Neurodegeneration in multiple sclerosis and neuromyelitis optica: an optical coherence tomography study

M. Pisa, M. Vabanesi, M. Radaelli, F. Ratti, S. Guerrieri, L. Moiola, V. Martinelli, G. Comi, L. Leocani

Neurological Dep.t & INSPE – Institute of Experimental Neurology; University Hospital-IRCCS S.Raffaele, Milano

Introduction

Neurodegeneration is the main determinant of disability in multiple sclerosis (MS). It occurs subclinically even in the very early stages of the disease and often becomes clinically apparent when the progressive phase supervenes. Disability in neuromyelitis optica spectrum disorders (NMOSD) follows a course of early incremental disability, due to frequent and severe relapses; a secondary progressive phase is deemed present only in less than 2% of cases [Wingerchuk, 2007]. This difference in clinical phenotype between the two diseases supports the classical hypothesis that only in MS neurodegeneration occurs independently from apparent inflammatory activity.

Recent findings suggested instead that brain atrophy in NMOSD might occur also independently from previous optic neuritis (ON) or myelitis [Tian, 2017]. Optical coherence tomography (OCT) is an emerging instrumental marker of neurodegeneration, since the thinning of neuroretinal layers has been associated with brain atrophy, disease activity and disability accumulation in MS [Pisa, 2017]. We aimed to assess through optical coherence tomography (OCT) monitoring, the patterns of retinal neurogeneration MS and NMOSD.

Methods

This is a prospective, single-center, observational, longitudinal study. Optic pathway monitoring -OCT, visual acuity and visual evoked potentials- was combined to routine clinical and MRI assessment in 26 NMOSD patients (mean follow-up 2,4 years). A propensity score analysis was used to find a 1:2 matched cohort of MS patients (n=52) with similar baseline characteristics: age, sex, baseline EDSS score, frequency of optic neuritis frequency and disease activity during follow-up (presence vs. absence). MS patients were divided according to NEDA-3 criteria (no relase, no EDSS progression, no new/enlarging or Gd enhancing lesion at brain MRI scan) while for NMOSD patients only clinical data were used.
Generalized estimating equation (GEE) taking into account inter-eye correlation was used for analyses: here reported average inner retinal loss with 95% confidence interval (CI) and Wald test p value.

Results

Population characteristics:

Baseline and follow-up characteristics of the NMOSD and MS cohorts are reported in the table on the right. Among 52 MS patients 23 (44%) were progressive MS and 29 (56%) were RRMS. Age, sex, EDSS at baseline and at follow-up, frequency of ON (ON history was present in the 61.5% of MS and in the 58.7% of NMOSD) and follow-up duration did not differ between the two groups as for inclusion criteria. Disease duration was higher in the MS cohort. At follow-up low-contrast acuity significantly improved at both 2.5% and 1.25% in both groups (p<0.001).

	NMOSD		MS		Mann-Whitney	
	Baseline	Follow-up	Baseline	Follow-up	p values	
Age (year)	46.2 ± 13.3		44.3 ± 9.0		p = n.s.	
Disease duration (year)	7,68 ± 13,3		13,47 ± 9,9		*p=0,026*	
FU duration (year)	2,43 ± 1,1		2,11 ± 0,6		p = n.s.	
EDSS (score)	3,7 ± 2,1	3,31 ± 2	3,2 ± 2,2	3,4 ± 2,4	p = n.s.	p = n.s.
HCVA	0,6 ± 0,5	0,7 ± 0,5	0,95 ± 0,2	0,97 ± 0,3	p=0,002	p=0,008
LCVA 2,5%	0,18 ± 0,23	0,35 ± 0,36	0,25 ± 0,23	0,39 ± 0,32	p = n.s.	p = n.s.
LCVA 1,25%	0,16 ± 0,2	0,32 ± 0,36	0,17 ± 0,24	0,28 ± 0,28	p = n.s.	p = n.s.
N. patients	26		52			
N. eyes studied	41		93			

Peripapilary RNFL:

The **NMOSD cohort** had significant pRNFL thinning at follow-up (-0.150 µm/yr; CI 95% -0.338 to 0.038 µm/yr; Wald p=0.119).
Dividing NMOSD group according to the clinical course, only those with active disease during follow-up had significant pRNFL thinning (n=12; -0.494 µm/yr; CI 95% -0.790 to -0.198 µm/yr; Wald p=0.001) (Figure 1).
In the **MS cohort**, mean pRNFL thinning was -0.521 µm/yr (CI 95% -0.745 to -0.298 µm/yr; Wald p<0.001), with significant loss in NEDA-3 (-0.409 µm/yr; CI 95% -0.801 to -0.017 µm/yr; Wald p=0.041) and those with disease activity during follow-up (-0.645 µm/yr; CI 95% -0.883 to -0.407 µm/yr; Wald p<0.001. Considering the possible interaction of progressive MS course (PMS) with disease activity, only NEDA-3 RRMS had preserved RNFL loss (Figure 2):

- NEDA RRMS: n=13; -0.271 µm/yr; CI 95% -0.892 to 0.349 µm/yr; Wald p=0.392
- NEDA PMS: n=11; -0.556 µm/yr; CI 95% -0.985 to -0.127 µm/yr; Wald p=0.011
- Active RRMS: n=16; -0.724 µm/yr; CI 95% -0.968 to -0.480 µm/yr; Wald p<0.001
- Active PMS: n=12; -0.586 µm/yr; CI 95% -1.002 to -0.169 µm/yr; Wald p=0.006

Macular GCIPL:

GCIPL thickness, available in a subgroup of patients (107/156 eyes: 38 NMOSD and 69 MS) with macular scans, was reduced at follow-up in both MS (-0.442 µm/yr; CI 95% -0.643 to -0.241 µm/yr; Wald p<0.001) and NMOSD (-0.279 µm/yr; CI 95% -0.458 to -0.099 µm/yr; Wald p=0.002), in all subgroups according to disease activity (Figure 3):

- NEDA MS: n=34; -0.476 µm/yr; CI 95% -0.819 to -0.133 µm/yr; Wald p=0.006
- Active MS: n=31; -0.418 µm/yr; CI 95% -0.604 to -0.231 µm/yr; Wald p<0.001
- Stable NMOSD: n=22; -0.251 µm/yr; CI 95% -0.490 to -0.012 µm/yr; Wald p=0.039
- Active NMOSD: n=7; -0.271 µm/yr; CI 95% -0.595 to 0.053 µm/yr; Wald p=0.102

Both active and stable PMS had significant GCIPL thinning (Walt p=0.024 and p=0.015 respectively) while RRMS had a significant reduction in the active (Walt p<0.001) but not in the stable group (Walt p=0.079)
(Figure 4).

Figure 1

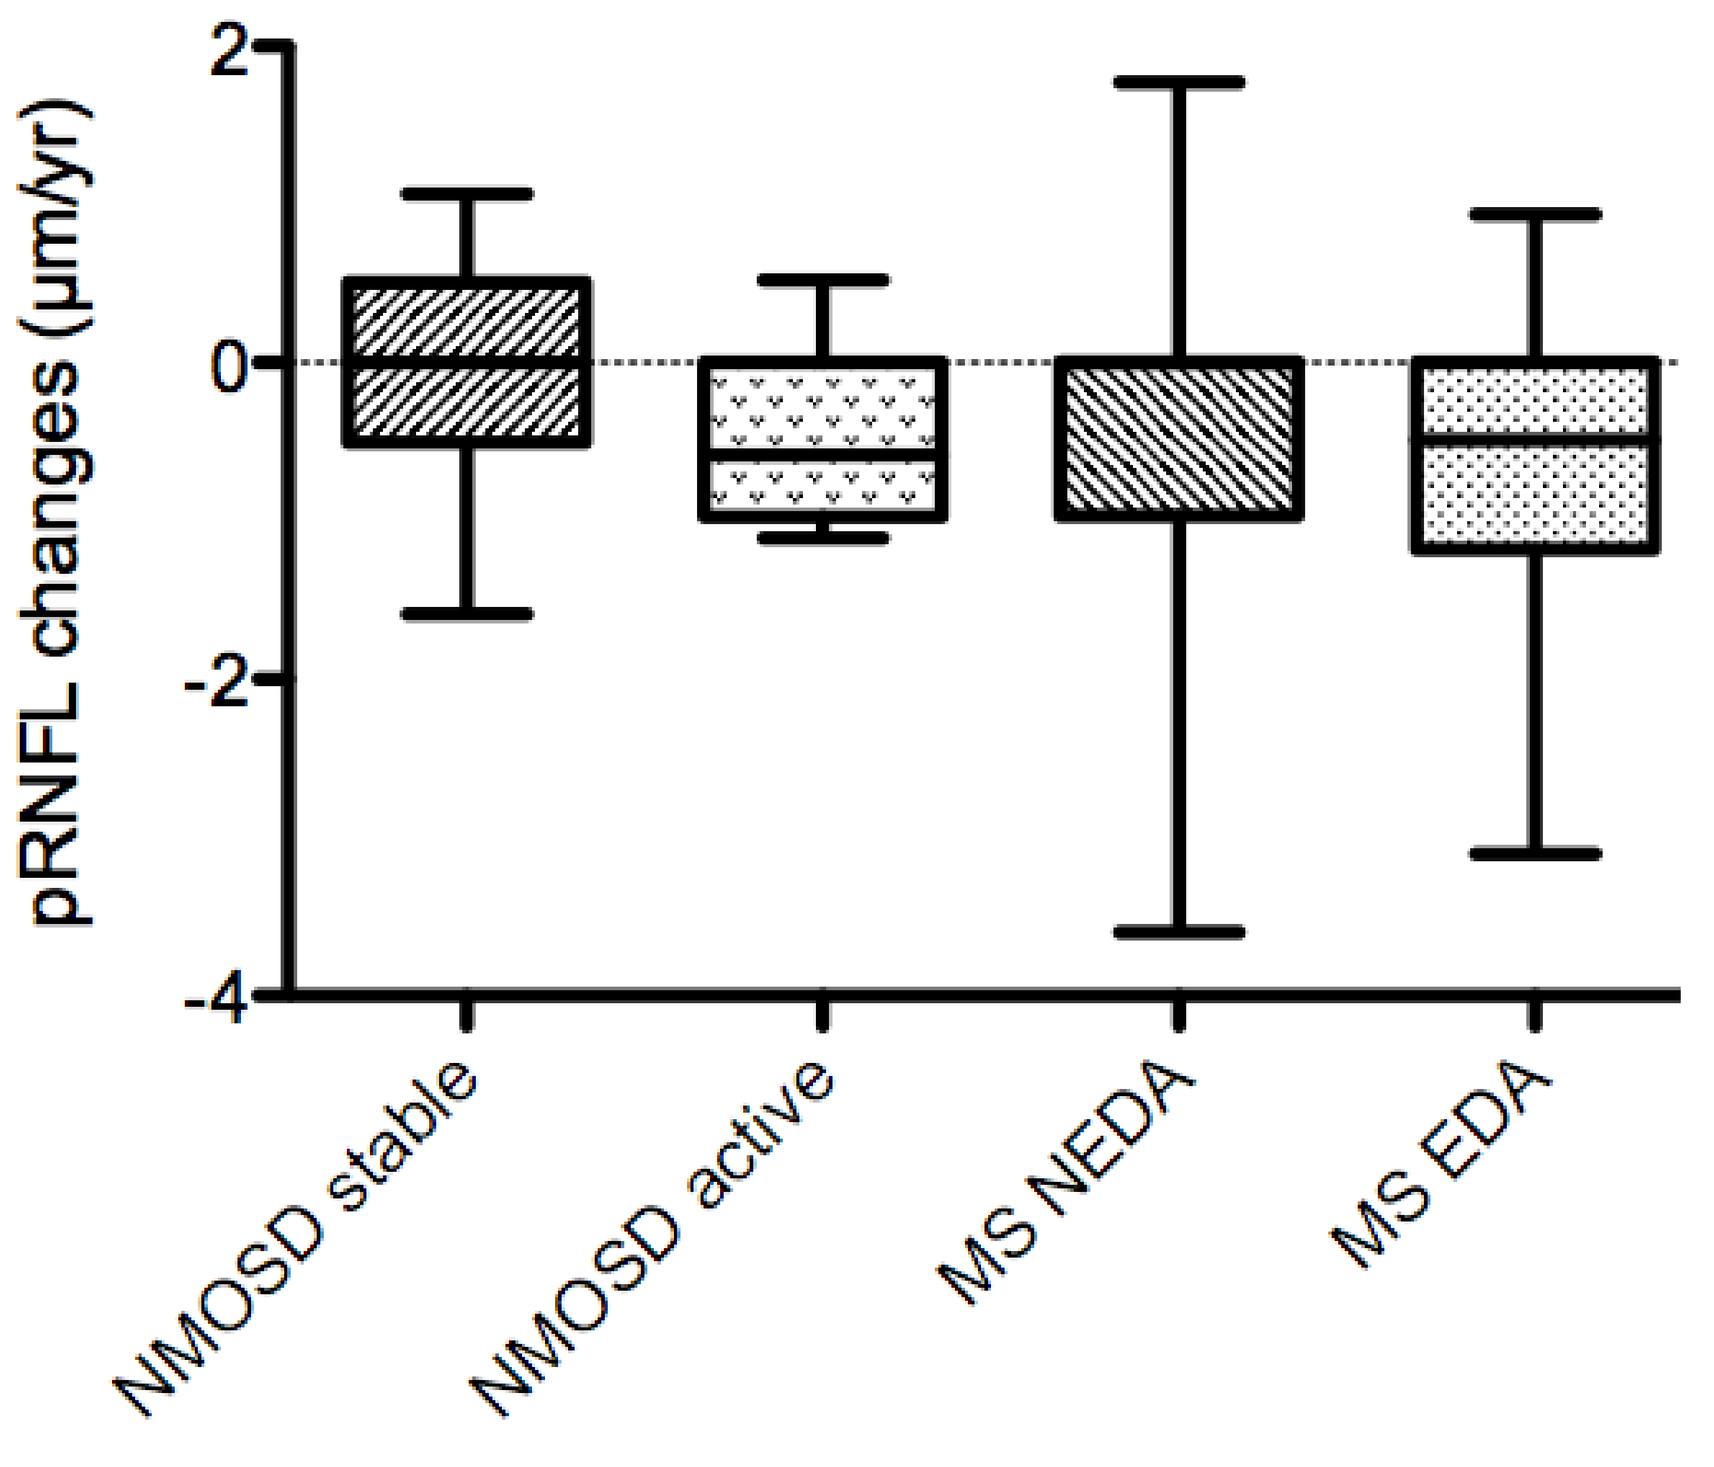
pRNFL changes (µm/yr)
2
0
-2
-4
NMOSD stable
NMOSD active
MS NEDA
MS EDA

Figure 2

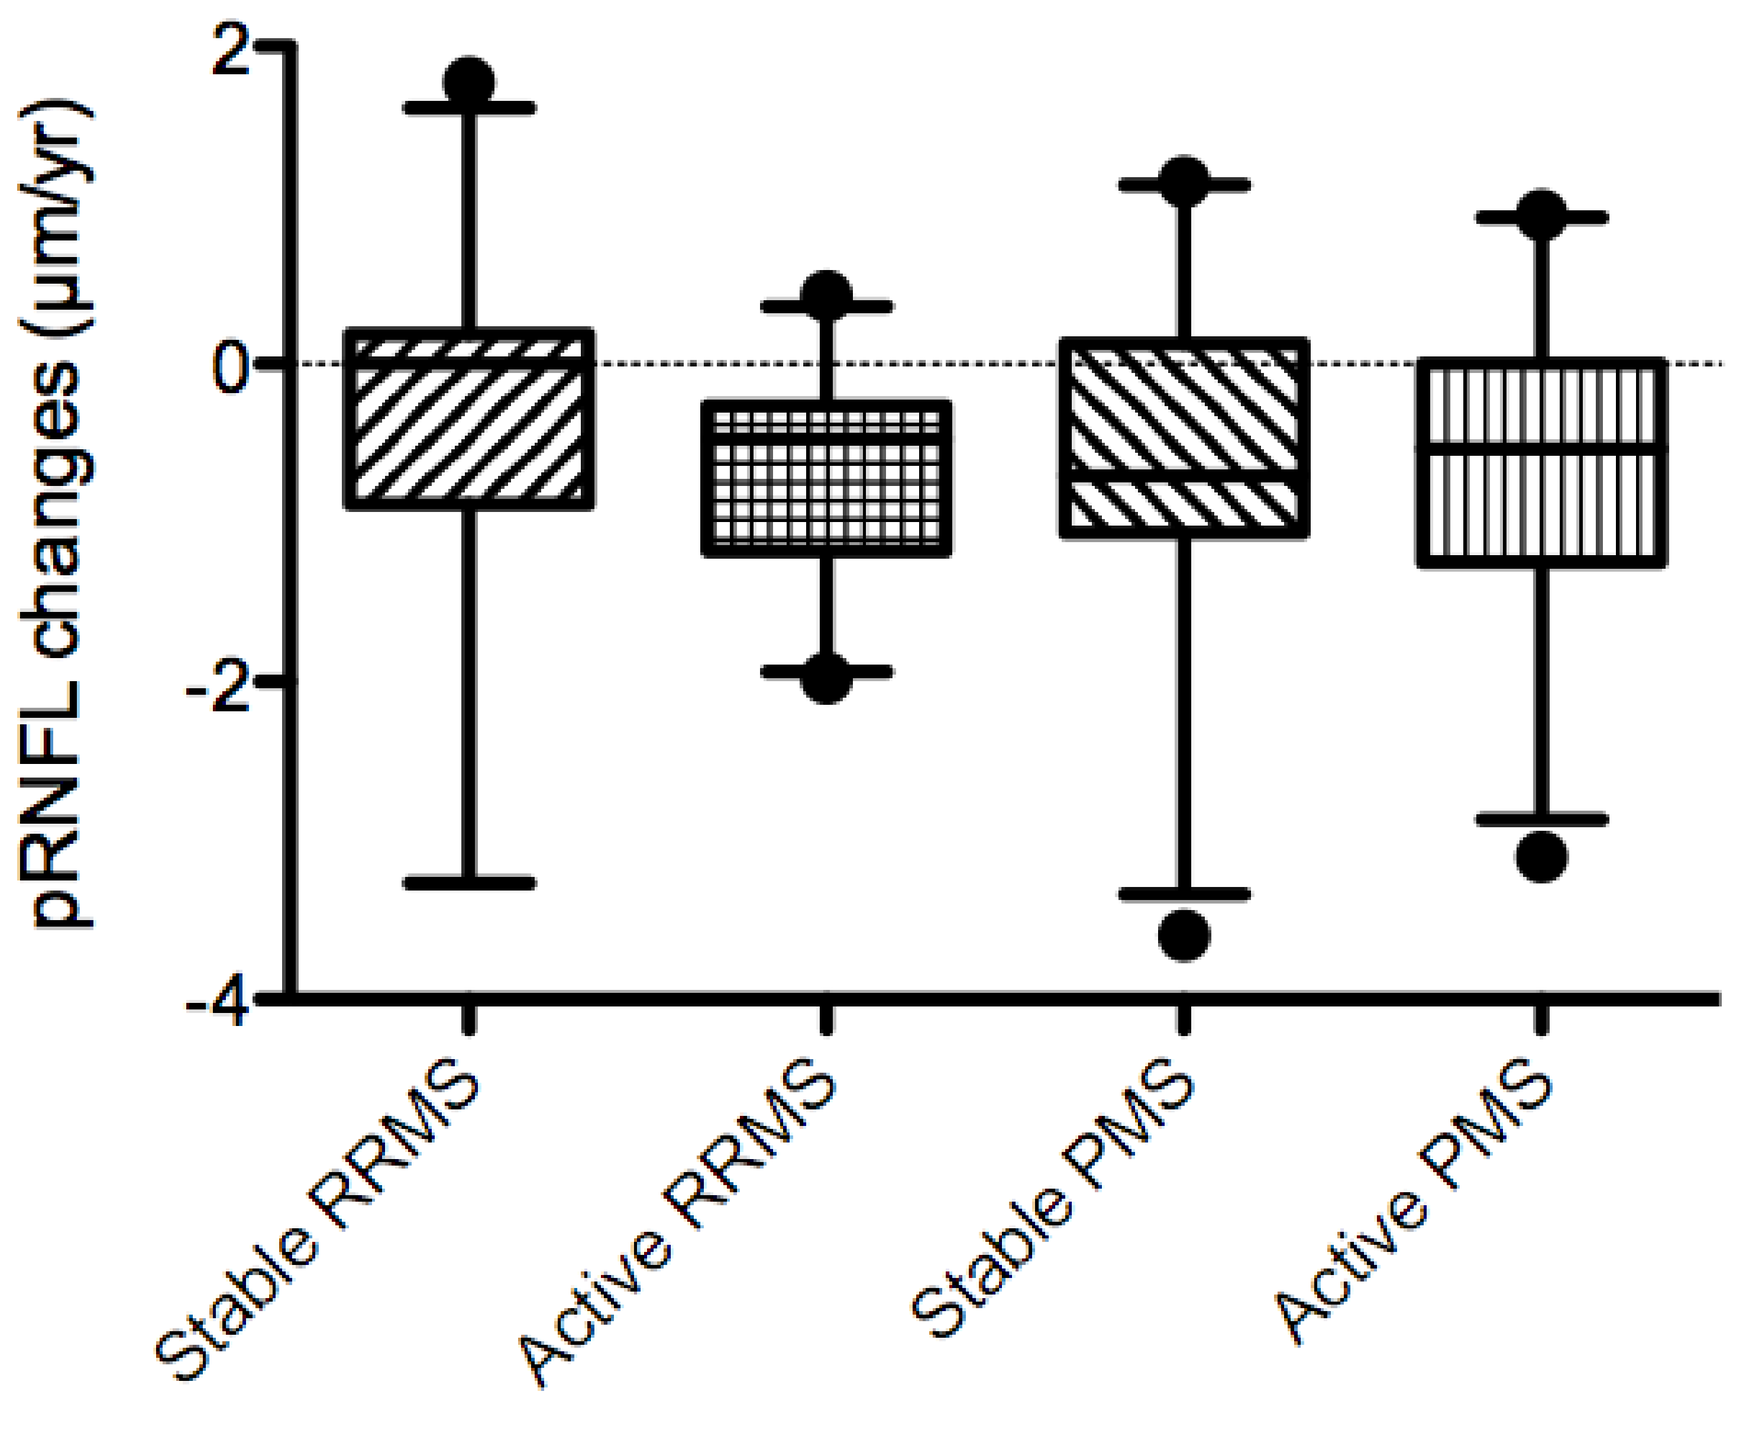
pRNFL changes (µm/yr)
2
0
-2
-4
Stable RRMS
Active RRMS
Stable PMS
Active PMS

Figure 3

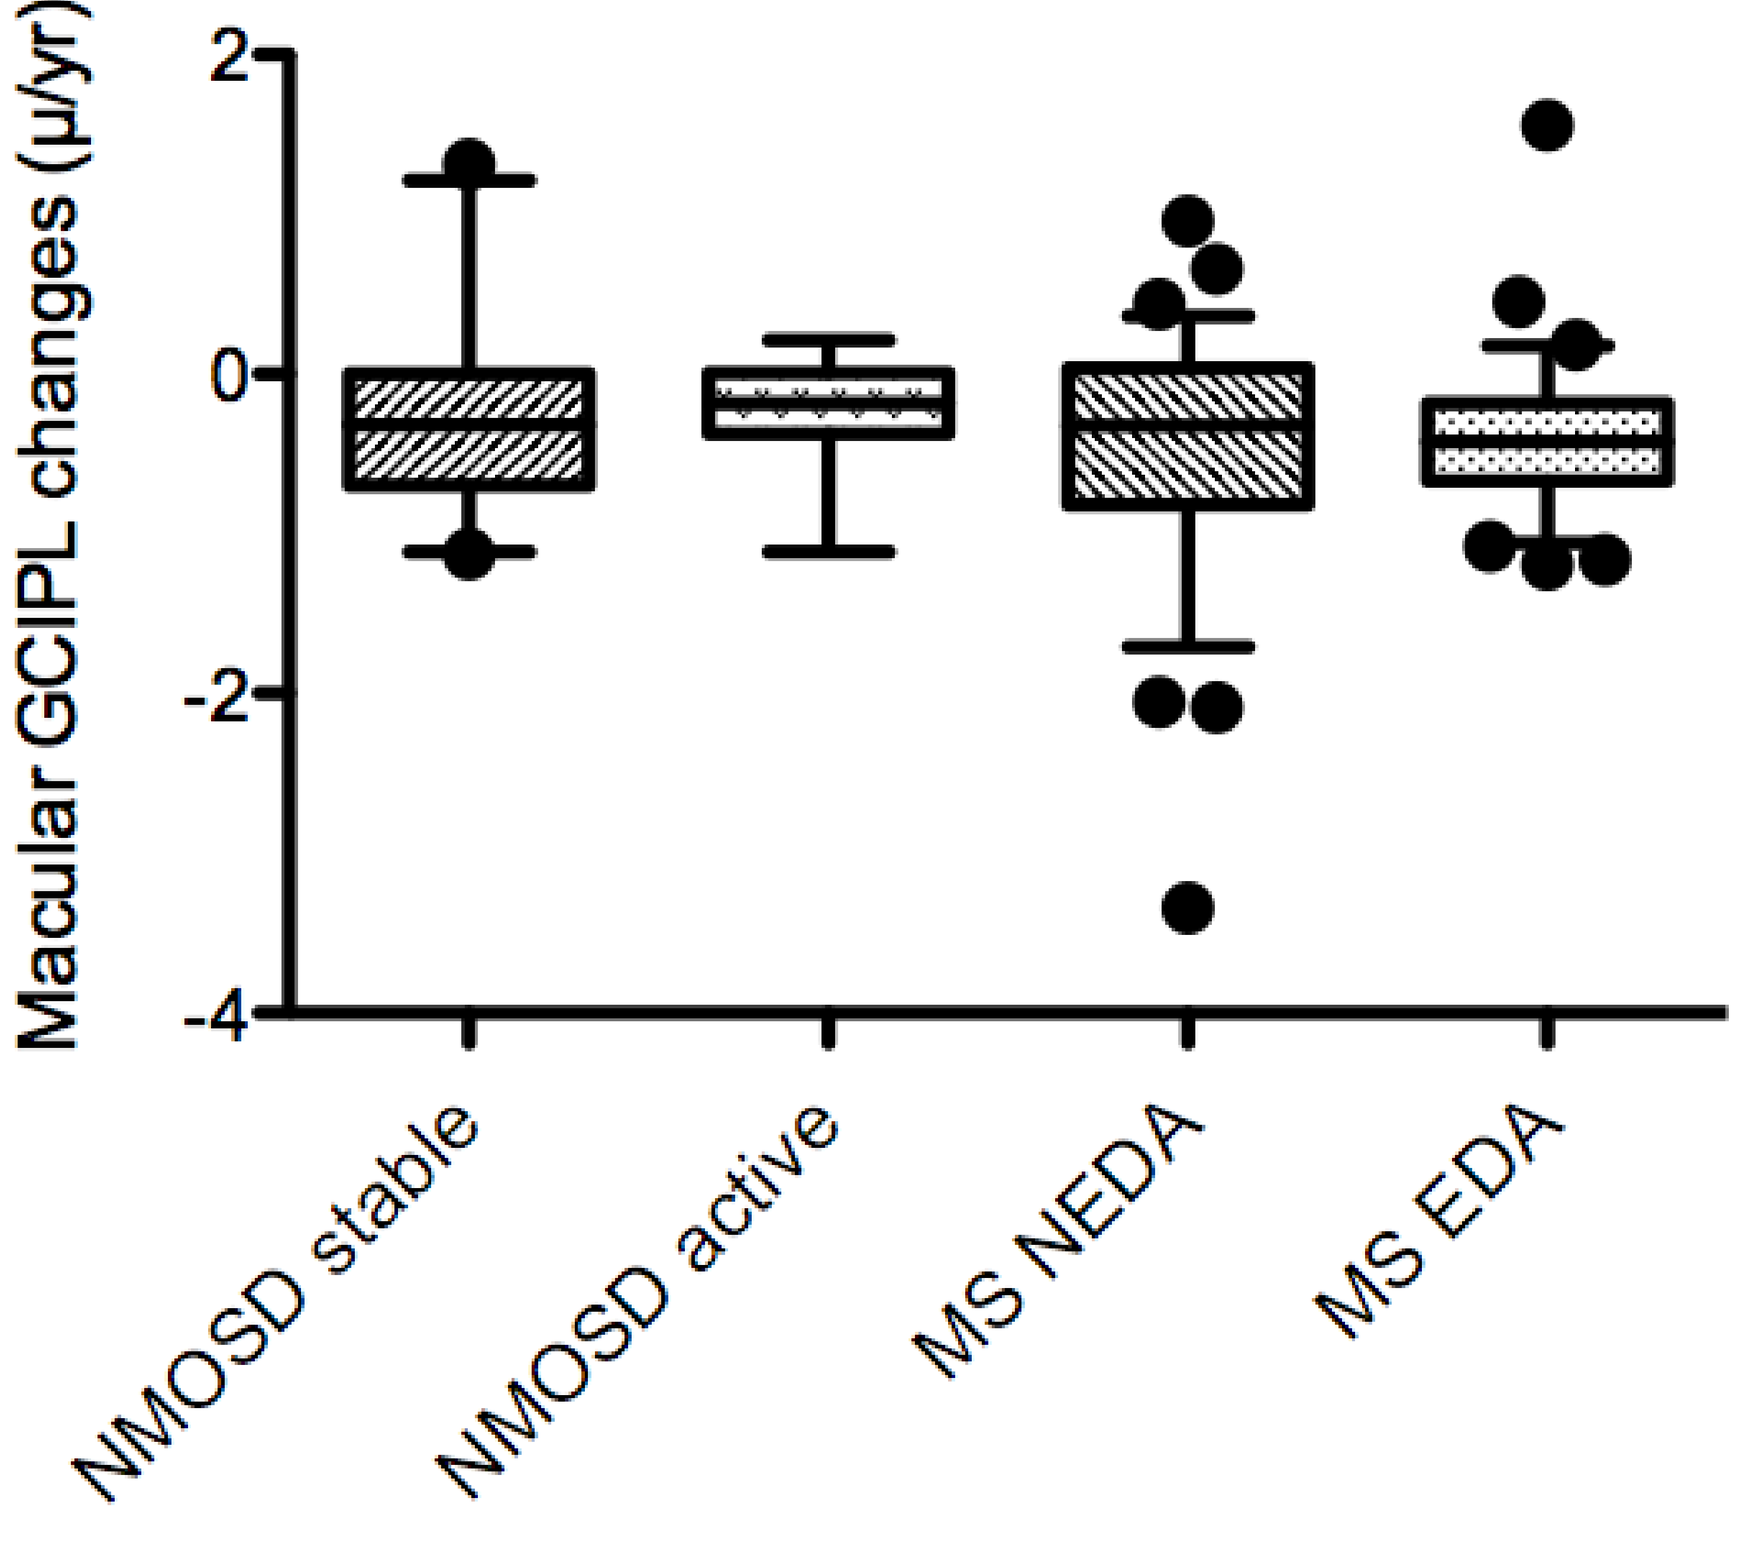
Macular GCIPL changes (µ/yr)
2
0
-2
-4
NMOSD stable
NMOSD active
MS NEDA
MS EDA

Figure 4

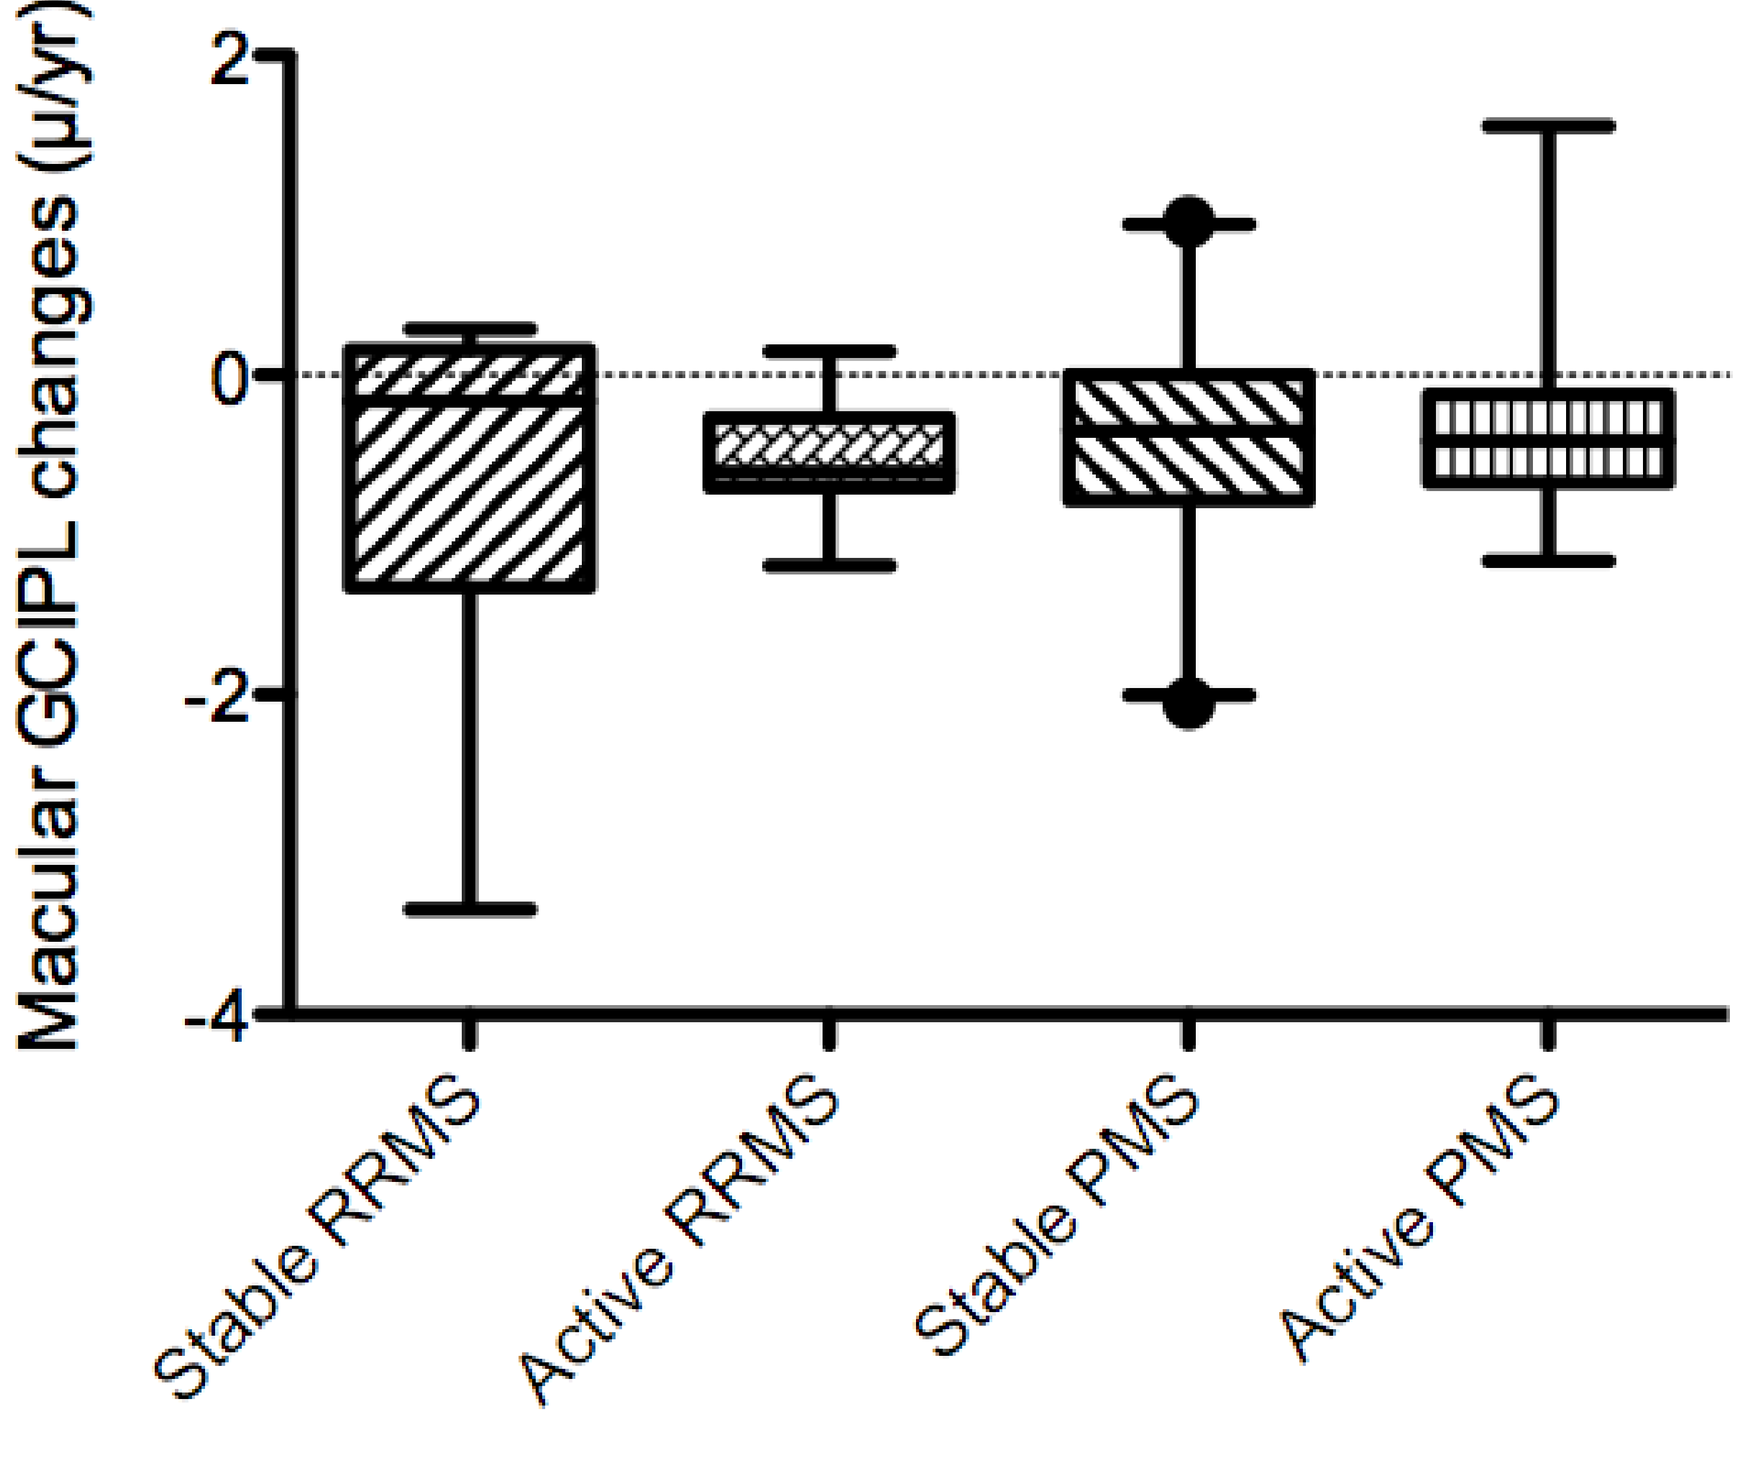
Macular GCIPL changes (µ/yr)
2
0
-2
-4
Stable RRMS
Active RRMS
Stable PMS
Active PMS

Discussion and conclusions

Retinal neuro-axonal loss occurs in both MS and NMOSD not only after acute optic neuritis but also in absence of optic nerve involvement. In relapsing remitting MS, retinal atrophy rates are associated with disease activity, whereas in progressive MS it may occur even without detection of MRI activity or EDSS progression. Instead, NMOSD cohort displays a progressive GCIPL thinning regardless of concurrent disease activity, probably reflecting chronic astrocytopathy, whereas the peripapillary RNFL shows subclinical axonal loss during inflammatory disease reactivation. These findings suggest that OCT may be more sensitive than NEDA3 criteria in detecting subclinical neurodegeneration in PMS. We also found that NMOSD patients GCIPL thinning occurs regardless of disease activity whilst disease relapses are associated with pRNFL thinning.

Disclosures: part of this work was supported by Merck.

Bibliography: Wingerchuk DM, Pittock SJ, Lucchinetti CF, et al. Secondary progressive clinical course is uncommon in neuromyelitis optica. Neurology. 2007 Feb 20;68(8):603-5. Pisa M, Guerrieri S, Di Maggio G, et al. No evidence of disease activity is associated with reduced rate of axonal retinal atrophy in MS. Neurology. 2017 Dec 12;89(24):2469-2475. Tian D-C, Su L, Fan M, et al. Bidirectional degeneration in the visual pathway in neuromyelitis optica spectrum disorder (NMOSD). Mult Scler. 2017;1352458517727604.