Abstract No. 44

Subcutaneous Interferon Beta-1a, 10-Year Results from the United Kingdom Multiple Sclerosis Risk Sharing Scheme

G. Harty[1], S.L Wong[1], A. Gillett[2], A. Davies[3]

[1]EMD Serono Research & Development Institute Inc., Billerica, MA, USA; [2]Merck KGaA, Darmstadt, Germany; [3]Stockbridge Economic Appraisal Ltd., London, UK.

INTRODUCTION

- Subcutaneous interferon beta-1a (sc IFNβ-1a; Rebif®) was introduced to the European market in 1998. In 2001, the National Institute for Health and Care Excellence (NICE) concluded there was considerable uncertainty regarding the long-term clinical and economic benefits of disease-modifying treatments (DMTs) for relapsing-remitting multiple sclerosis (RRMS) and that DMTs may not represent a cost-effective use of National Health Service (NHS) resources.
- In response, the UK Department of Health (DoH) established the MS Risk Sharing Scheme (RSS), to provide patient access to DMTs, whilst monitoring patient outcomes in routine care, with provision that, if actual outcomes fell short of expectations, for price adjustments to maintain cost-effective provision of DMTs.[1]
- Interim, aggregate RSS results, for all DMTs have been published previously by the RSS Scientific Advisory Group.[1,2]
- Final RSS results for sc IFNβ-1a, which had an estimated cumulative 1,616,700 patient-years in the European post-marketing setting to 03 May 2018, have not previously been reported.
- In the European Union (EU), the recommended posology of Rebif® is 44 µg given three times per week by subcutaneous injection. A lower dose of 22 µg, also given three times per week by subcutaneous injection, is recommended for patients who cannot tolerate the higher dose in view of the treating specialist.
- In the RSS, both sc IFNβ-1a 22 µg & 44 µg dosages were assessed.

OBJECTIVE

- Our aim is to present the final year 10 results of the RSS for patients treated with sc IFNβ-1a.

METHODS

- The primary objective of the RSS was to monitor EDSS progression and compare this with expected outcomes in the absence of treatment.
- EDSS scores were monitored annually and analysed every 2 years. Monitoring continued over a 10-year follow up.
- To minimise 'drop-out bias' patients continued to be monitored following DMT discontinuation, or a switch to another DMT.

Patient Cohorts

- Patients were recruited from 72 specialist MS centres in UK NHS hospitals between May 2002 and April 2005. Adults with RRMS who met the Association of British Neurologist (ABN) criteria for DMT, aged ≥ 18 years, with two clinically significant relapses in the preceding two years and an Expanded Disability Status Scale (EDSS) score ≤ 5.5 were eligible for inclusion.
- A control cohort was developed from the British Columbia Multiple Sclerosis (BCMS) Database, which recorded EDSS scores in a group of MS patients in British Columbia, Canada.[3]

Analyses and Outcomes

- The relevant outcome statistic was the change relative to baseline in the distribution of EDSS scores weighted by the expected health related quality of life weights (utilities) of patients across EDSS levels.
- Utility values based on the EuroQoL EQ-5-dimension scale for integer EDSS scores, were derived from the MS survey.[1]
- An EDSS progression ratio was defined as the ratio of mean change in EDSS score from baseline to end of follow up compared with the expected change if the patient had been untreated.
- This expected change was based on the BCMS natural history cohort as outlined by Palace et al.[1]
- 'Target' hazard ratios (HRs) applied to the BCMS natural history cohort data allowed comparison of actual vs. target health related quality of life (utility) weighted EDSS.
 - HRs which would produce zero shortfall were deemed the 'implied HRs' (HRs < 1 indicate treatment benefit).[4]
- A shortfall in treatment benefit that exceeded 10% would trigger price adjustments to restore cost-effectiveness (**Figure 1**).
- For the full details of calculations, please refer to the UK DoH, 2002 Health Service Circular (HSC) relating to the scheme.[4]

Figure 1. The UK MS Risk Share Scheme: The basic premise

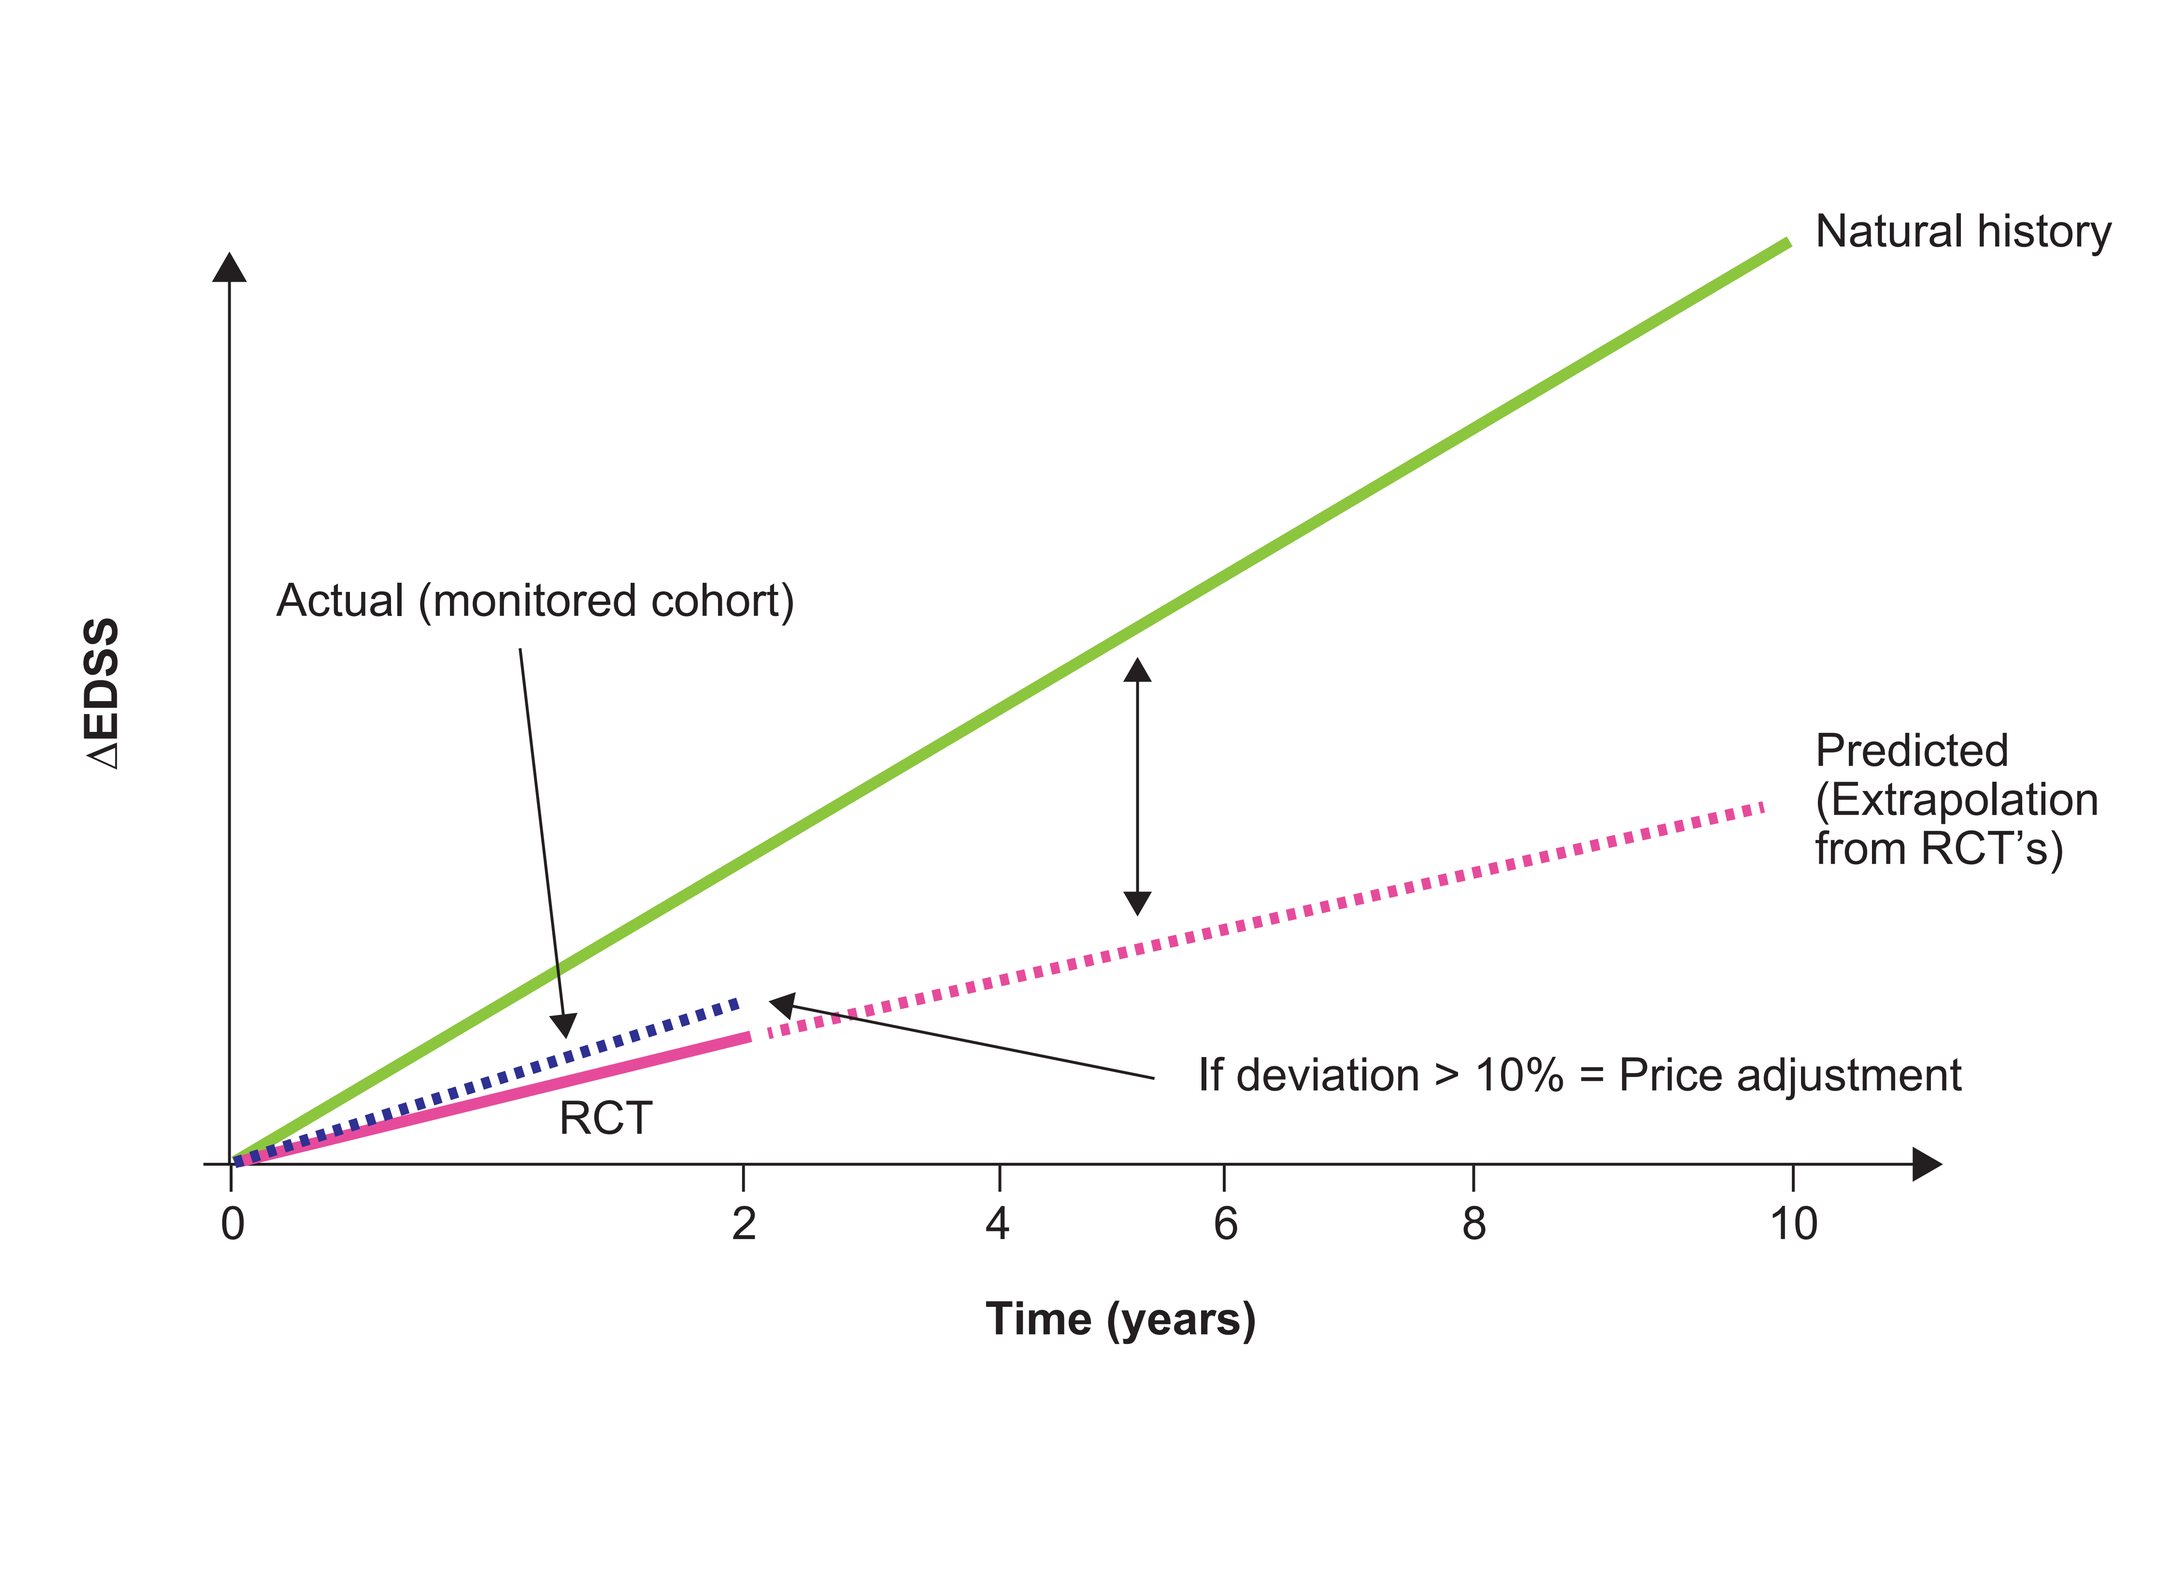
Natural history
Actual (monitored cohort)
ΔEDSS
Predicted (Extrapolation from RCT's)
If deviation > 10% = Price adjustment
RCT
0
2
4
6
8
10
Time (years)

EDSS, Expanded disability status scale; **MS**, multiple sclerosis; **RCT,** randomised controlled trial.

RESULTS

- A total of 4862 patients were included in the 10-year primary analysis cohort; 1635 patients were treated with sc IFNβ-1a (22 µg & 44 µg).
- At baseline, the mean EDSS score for sc IFNβ-1a patients with RRMS was 2.92.

Table 1. Baseline, Expected and Actual Disability Progression for all sc IFNβ-1a (22 µg & 44 µg) Patients (N = 1635) at Year 10

EDSS	Baseline	Expected without treatment†	Actual
0	0.0288	0.0355	0.0220
1	0.1609	0.1221	0.1156
2	0.2489	0.1403	0.1523
3	0.2202	0.1314	0.1162
4	0.1706	0.0768	0.1303
5	0.1052	0.0628	0.1101
6	0.0654	0.2152	0.2618
7		0.0892	0.0526
8		0.1029	0.0245
9		0.0118	0.0049
10		0.0121	0.0098

†BCMS natural history cohort.
EDSS, Expanded Disability Status Scale; **IFN**, interferon; **sc,** subcutaneous.

- In addition to the expected EDSS distributions with target levels of treatment effect, the implied HRs for sc IFNβ-1a (after 10 years) were 0.78 and 0.77 for patients treated with 22 µg or 44 µg doses, respectively.
- Baseline EDSS distributions for all patients treated with sc IFNβ-1a (22 µg & 44 µg) are shown in **Table 1** alongside the expected EDSS distributions of progression in the absence of treatment and the actual EDSS distribution of progression at the final year 10 follow up.
- These results were acceptable within the margin allowed under the scheme, thereby confirming the long-term clinical and cost-effectiveness of sc IFN-β-1a in UK clinical practice.
- The cumulative benefit of sc IFNβ-1a treatment was 1.6 EDSS years over the 10 year period compared to expected no treatment.
- Progression with sc IFNβ-1a was 6.49 EDSS years compared with 8.09 EDSS years in the 'natural history' cohort).
- A sensitivity analysis excluding patients initially diagnosed with secondary progressive MS did not impact the findings.

LIMITATIONS

- The RSS was an observational study. Patients were not randomised between treatments and in general it was up to patient and clinical discretion, to determine a DMT option.
- This possible selection bias, should be considered when interpreting results from the scheme.

CONCLUSIONS

- **The RSS scheme involves detailed monitoring of a cohort of patients to confirm the cost-effectiveness of these DMTs.**
- **Providing patients access to these products in a cost-effective manner was the key aim of the scheme.**
- **Sc IFNβ-1a succeeded in meeting the expectations required of the RSS and demonstrated its value in the treatment of MS patients in the UK.**
- **The findings of the 10-year analysis of this real-world-study confirm that treatment with sc IFNβ-1a in patients with MS has the effect of slowing disease progression.**
- **This long-term observation study supports the findings of earlier randomised controlled trials of the clinical benefits of sc IFNβ-1a.**

REFERENCES

1. Palace J, et al. *J Neurol Neurosurg Psychiatry.* 2018 [Epub ahead of print].
2. Duddy M, et al. *Pract Neurol*. 2016;16:4-6.
3. Tremlett H, Paty D, Devonshire V. *Neurol.* 2005;65:1919-1923.
4. http://webarchive.nationalarchives.gov.uk/20120503110731/http://www.dh.gov.uk/en/Publicationsandstatistics/Lettersandcirculars/Healthservicecirculars/DH_4004332 [Accessed September 2018]

ACKNOWLEDGEMENTS

The study was sponsored by the Health Departments of England, Wales, Scotland and Northern Ireland, Biogen Idec, Merck Serono (an affiliate of Merck KGaA), Bayer Schering Pharmaceuticals, Teva Pharmaceutical Industries, UK National Institute of Health Research's Health Technology Assessment Programme. The authors would like to thank patients, investigators, co-investigators and the study teams at each of the participating centres. Medical writing assistance was provided by Ewan Smith, Keith Evans and Claire Mwape of inScience Communications, Springer Healthcare, Chester, UK and funded by Merck KGaA, Darmstadt, Germany.

DISCLOSURES

GH and **SLW** are employees of EMD Serono Research & Development Institute Inc., a division of Merck KGaA, Darmstadt, Germany. **AG** is an employee of Merck KGaA, Darmstadt, Germany. **AD** provided consulting services to Merck through Stockbridge Economic Appraisal Ltd.

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors.

GET POSTER PDF

Presented at European Charcot Foundation (ECF) 2018; 15-17 November; Baveno, Italy.