or complex of flushing, fever and throat pain)	If a patient experiences a severe IRR or a complex of flushing, fever, and throat pain symptoms, the infusion should be interrupted immediately and the patient should receive symptomatic treatment. The infusion should be restarted only after all symptoms have resolved. The initial infusion rate at restart should be half of the infusion rate at the time of onset of the reaction
Life-threatening or disabling IRR (e.g. anaphylaxis)	Immediately stop ocrelizumab if there are signs of a life-threatening or disabling IRR during an infusion, such as acute hypersensitivity or acute respiratory distress syndrome. The patient should receive appropriate treatment. Permanently discontinue ocrelizumab in these patients

IRR, infusion-related reaction.

- The impact of reducing infusion time on ocrelizumab-related IRRs will be investigated in ENSEMBLE PLUS
- ENSEMBLE PLUS is a substudy of an ongoing Phase IIIb, open-label, single-arm investigation of the effectiveness and safety of ocrelizumab in patients with early-stage relapsing-remitting MS (RRMS) (ENSEMBLE [NCT03085810])[4]
- The objective of this presentation is to describe the design of the ENSEMBLE PLUS substudy

METHODS

Study Design

- In the ENSEMBLE PLUS substudy, a subgroup of eligible patients from the main ENSEMBLE[4] study will be randomised with equal allocation into a conventional infusion group (infusion duration: 3.5 hours) and a shorter infusion group (infusion duration: 2 hours) at the next scheduled infusion under a double-blind setting
 - — In addition to the optional randomisation of patients from ENSEMBLE, ENSEMBLE PLUS will recruit an additional 300 patients (first enrolled within ENSEMBLE) for the evaluation of IRRs starting at the Week 24 infusion
- ENSEMBLE PLUS inclusion criteria are:
 - — Diagnosis of early-stage RRMS (per revised McDonald 2010 criteria)[5]
 - — Age 18–55 years (inclusive) and disease duration ≤3 years
 - — Expanded Disability Status Scale score 0–3.5 (inclusive) and treatment naive
 - — ≥1 relapse or T1 gadolinium-enhancing lesion in prior 12 months
 - — Inclusion in double-blind randomisation
 - — Disease-modifying therapy treatment naive
- Patients with prior serious ocrelizumab-related IRRs will be excluded from ENSEMBLE PLUS
- IRR frequency and severity will be assessed with each infusion administered after randomisation
- The design of ENSEMBLE PLUS is presented in **Figure 2**
- The location of ENSEMBLE PLUS study sites is presented in **Figure 3**

Figure 2. ENSEMBLE PLUS study design

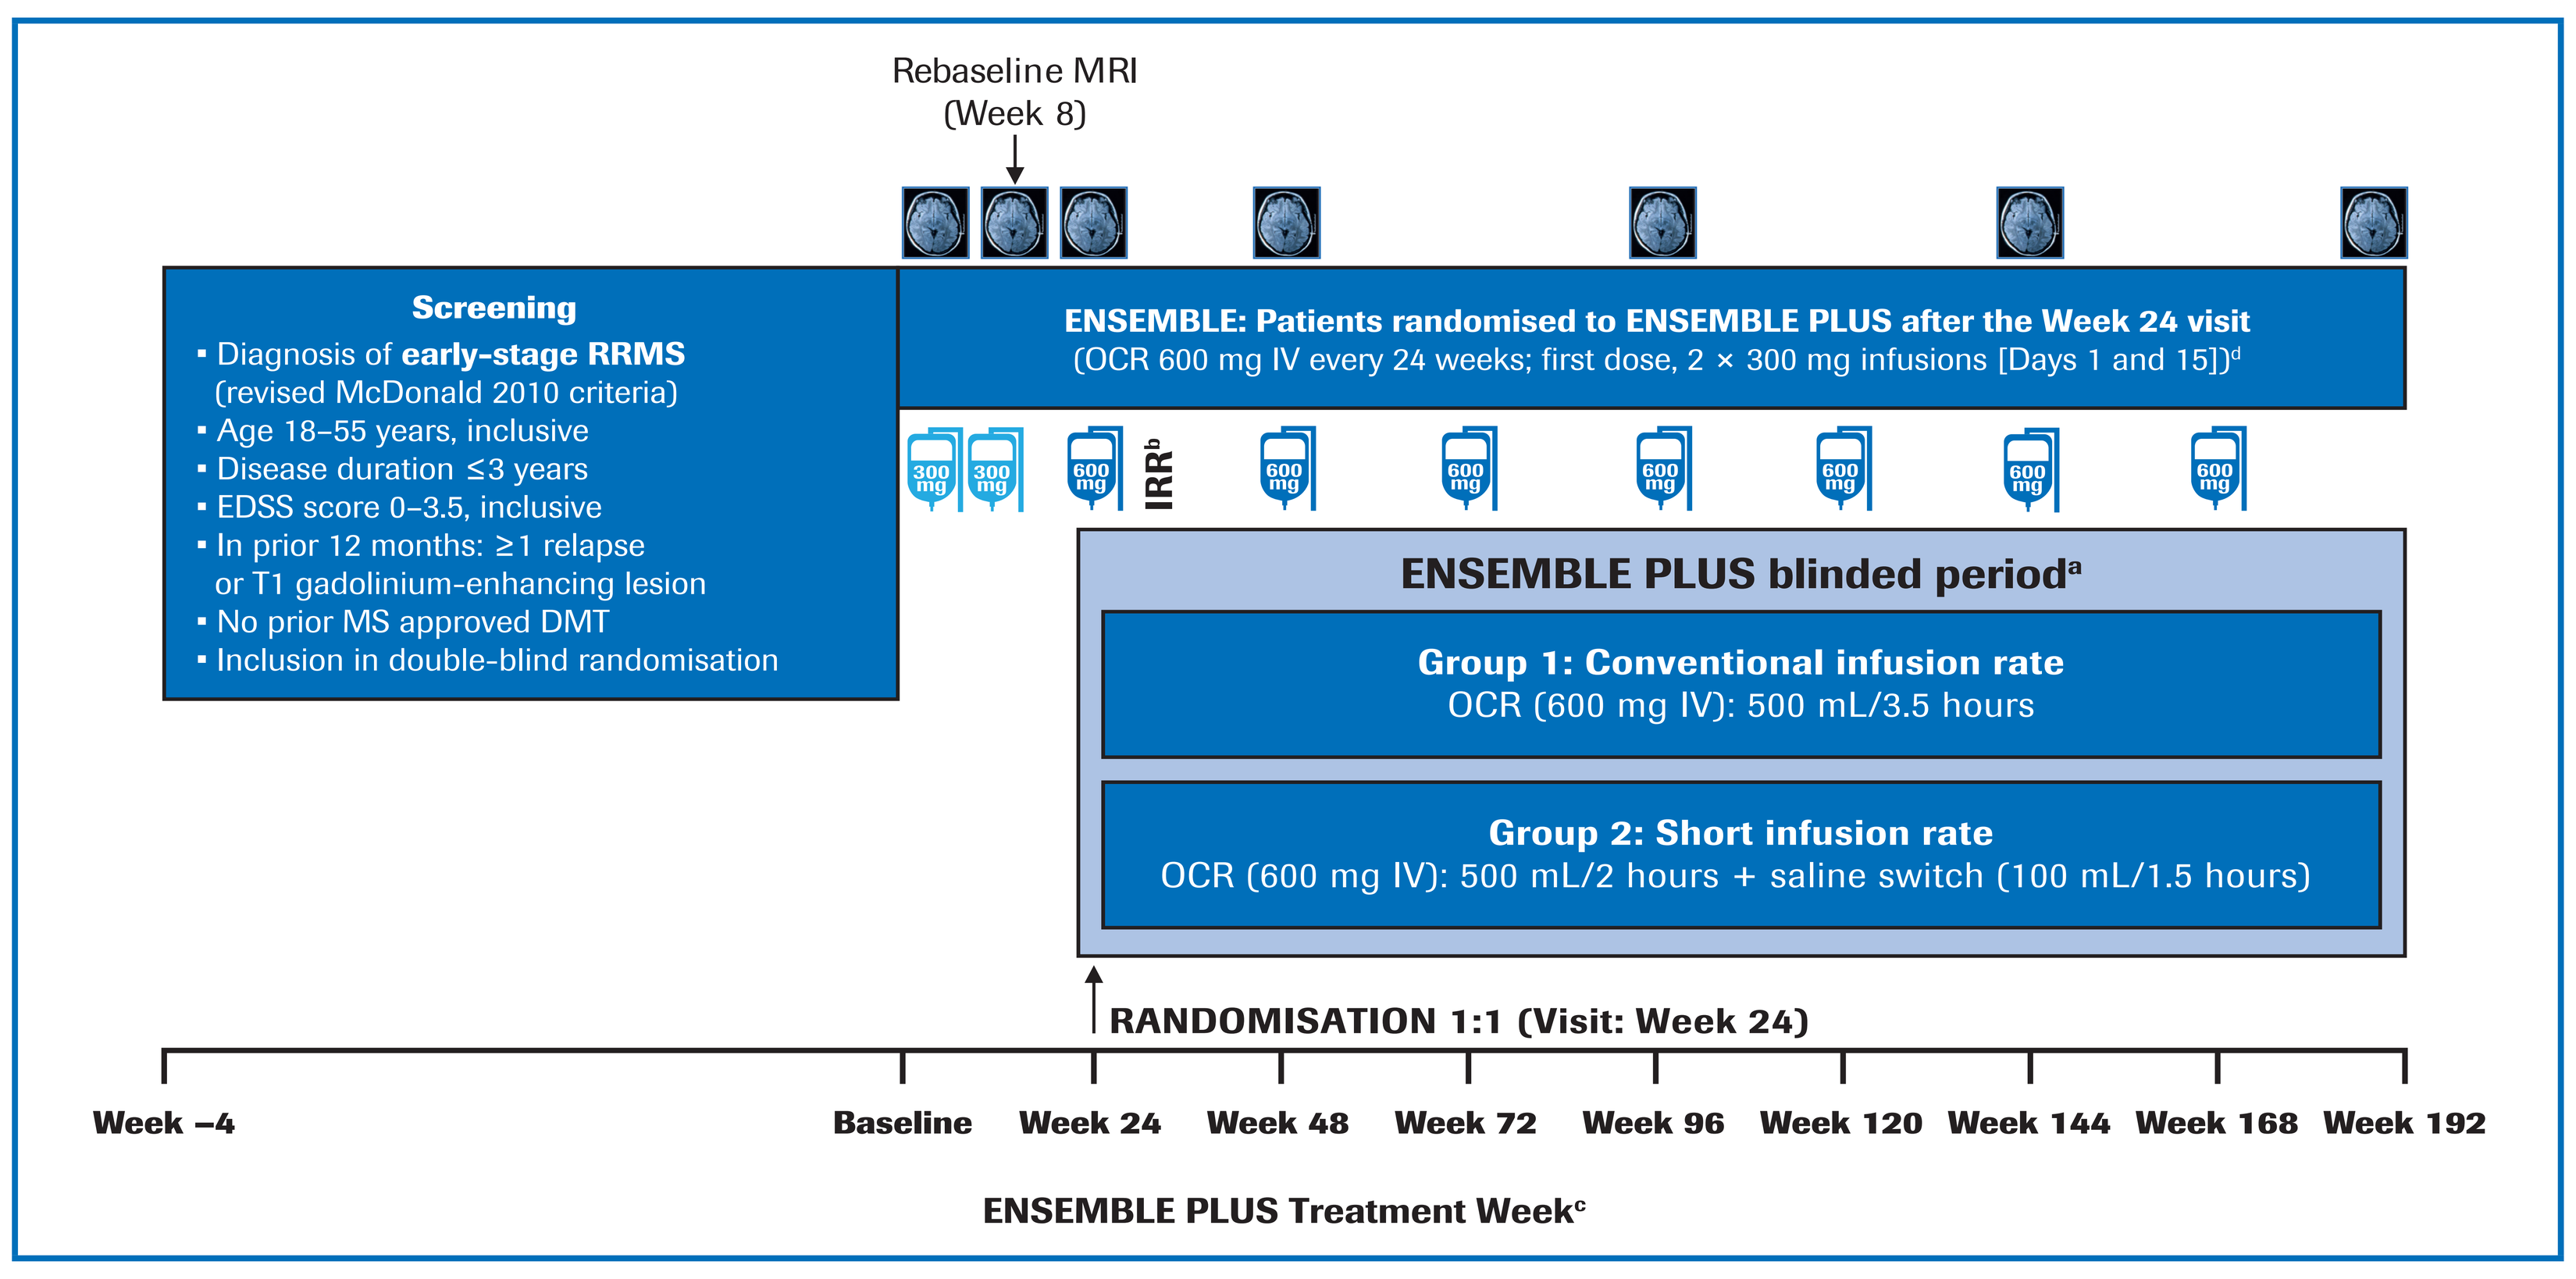
Rebaseline MRI (Week 8)
Screening
• Diagnosis of early-stage RRMS (revised McDonald 2010 criteria)
• Age 18–55 years, inclusive
• Disease duration ≤3 years
• EDSS score 0–3.5, inclusive
• In prior 12 months: ≥1 relapse or T1 gadolinium-enhancing lesion
• No prior MS approved DMT
• Inclusion in double-blind randomisation
ENSEMBLE: Patients randomised to ENSEMBLE PLUS after the Week 24 visit
(OCR 600 mg IV every 24 weeks; first dose, 2 × 300 mg infusions [Days 1 and 15])[d]
300 mg
600 mg
IRR[b]
ENSEMBLE PLUS blinded period[a]
Group 1: Conventional infusion rate
OCR (600 mg IV): 500 mL/3.5 hours
Group 2: Short infusion rate
OCR (600 mg IV): 500 mL/2 hours + saline switch (100 mL/1.5 hours)
RANDOMISATION 1:1 (Visit: Week 24)
Week −4
Baseline
Week 24
Week 48
Week 72
Week 96
Week 120
Week 144
Week 168
Week 192
ENSEMBLE PLUS Treatment Week[c]

[a]The study design is not final and might be changed based upon the recommendations of health authorities. Patients completing the blinded period can enter a long-term extension period (evaluation of long-term outcomes). Patients discontinuing early entered a safety follow-up period (48 weeks or until B-cell repletion); [b]IRR analyses were performed after the first randomised dose in ENSEMBLE PLUS; [c]Treatment week for patients first treated in ENSEMBLE will differ (for randomisation). Patients from the existing ENSEMBLE study can enter the ENSEMBLE PLUS study and can be randomised at any upcoming infusion; [d]Inclusion of ENSEMBLE patients in ENSEMBLE plus is optional and can be done at any infusion following provision of informed consent and randomisation.
DMT, disease-modifying therapy; Expanded Disability Status Scale; IRR, infusion-related reaction; IV, intravenous; MRI, magnetic resonance imaging; MS, multiple sclerosis; OCR, ocrelizumab; RRMS, relapsing-remitting multiple sclerosis.

Figure 3. ENSEMBLE PLUS study site locations

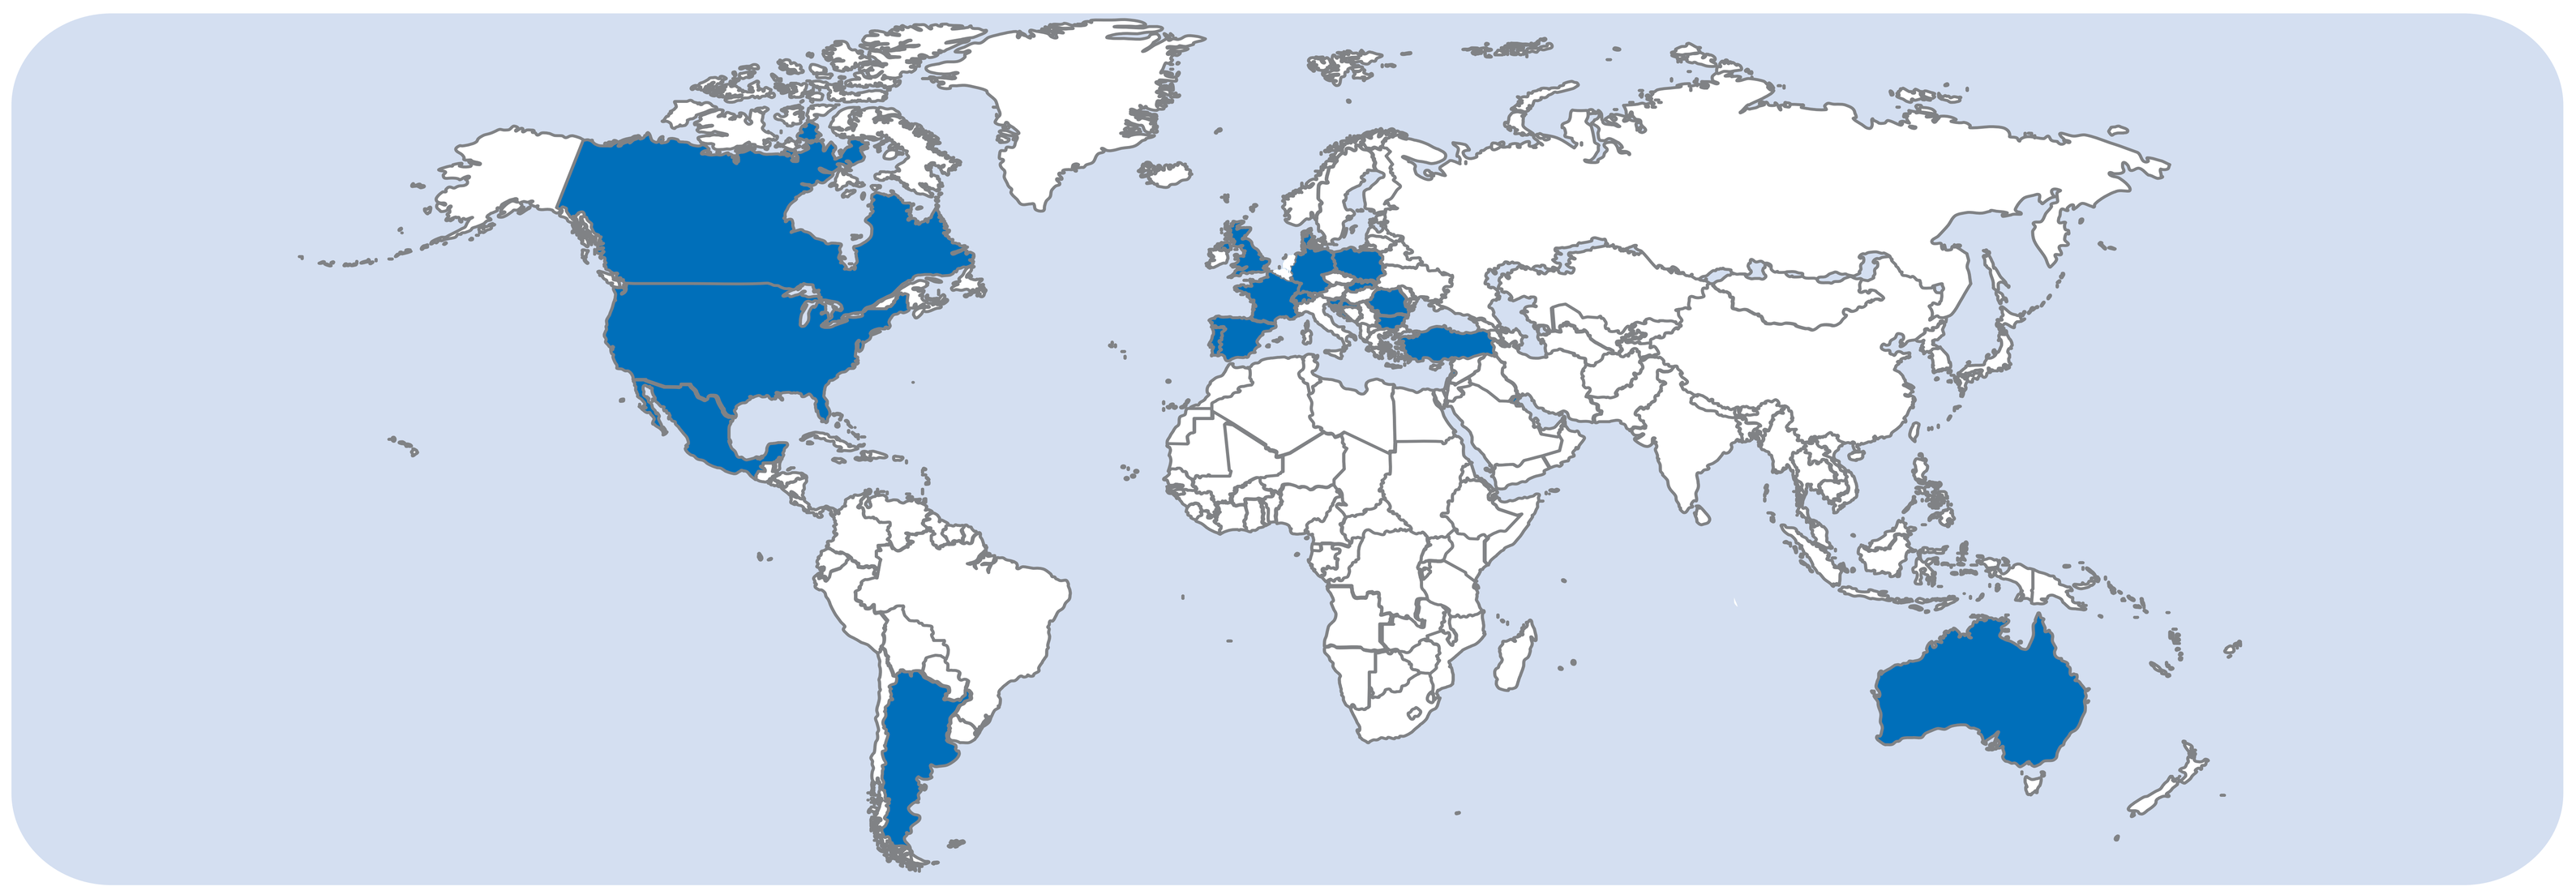

Study Treatments

- Patients in ENSEMBLE receive ocrelizumab 300 mg infusions on Day 1 and on Weeks 2, and ocrelizumab 600 mg infusions on week 24, 48, 72, 96, 120, 144 and 168. Patients already included in ENSEMBLE can be randomised directly to the ENSEMBLE PLUS conventional and short infusion protocols at any upcoming infusion.
- Patients entering ENSEMBLE PLUS who were already enrolled in ENSEMBLE will enter ENSEMBLE PLUS at any visit after the Week 24 visit and receive ocrelizumab per the schedule above
- The first dose will be administered as 2 × 300 mg infusions 14 days apart
- Ocrelizumab 600 mg infusions are administered as:
 - — Conventional infusion group: 500 mL infusions over 3.5 hours
 - — Short infusion group: 500 mL infusions over 2 hours with an additional saline 100 mL infusion administered over 1.5 hours to maintain blinded conditions
 - — The saline switch in the short infusion group will be mimicked in the conventional infusion group
 - — Infusions will be administered by unblinded infusion staff
- ENSEMBLE PLUS infusion schedule is presented in **Figure 4**

Figure 4. Infusion schedule

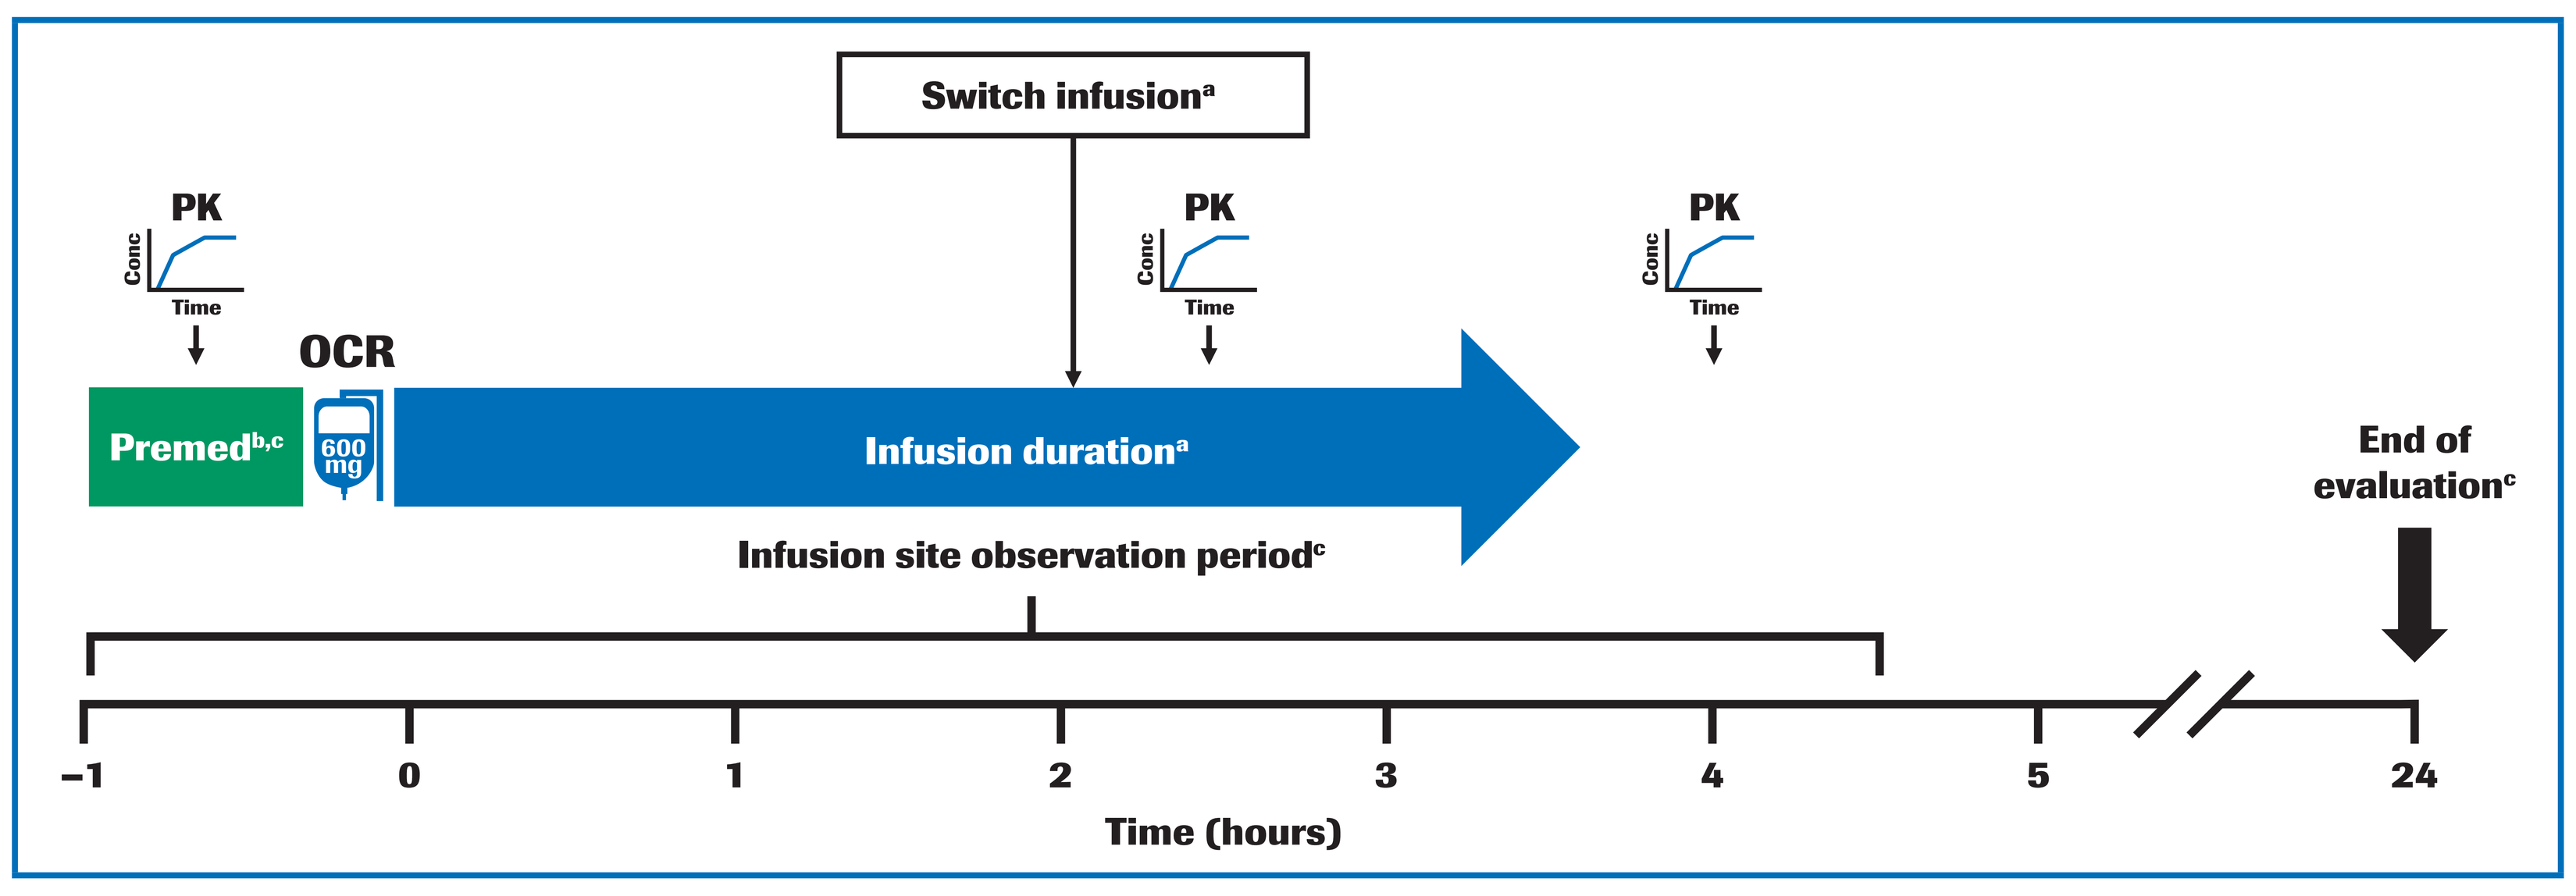
Switch infusion[a]
PK
Conc
Time
OCR
Premed[b,c]
600 mg
Infusion duration[a]
Infusion site observation period[c]
End of evaluation[c]
−1
0
1
2
3
4
5
24
Time (hours)

[a]Performed by an unblinded infusion nurse; [b]Predmedication with methylprednisolone 100 mg IV (or equivalent) approximately 30 minutes prior to each infusion and an antihistamine (e.g. diphehylhydramine) approximately 30–60 minutes prior to each infusion. Additional antipyretics (e.g. paracetamol) may also be considered; [c]Performed by a blinded clinical nurse (all clinical assessments and follow-up).
IV, intravenous; OCR, ocrelizumab; PK, pharmacokinetic assessment; Premed, premedications.

Study Assessments: IRRs

- The primary IRR assessment in ENSEMBLE PLUS is the proportion of patients with an IRR up to 24 hours after the first randomised infusion in both the conventional and short infusion group
- Secondary IRR assessments include:
 - — The proportion of patients with an IRR by dose and over the total study
 - — IRR severity by infusion
 - — Description of IRR

RESULTS

- The first patient enrolled in ENSEMBLE PLUS: October 2018

DISCUSSION

- ENSEMBLE PLUS will help to establish the safety profile of ocrelizumab when administered with a shorter infusion period

DISCLOSURES

B Brochet or his institution has received honoraria for consulting, speaking at scientific symposia and serving in advisory boards; has received research support from Actelion, Biogen Idec, Merck Serono, Sanofi Genzyme, Bayer, MedDay, F. Hoffmann-La Roche Ltd, Teva, Novartis, and institutional support from ANR, ARSEP and LFSEP (all with approval by the general director of CHU de Bordeaux). C Nos has received speaking honoraria from Novartis, and travel funding from Biogen Idec, Merck, F. Hoffmann-La Roche and Teva. MS Freedman has received research or educational grants from Genzyme Canada and honoraria or consultation fees from Actelion/Johnson & Johnson, BayerHealthcare, Biogen Idec, Chugai, Celgene Canada, Clene Nanomedicine, EMD Canada, Genzyme, Merck Serono, Novartis, F. Hoffmann-La Roche, Sanofi-Aventis and Teva Canada Innovation; has served as a member of company advisory boards, boards of directors or other similar groups for Actelion/Johnson & Johnson, BayerHealthcare, Biogen Idec, Clene Nanomedicine, Genzyme, F. Hoffman-La Roche, Merck Serono, MedDay, Novartis, Sanofi-Aventis; and has participated in a company-sponsored speakers bureau for Sanofi-Genzyme. J Killestein has accepted speaker and consulting fees from Merck, Biogen, Teva, Genzyme, Roche and Novartis. J Overell has received honoraria for speaking engagements and attendance at advisory boards, and conference travel support, from Teva, Novartis, Merck Serono, Genzyme, Roche, Allergan, WebMD Global and Biogen; his department has received educational funds, research support and funds to provide nursing and administrative staff from Novartis, Genzyme, Roche and Biogen. R Buffels is an employee of F. Hoffmann-La Roche Ltd. K Fitovski is an employee of F. Hoffmann-La Roche Ltd. L Mehta is an employee of Genentech, Inc./F. Hoffmann-La Roche Ltd. Q Wang is an employee of F. Hoffmann-La Roche Ltd. H-P Hartung has received honoraria for consulting, serving on steering committees and speaking at scientific symposia with approval by the Rector of Heinrich-Heine University Düsseldorf from Bayer, Biogen, F. Hoffmann-La Roche Ltd, GeNeuro SA, Genzyme, MedImmune, Merck Serono, Novartis, Octapharma, Opexa, Teva and Sanofi.

ACKNOWLEDGEMENTS

We would like to thank all patients, their families, and the investigators who participated in these trials. This research was funded by F. Hoffmann-La Roche Ltd, Basel, Switzerland. Writing and editorial assistance for this presentation was provided by Articulate Science, UK, and funded by F. Hoffmann-La Roche Ltd, Basel, Switzerland.

LITERATURE

1. Hauser SL, *et al. N Engl J Med* 2017;376:221–234.
2. Montalban X, *et al. N Engl J Med* 2017;376:209-220.
3. de Seze J, *et al. ECTRIMS* 2017;ePoster 1671.
4. Carroll W, *et al. PACTRIMS* 2018;Poster 76.
5. Polman CH, *et al. Ann Neurol* 2011;69:292–302.

Presented at the 26th Annual Meeting of the European Charcot Foundation (ECF); 15–17 November 2018; Baveno, Italy

Poster 68