Proportions of Patients With Highly Active RMS Achieving No Evidence of Disease Activity in Response to Cladribine Tablets in CLARITY

G. Giovannoni[1], K. Rammohan[2], S. Cook[3], G. Comi[4], P. Rieckmann[5], P. Soelberg-Sorensen[6], P. Vermersch[7], F. Dangond[8], C. Hicking[9]

[1]Queen Mary University of London, Blizard Institute, Barts and The London School of Medicine and Dentistry, London, UK; [2]Department of Neurology, MS Research Center University of Miami School of Medicine, Miami, FL, USA; [3]Rutgers, The State University of New Jersey, New Jersey Medical School, Newark, NJ, USA; [4]Department of Neurology and Institute of Experimental Neurology, Università Vita-Salute San Raffaele, Ospedale San Raffaele, Milan, Italy; [5]Department of Neurology, Medical Park Loipl and University of Erlangen, Erlangen, Germany; [6]Danish MS Center, Department of Neurology, University of Copenhagen, Rigshospitalet, Copenhagen, Denmark; [7]Université de Lille, CHU Lille, LIRIC-INSERM U995, FHU Imminent, Lille, France; [8]EMD Serono, Inc., Billerica, MA, USA; [9]Merck KGaA, Darmstadt, Germany

INTRODUCTION

- CLARITY, a large study (1326 patients, **Figure 1**) in patients with early and moderately advanced active relapsing multiple sclerosis (RMS), demonstrated that cladribine tablets 3.5 mg/kg, given in short courses annually over 2 years, significantly reduced relapse rates, disability worsening, and outcomes assessed by magnetic resonance imaging (MRI) versus placebo.[1]
- Patients with RMS who show an increased rate of relapse or disability progression can be described as having high disease activity (HDA). Analysis of CLARITY may provide insights into the efficacy of cladribine tablets in patients with evidence of HDA.
- The high efficacy of new treatments like cladribine tablets may warrant modification of treatment aims to go beyond reduction of relapse rates and disability progression. No evidence of disease activity (NEDA) is a composite of measures that includes no relapses, no disability worsening, and no MRI activity. Previous *post hoc* analyses have analyzed individual and composite NEDA measures in CLARITY using alternative definitions of HDA at baseline.[2]

Figure 1. CLARITY Study Design

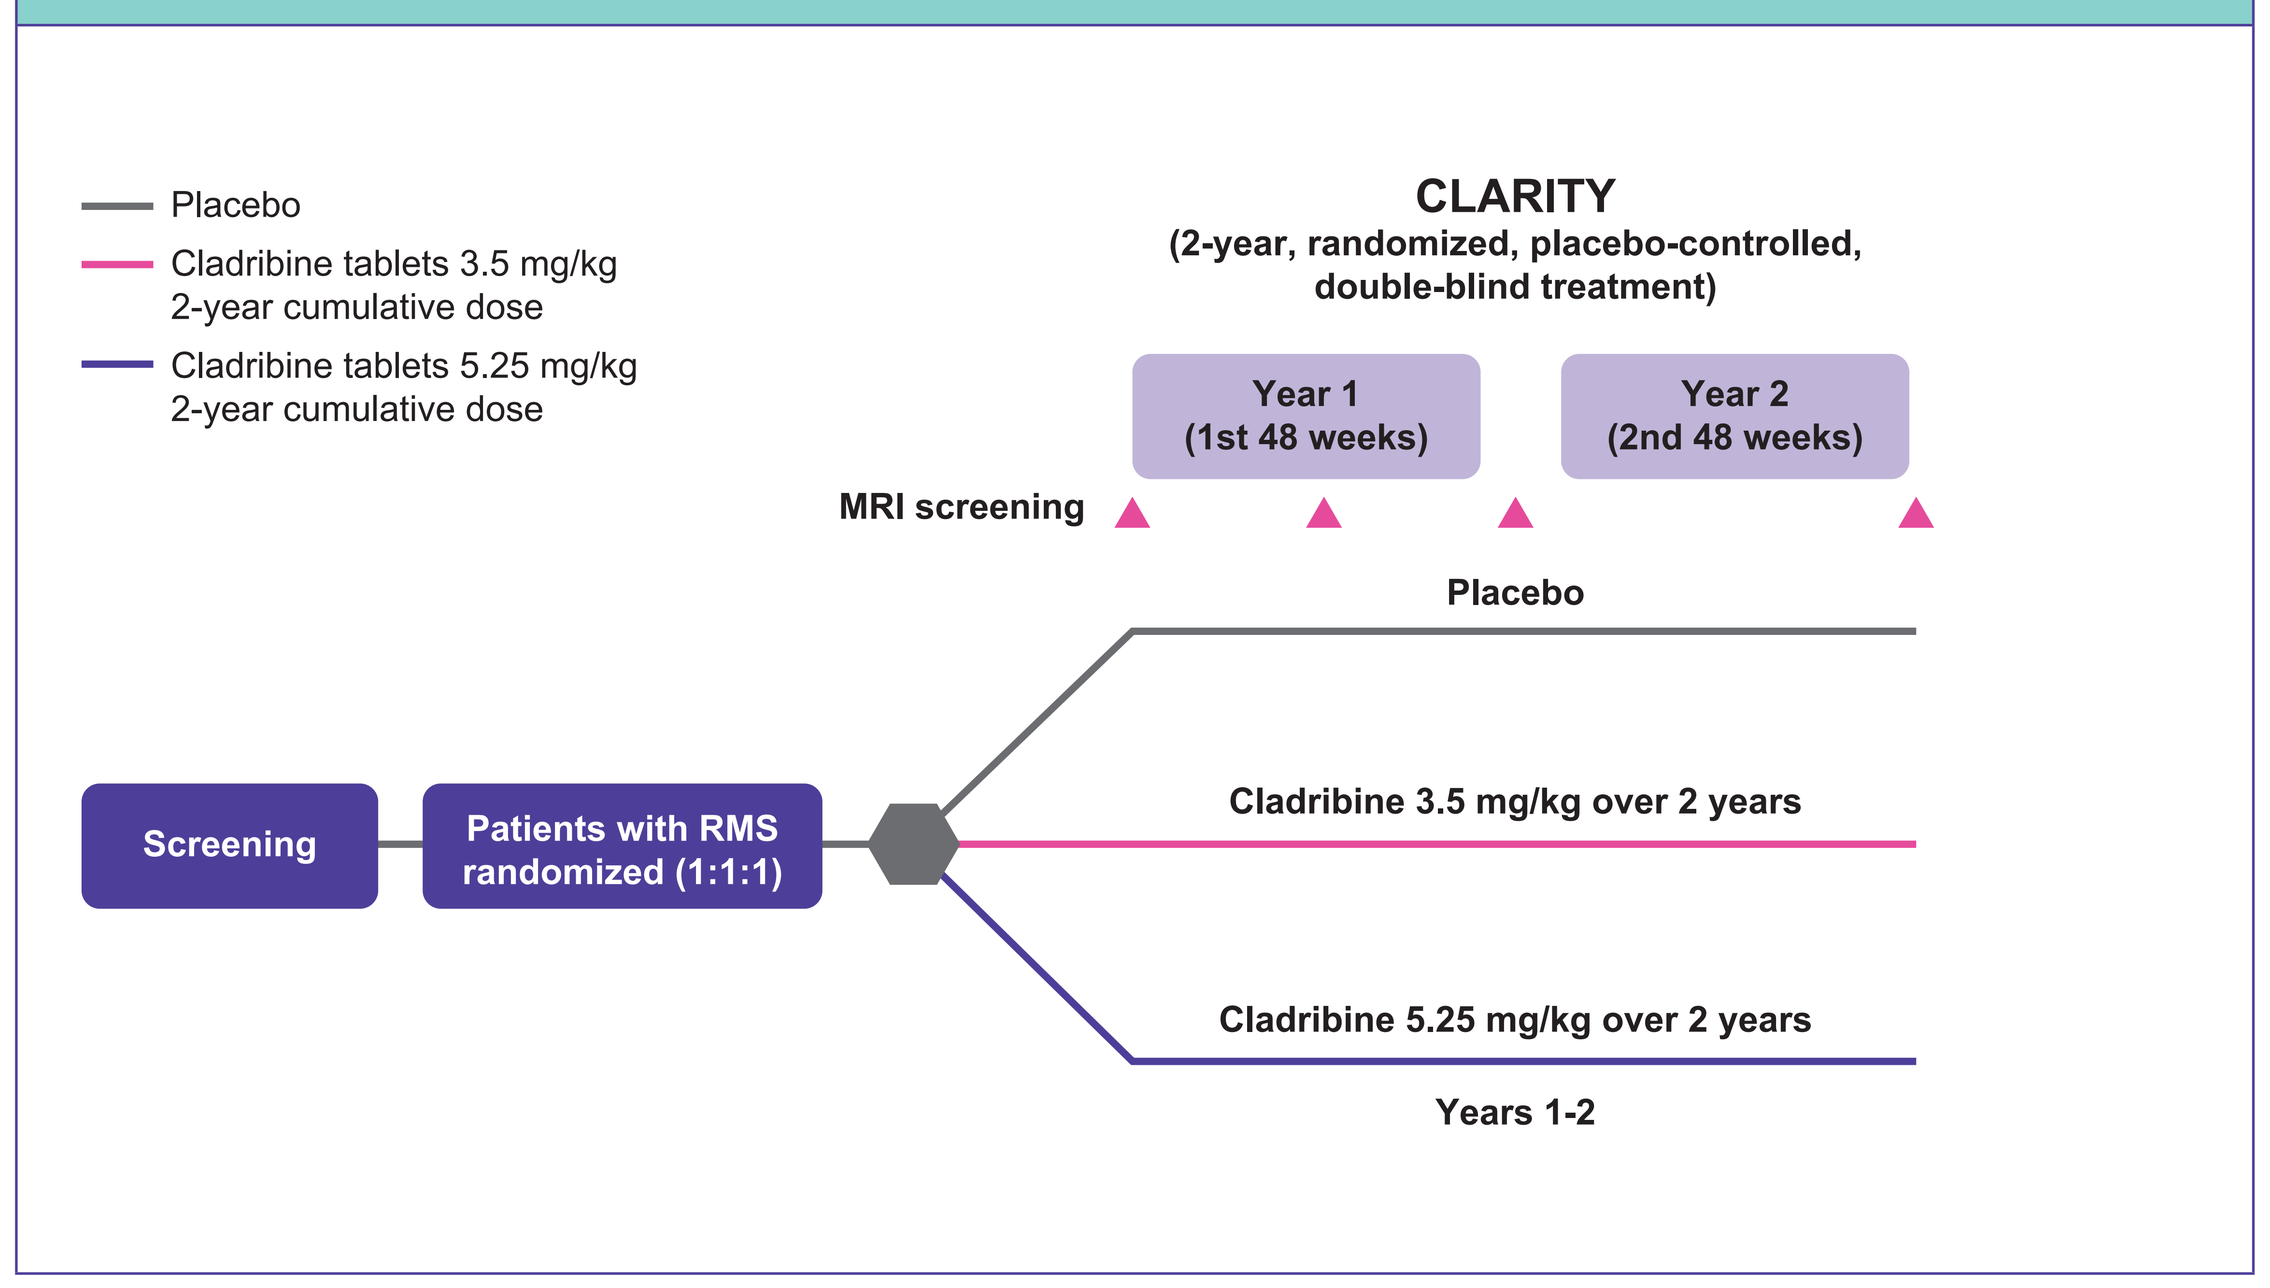
Placebo
Cladribine tablets 3.5 mg/kg 2-year cumulative dose
Cladribine tablets 5.25 mg/kg 2-year cumulative dose
CLARITY
(2-year, randomized, placebo-controlled, double-blind treatment)
Year 1 (1st 48 weeks)
Year 2 (2nd 48 weeks)
MRI screening
Placebo
Screening
Patients with RMS randomized (1:1:1)
Cladribine 3.5 mg/kg over 2 years
Cladribine 5.25 mg/kg over 2 years
Years 1-2

MRI, magnetic resonance imaging; **RMS**, relapsing multiple sclerosis.

OBJECTIVE

- The aim of this *post hoc* analysis was to compare the effects of cladribine tablets 3.5 mg/kg versus placebo on the proportion of patients with NEDA in subgroups of CLARITY patients at higher risk of disease progression than the intent-to-treat (ITT) population, selected using two HDA definitions.

METHODS

- The CLARITY study enrolled patients aged 18–65 years with a definite diagnosis of relapsing-remitting multiple sclerosis (RRMS):[3]
 - ≥ 1 relapse in the 12 months before study entry, but no relapses within the 28 days before entry.
 - Neurological lesions detectable by MRI consistent with MS.
 - An Expanded Disability Status Scale (EDSS) score of 0−≤ 5.5.
- Patients were excluded if they had received a disease-modifying drug (DMD) within 3 months before study entry, or if treatment with > 1 DMD had failed.
- In CLARITY, MRI scans were carried out at the pre-trial assessment and at Weeks 24, 48, and 96 (or early termination).

HDA Subgroups

- CLARITY patients randomized to cladribine tablets 3.5 mg/kg (N = 433) or placebo (N = 437) were retrospectively analyzed using two different HDA definitions based on relapse history, prior treatment, and MRI characteristics.
- Two overlapping sets of criteria (**Figure 2**) were applied in the analysis of baseline disease characteristics to subdivide patients into HDA groups based upon:
 1. High relapse activity (HRA); patients with ≥ 2 relapses in the year before study entry, regardless of prior use of DMD.
 2. HRA plus treatment nonresponse (HRA+TNR); patients with ≥ 2 relapses in the year before study entry, regardless of prior use of DMD, plus patients with ≥ 1 relapse AND ≥ 1 T1 gadolinium-enhancing (Gd+) or ≥ 9 T2 lesions in the year before study entry while on DMD therapy.
- Data from the 2-year, double-blind periods of CLARITY were used to compare the efficacy of cladribine tablets versus placebo in patients with RMS in the overall population and in subgroups defined by these baseline characteristics.
- The ability of these criteria to retrospectively identify patients with HDA and the effects of cladribine tablets 3.5 mg/kg versus placebo on relapse rates and disability outcomes in these patients has been presented previously.[4,5]

Figure 2. Definitions of High Disease Activity

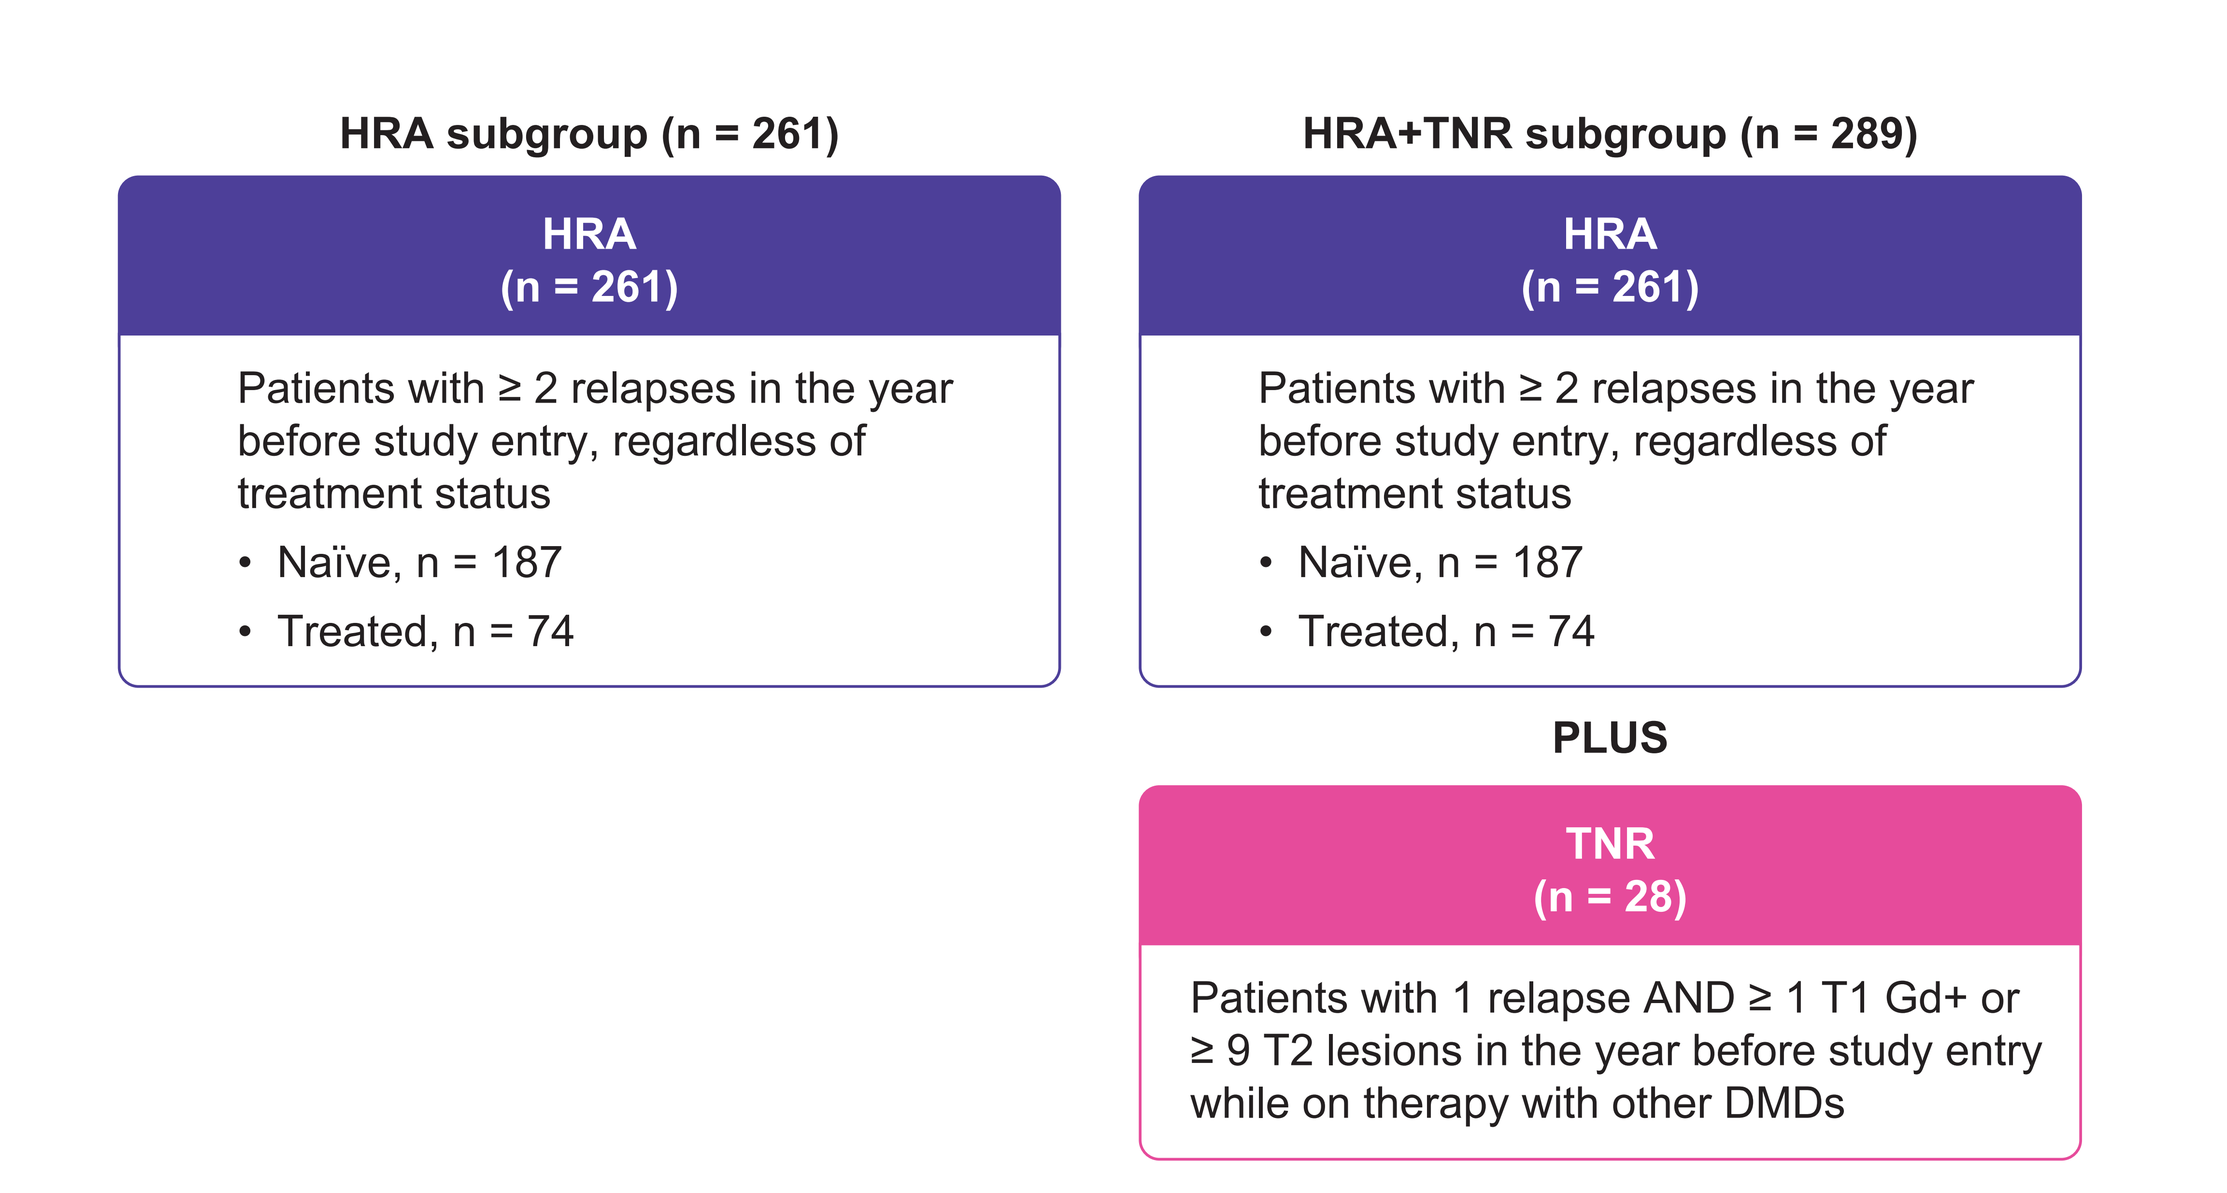
HRA subgroup (n = 261)
HRA
(n = 261)
Patients with ≥ 2 relapses in the year before study entry, regardless of treatment status
• Naïve, n = 187
• Treated, n = 74
HRA+TNR subgroup (n = 289)
HRA
(n = 261)
Patients with ≥ 2 relapses in the year before study entry, regardless of treatment status
• Naïve, n = 187
• Treated, n = 74
PLUS
TNR
(n = 28)
Patients with 1 relapse AND ≥ 1 T1 Gd+ or ≥ 9 T2 lesions in the year before study entry while on therapy with other DMDs

NB These definitions are not exclusive; to a large extent, the subgroups comprise the same patients.
DMD, disease modifying drug; **Gd+**, gadolinium-enhancing; **HRA**, high relapse activity; **HRA+TNR**, high relapse activity plus treatment nonresponse.

- All HDA analyses were *post hoc* and not pre-specified; no multiplicity adjustments were done to the resulting *P* values. Comparisons with a *P* value < 0.05 by statistical testing should be regarded as nominally significant.

NEDA

- NEDA was defined as qualifying relapse-free, 3-month confirmed EDSS progression-free, T1 Gd+ lesion-free, and active T2 lesion-free.
- The composite analysis looked at patients who achieved all components of NEDA over the full time period in CLARITY, and received treatment with cladribine tablets 3.5 mg/kg or placebo. Patients who were missing data for one or more components of NEDA were reported as unknown. Patients who withdrew early(< 83 weeks) with no disease activity were reported as unknown.
- Odds ratios (ORs) and 95% confidence intervals (95% CI) were calculated for NEDA in patients treated with cladribine tablets 3.5 mg/kg or placebo, in the ITT population and in patient subgroups, from a logistic regression with treatment as fixed effect and study as covariate.

RESULTS

- The current definitions of HRA and HRA+TNR were validated through examination of their ability to identify placebo-treated patients at increased risk of relapses or EDSS progression and show consistency with the previous observations using alternative definitions of HDA at baseline.[2]

Patients

- Demographics and disease characteristics were similar across HDA subgroups (**Table 1**). The characteristics of patients who did not fulfill the HDA criteria were also similar across the subgroups (data not shown).
- The overall analysis involved 870 patients randomized to placebo (n = 437) or cladribine tablets 3.5 mg/kg (n = 433):
 - Among the non-HRA patients, 306 received placebo and 303 received cladribine tablets 3.5 mg/kg.
 - Among the non-HRA+TNR patients, 288 received placebo and 293 received cladribine tablets 3.5 mg/kg.

Table 1. Patient Demographics and Disease Characteristics by HDA Subgroup

	Placebo		Cladribine Tablets 3.5 mg/kg	
	HRA (n = 131)	HRA+TNR (n = 149)	HRA (n = 130)	HRA+TNR (n = 140)
Age, years; mean (SD)	36.7 (10.3)	37.1 (10.2)	36.5 (9.5)	36.3 (9.5)
Female, n (%)	82 (62.6)	94 (63.1)	96 (73.8)	102 (72.9)
Disease duration, years; mean (SD)	4.51 (5.48)	4.75 (5.34)	3.90 (5.07)	3.94 (4.92)
Prior use of DMDs, n (%)	38 (29.0)	56 (37.6)	36 (27.7)	46 (32.9)
Relapses in prior 12 months, n (%)				
0	0 (0.0)	0 (0.0)	0 (0.0)	0 (0.0)
1	0 (0.0)	18 (12.1)	0 (0.0)	10 (7.1)
2	110 (84.0)	110 (73.8)	105 (80.8)	105 (75.0)
≥ 3	21 (16.0)	21 (14.1)	25 (19.2)	25 (17.9)
EDSS score; mean (SD)	2.91 (1.37)	2.97 (1.35)	2.90 (1.33)	2.86 (1.32)
Number of T1 Gd+ lesions; mean (IQR)	1.1 (0.0;1.0)	1.0 (0.0;1.0)	1.4 (0.0;1.0)	1.3 (0.0;1.0)
Number of T2 lesions; mean (IQR)	30.0 (15.0;39.0)	29.9 (16.0;39.0)	25.3 (12.0;33.0)	25.2 (12.0;33.5)

DMD, disease modifying drug; **EDSS**, Expanded Disability Status Scale; **Gd+**, gadolinium-enhancing; **HDA**, high disease activity; **HRA**, high relapse activity; **HRA+TNR**, high relapse activity plus treatment nonresponse; **IQR**, interquartile range; **SD**, standard deviation.

Patients Who Achieved NEDA and Each Component

Figure 3. Effects of Placebo and Cladribine Tablets 3.5 mg/kg on the Proportions of Patients Who Fulfilled the Individual Components of NEDA, Excluding Unknowns

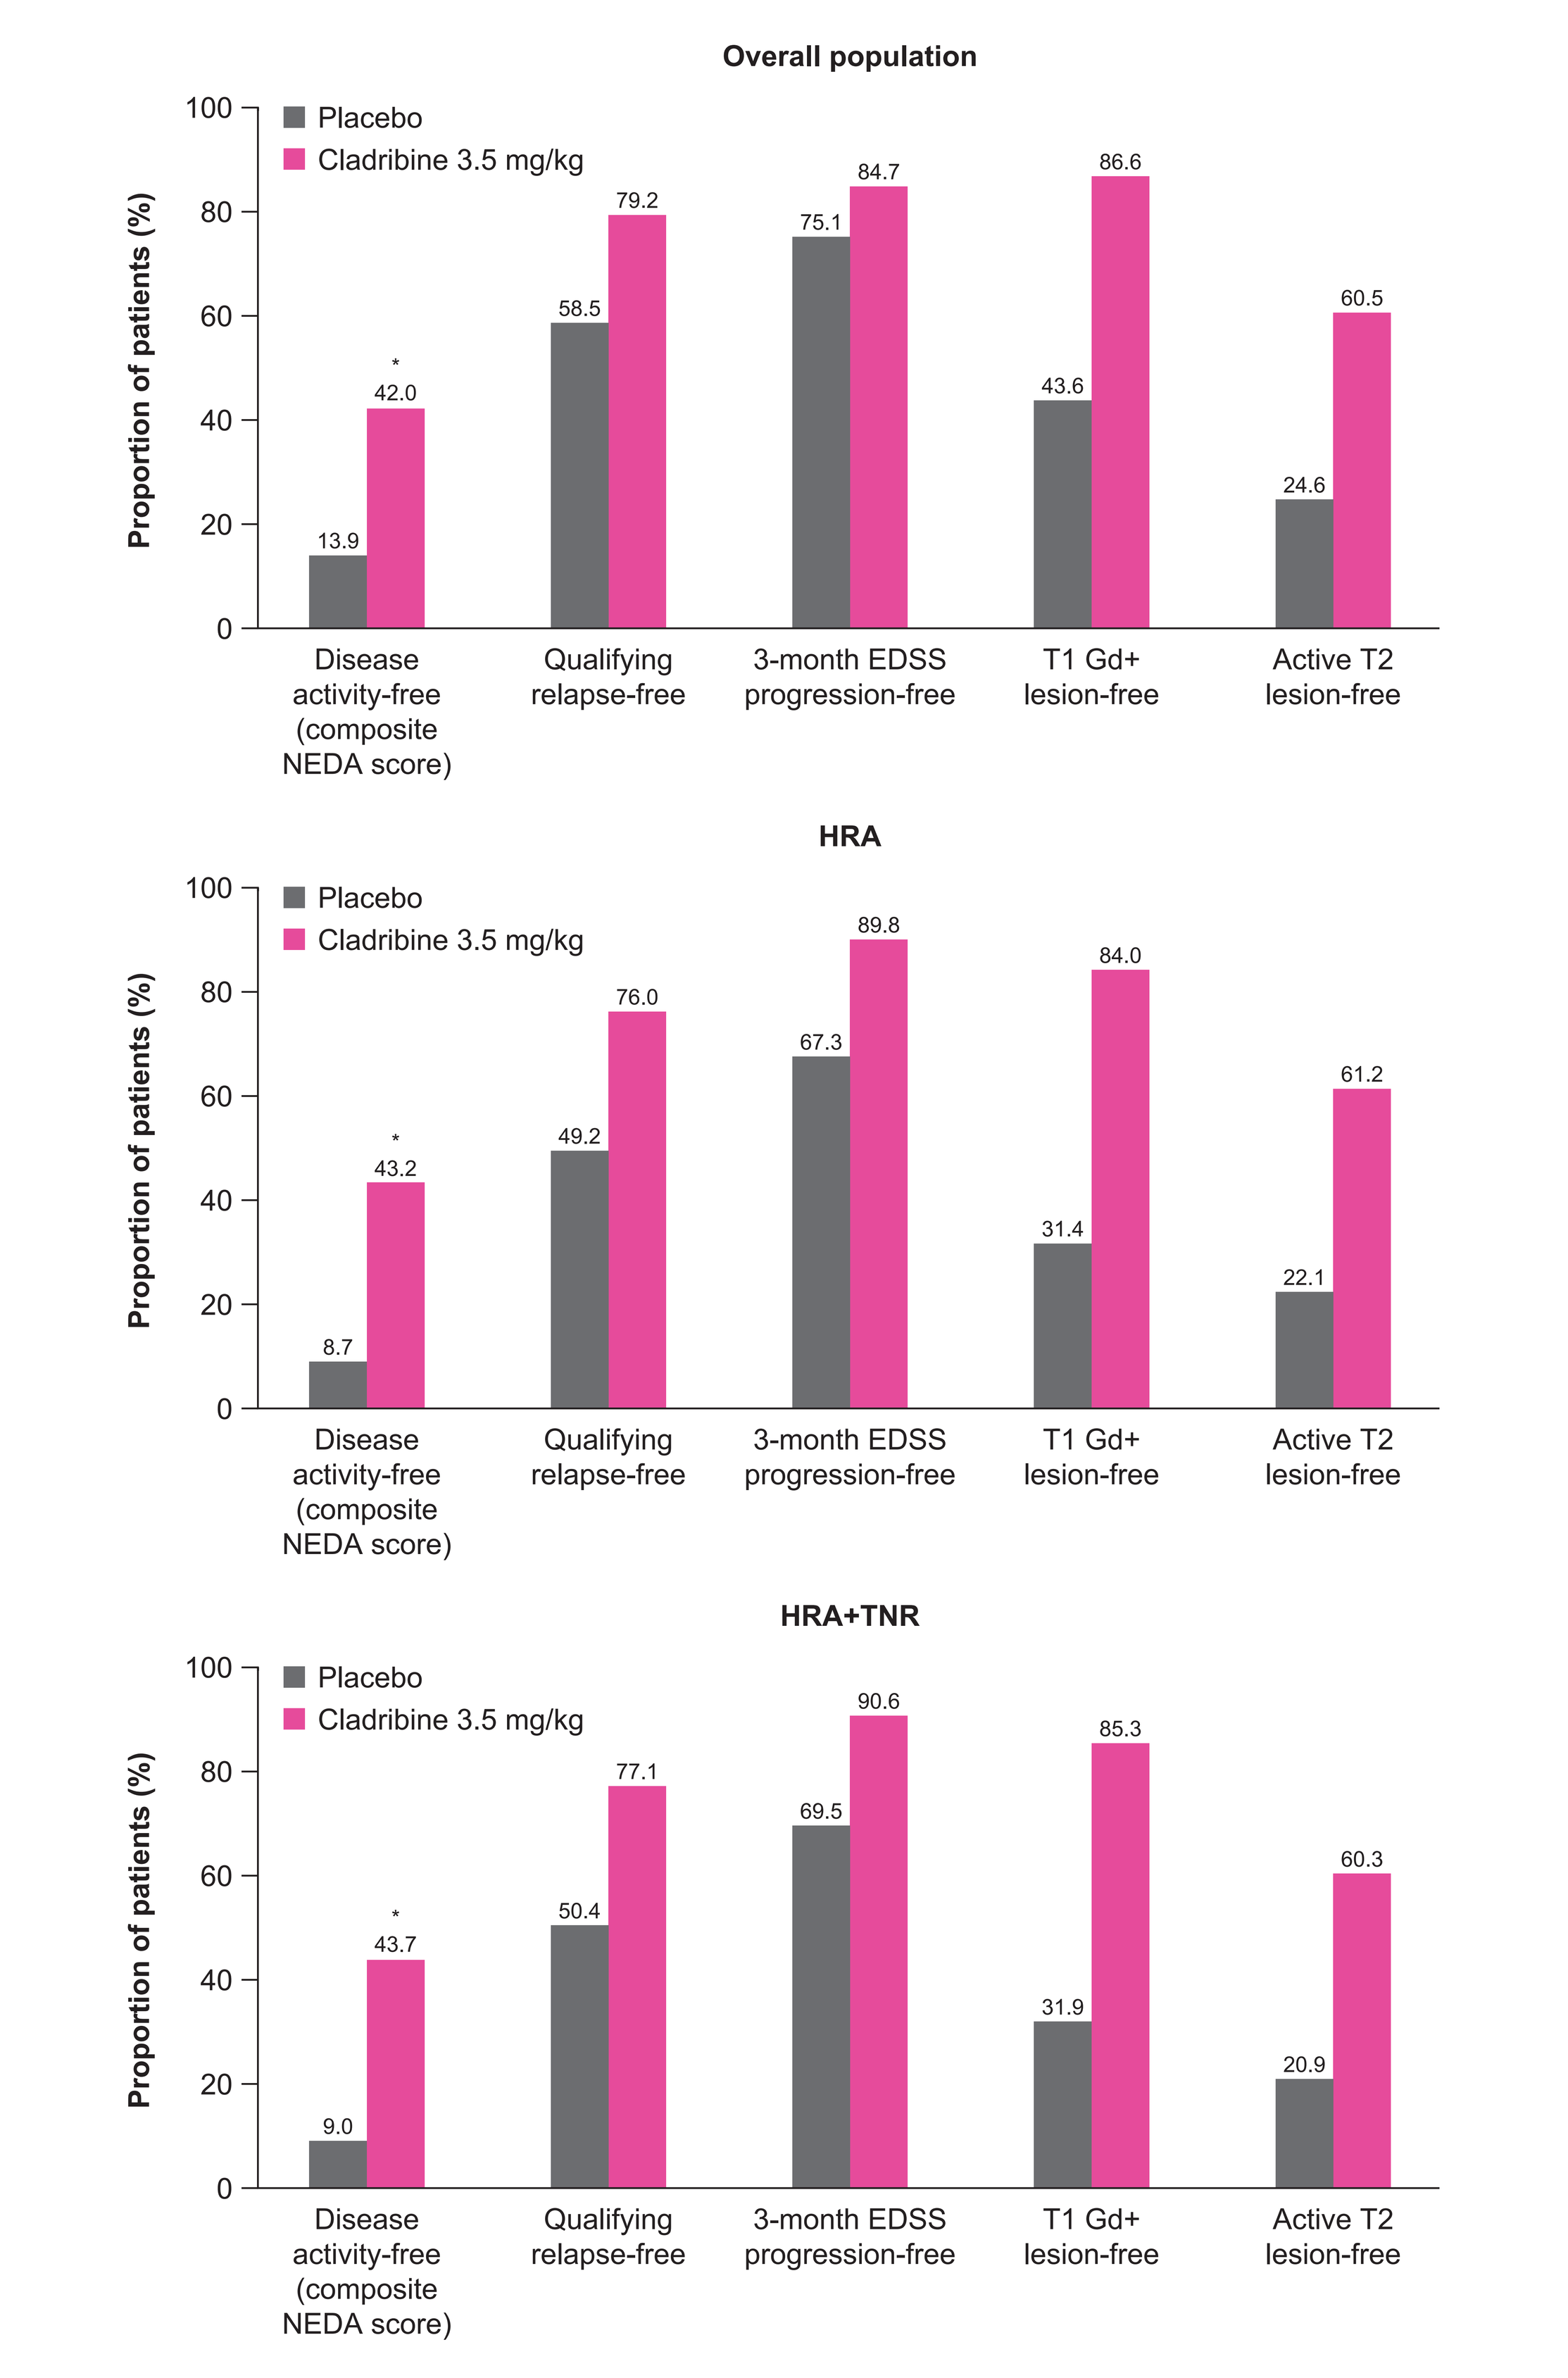
Overall population
Placebo
Cladribine 3.5 mg/kg
Proportion of patients (%)
100
80
60
40
20
0
13.9
42.0
*
58.5
79.2
75.1
84.7
43.6
86.6
24.6
60.5
Disease activity-free (composite NEDA score)
Qualifying relapse-free
3-month EDSS progression-free
T1 Gd+ lesion-free
Active T2 lesion-free
HRA
8.7
43.2
*
49.2
76.0
67.3
89.8
31.4
84.0
22.1
61.2
HRA+TNR
9.0
43.7
*
50.4
77.1
69.5
90.6
31.9
85.3
20.9
60.3

* *P* <0.0001 vs placebo.
EDSS, expanded Disability Status Scale; **Gd+**, gadolinium-enhancing; **HRA**, high relapse activity; **HRA+TNR**, high relapse activity plus treatment nonresponse; **NEDA**, no evidence of disease activity.

- In the overall CLARITY population, and in each HDA subgroup, the proportion of patients who fulfilled each component of NEDA (patients qualifying relapse-free, 3-month confirmed EDSS progression-free, T1 Gd+ lesion-free, and active T2 lesion-free) over 2 years was higher with cladribine tablets 3.5 mg/kg than placebo (**Figure 3**).
- In general, the effects of treatment with cladribine tablets 3.5 mg/kg versus placebo were similar for each HDA subgroup and the overall CLARITY population.

Odds of Achieving NEDA

- Odds ratios for achieving NEDA status in the overall population and the two HDA subgroups are shown in **Figure 4**.
 - These values were significantly more favorable than the non-HDA counterpart subgroup for the HRA+TNR definition (interaction *P* value = 0.0435).

Figure 4. Effects of Cladribine Tablets 3.5 mg/kg Versus Placebo on the Odds Ratio for Composite NEDA Score in the Overall Patient Population and in Each HDA Subgroup

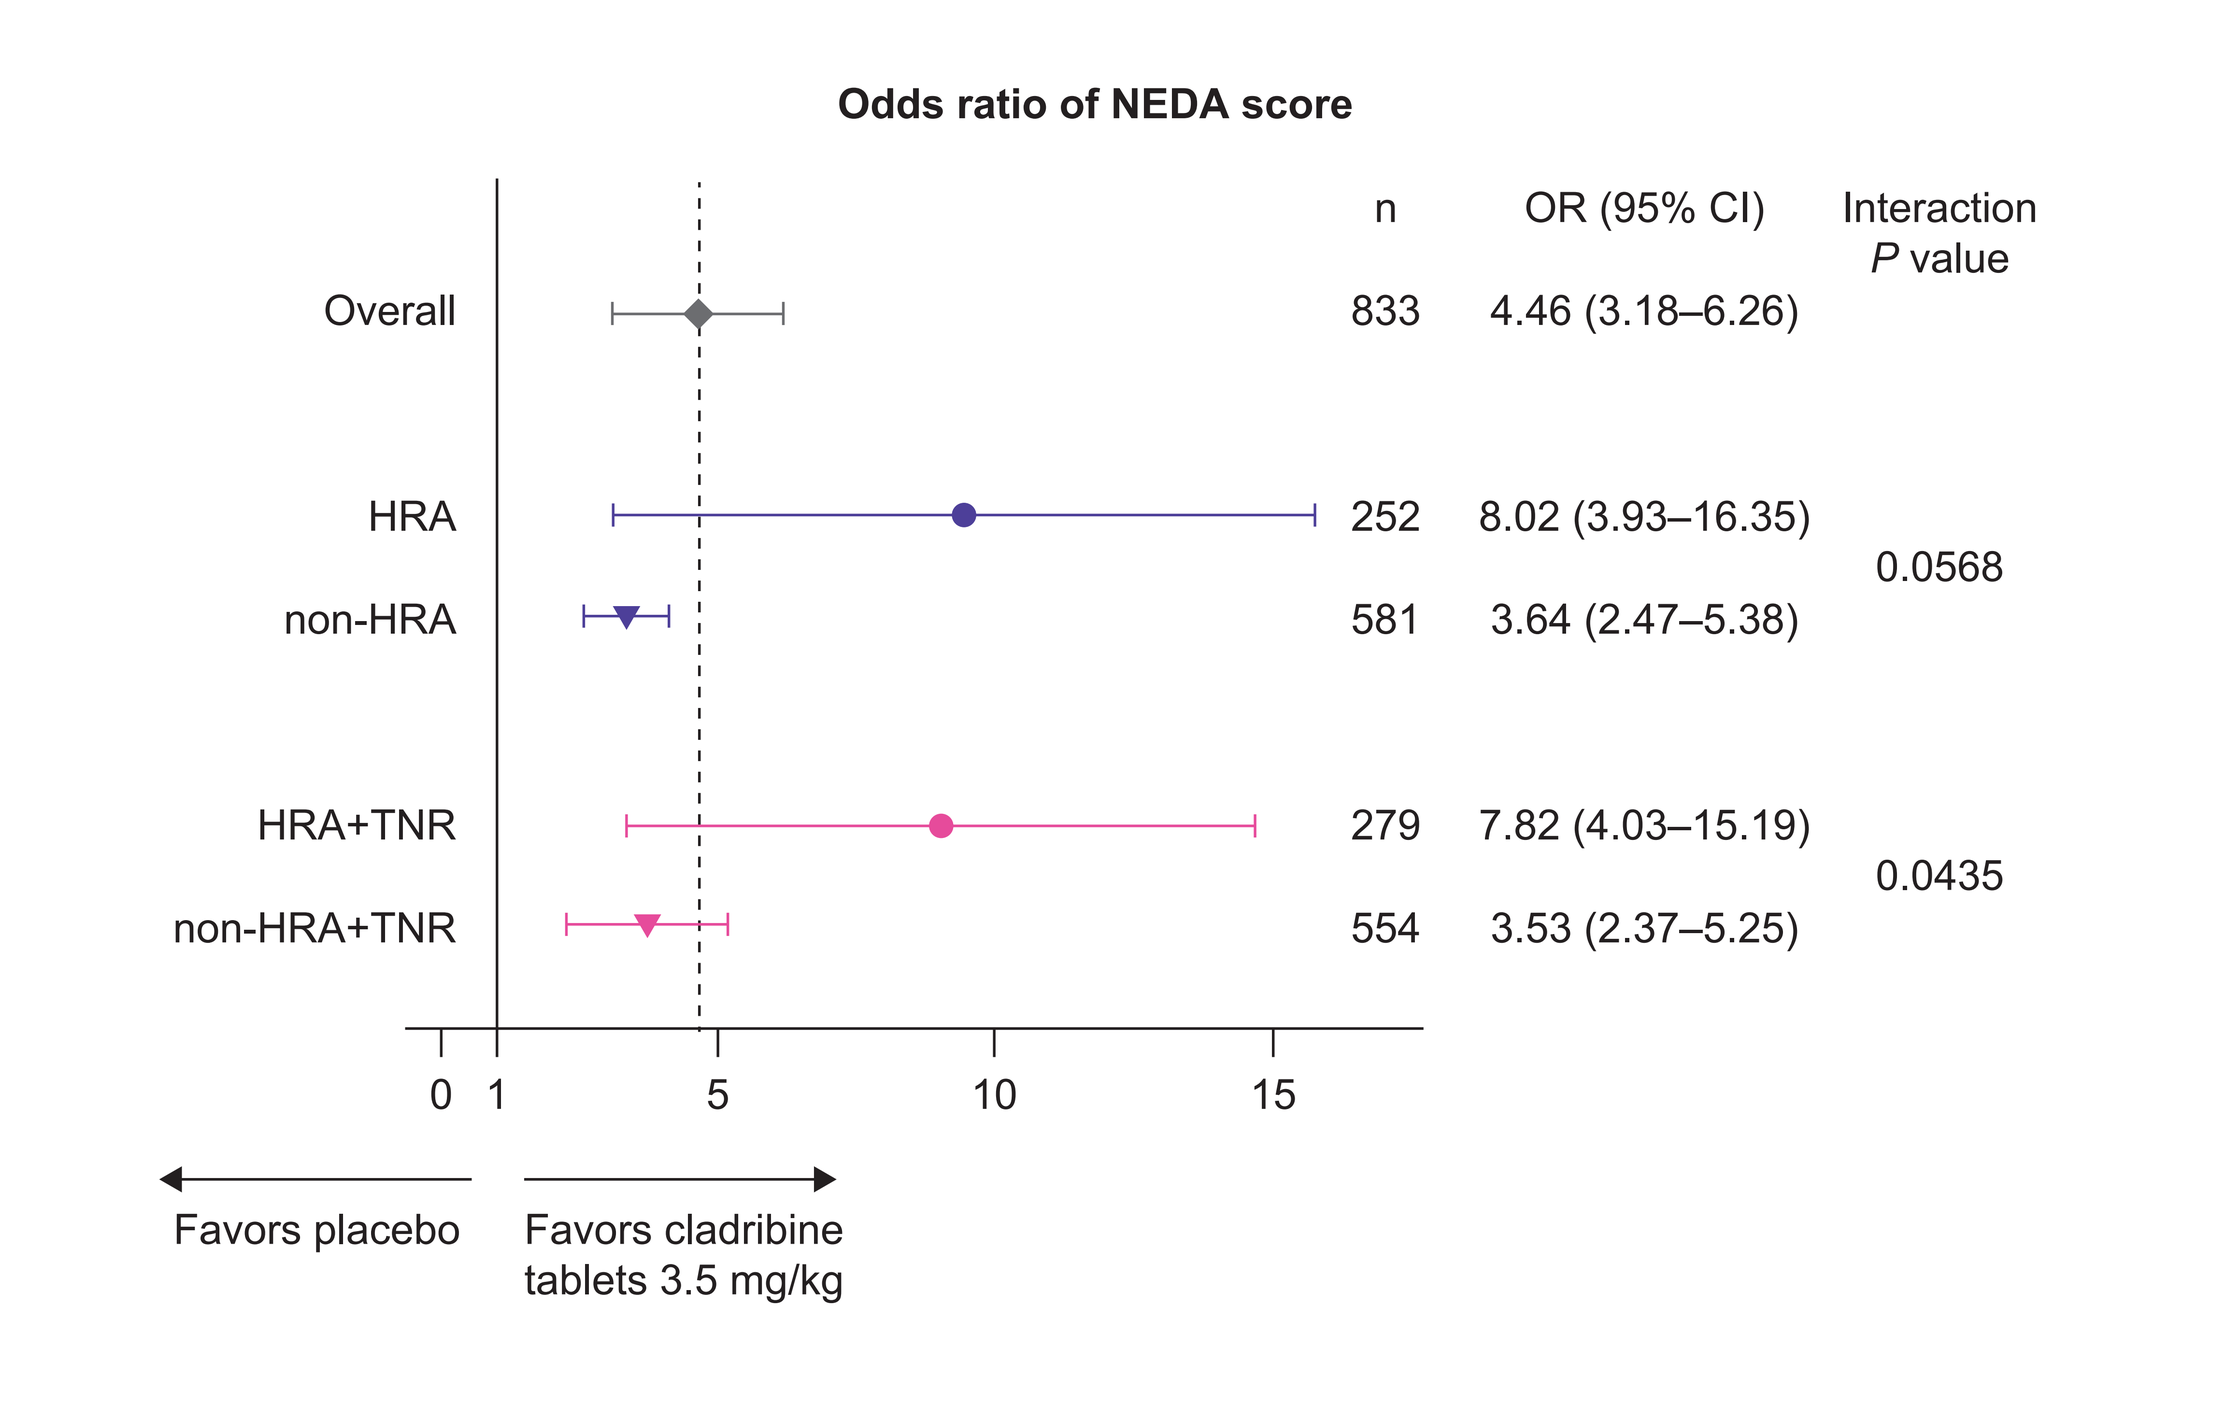
Odds ratio of NEDA score
n
OR (95% CI)
Interaction P value
Overall
833
4.46 (3.18–6.26)
HRA
252
8.02 (3.93–16.35)
non-HRA
581
3.64 (2.47–5.38)
0.0568
HRA+TNR
279
7.82 (4.03–15.19)
non-HRA+TNR
554
3.53 (2.37–5.25)
0.0435
0
1
5
10
15
Favors placebo
Favors cladribine tablets 3.5 mg/kg

CI, confidence interval; **HDA**, high disease activity; **HRA**, high relapse activity; **HRA+TNR**, high relapse activity plus treatment nonresponse; **NEDA**, no evidence of disease activity; **OR**, odds ratio.

CONCLUSIONS

- **In a large study of patients with active RMS, treatment with cladribine tablets significantly increased the proportion of HDA patients with no evidence of disease activity compared with placebo.**
- **Compared with the overall study population, each HDA subgroup showed higher odds of achieving NEDA with cladribine tablets 3.5 mg/kg versus placebo.**

REFERENCES

1. Giovannoni G, et al. *N Engl J Med.* 2010;362:416-426.
2. Giovannoni G, et al. *Lancet Neurol.* 2011;10:329-337.
3. McDonald W, et al. *Ann Neurol.* 2001;50:121-127.
4. Giovannoni G, et al. *Neurology.* 2017;88(16 suppl):P6.351.
5. Giovannoni G, et al. *Neurology.* 2017;88(16 suppl):P6.360.

ACKNOWLEDGMENTS

This study was sponsored by EMD Serono, Inc., a business of Merck KGaA, Darmstadt, Germany (in the USA), and Merck Serono SA – Geneva, an affiliate of Merck KGaA, Darmstadt, Germany (ROW). The authors would like to thank patients and their families, investigators, co-investigators, and the study teams at each of the participating centers and at Merck KGaA, Darmstadt, Germany. Medical writing assistance was provided by Jackie Campbell of inScience Communications, Springer Healthcare, Chester, UK, and was funded by Merck KGaA, Darmstadt, Germany.

DISCLOSURES

GG has received speaker honoraria and consulting fees from Abbvie, Atara Bio, Almirall, Bayer Schering Pharma, Biogen Idec FivePrime, GlaxoSmithKline, GW Pharma, Merck, Pfizer Inc, Protein Discovery Laboratories, Teva Pharmaceutical Industries Ltd, Sanofi-Genzyme, UCB, Vertex Pharmaceuticals, Ironwood, and Novartis; and has received research support unrelated to this study from Biogen Idec, Merck, Novartis, and Ironwood. **KR** has received honoraria for lectures and steering committee meetings from EMD Serono, Biogen Idec, Sanofi-Aventis, Genzyme, Novartis, Teva Neurosciences, Acorda, and Roche/Genentech. **SC** has received honoraria for lectures/consultations from Merck, Bayer HealthCare, Sanofi-Aventis, Neurology Reviews, Biogen Idec, Teva Pharmaceuticals, and Actinobac Biomed Inc.; has served on advisory boards for Bayer HealthCare, Merck, Actinobac Biomed, Teva Pharmaceuticals, and Biogen Idec; and received grant support from Bayer HealthCare. **GC** has received consulting fees from Novartis, Teva Pharmaceutical Industries Ltd., Sanofi-Aventis, Merck, Receptos, Biogen Idec, Genentech-Roche, and Bayer Schering; lecture fees from Novartis, Teva Pharmaceutical Ind. Ltd., Sanofi-Aventis, Merck, Biogen Dompè, Bayer Schering, and Serono Symposia International Foundation; and trial grant support from Novartis, Teva Pharmaceutical Ind. Ltd., Sanofi-Aventis, Receptos, Biogen Idec, Genentech-Roche, Merck, Biogen Dompè, and Bayer Schering. **PR** has received honoraria for lectures/steering committee meetings from Merck, Biogen Idec, Bayer Schering Pharma, Boehringer-Ingelheim, Sanofi-Aventis, Genzyme, Novartis, Teva Pharmaceutical Industries, and Serono Symposia International Foundation. **PS-S** has served on advisory boards for Biogen, Merck, Novartis, Teva, MedDay Pharmaceuticals, and GSK; on steering committees or independent data monitoring boards in trials sponsored by Merck, Teva, GSK, and Novartis; has received speaker honoraria from Biogen Idec, Merck, Teva, Sanofi-Aventis, Genzyme, and Novartis. His department has received research support from Biogen, Merck, Teva, Novartis, Roche, and Genzyme. **PV** has received honoraria or consulting fees from Biogen, Sanofi-Genzyme, Bayer, Novartis, Merck, Celgen, Roche, and Almirall; and research support from Biogen, Sanofi-Genzyme, Bayer, and Merck. **CH** is an employee of Merck KGaA, Darmstadt, Germany. **FD** is an employee of EMD Serono, Inc., a business of Merck KGaA, Darmstadt, Germany.

Supported by Merck KGaA, Darmstadt, Germany.

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors.

GET POSTER PDF

Presented at European Charcot Foundation (ECF) 2017; 30 November - 2 December; Baveno, Italy