Teriflunomide (Aubagio®) International Pregnancy Registry: Enrollment Update

Christine Lebrun-Frénay,[1] David Rog,[2] Myriam Benamor,[3] Stephanie Jurgensen,[4] Philippe Truffinet,[3] Angelo Ghezzi[5]

[1]Hôpital Pasteur 2, Nice, France; [2]Greater Manchester Neurosciences Centre, Salford Royal NHS Foundation Trust, Salford, UK; [3]Sanofi, Chilly-Mazarin, France; [4]Sanofi, Cambridge, MA, USA; [5]Ospedale di Gallarate, Gallarate, Italy

OBJECTIVE

- **To provide an enrollment update for the International Teriflunomide Pregnancy Exposure Registry. Table 1 outlines the primary and secondary objectives of the registry**

CONCLUSIONS

- **The International Teriflunomide Pregnancy Exposure Registry will provide outcomes on teriflunomide-exposed pregnancies, in addition to infant development during the first year of life**
- **Findings from this registry, together with those of the US/Canadian Teriflunomide Pregnancy Exposure Registry, will inform HCPs when counseling women exposed to teriflunomide during pregnancy**

Table 1. International Teriflunomide Pregnancy Exposure Registry Objectives[1]

Primary objective	• Compare rate of birth defects in teriflunomide-exposed pregnant women with those reported by the population-based European surveillance system, EUROCAT[2]
Secondary objectives	• Compare rate of birth defects in teriflunomide-exposed pregnant women with those reported by the population-based US surveillance system, MACDP[3] • Estimate proportions of pregnancy outcomes, including live-born infants, in teriflunomide-exposed pregnant women • Estimate proportions of preterm live births (<37 weeks of gestation) among live-born infants of teriflunomide-exposed pregnant women • Estimate proportions of alterations in fetal/infant growth, indications of delayed development, and functional deficits observed during first year of life in live-born infants of teriflunomide-exposed pregnant women

EUROCAT, European Surveillance of Congenital Anomalies; MACDP, Metropolitan Atlanta Congenital Defects Program.

INTRODUCTION

- Teriflunomide is a once-daily oral immunomodulator approved for relapsing-remitting MS in 80 countries. As of October 2017, over 80,000 patients are being treated with teriflunomide, with a total real-world exposure of approximately 162,000 patient-years since approval
- Alongside a consistent, well-characterized safety and tolerability profile,[4–7] teriflunomide has demonstrated consistent efficacy on clinical and MRI disease activity in patients with relapsing forms of MS[4–6] and in those who experienced a first clinical episode suggestive of MS[7]
- Use of teriflunomide is contraindicated in pregnant women and women of childbearing potential not using reliable contraception, owing to the observation of teratogenicity and embryo-lethality in the offspring of teriflunomide-treated rats and rabbits[8]
 - Rats exhibit greater sensitivity to the effects of teriflunomide than humans, which may explain why similar plasma exposures of teriflunomide have resulted in teratogenicity in animals but not, to date, in humans[9]
- Teriflunomide elimination can be accelerated in patients by the administration of cholestyramine or activated charcoal after stopping teriflunomide treatment[8]
- Teriflunomide is the principal active metabolite of leflunomide (approved since 1998[10] for the treatment of rheumatoid arthritis)
 - A prospective study conducted by the Organization of Teratology Information Specialists (OTIS) found no significant differences in rates of major structural defects, and no pattern of minor or major anomalies, in newborns of women exposed to leflunomide compared with disease-matched or healthy comparator groups.[11] These observations were confirmed in a subsequent OTIS case series[12]
- Despite the requirement for contraceptive use, a number of pregnancies were reported during the teriflunomide clinical trial program; an update on these outcomes together with those in the postmarketing setting was presented recently at ECTRIMS-ACTRIMS 2017[13]
- Although there is no evidence of a signal for teratogenicity or other adverse outcomes, it is important to collect data regarding teriflunomide exposure in pregnancy to evaluate any potential adverse outcomes[14]
- Global teriflunomide pregnancy registries have been established and will capture prospective data from pregnancies within the postmarketing setting

METHODS

Registry Design

- The registry is an ongoing, voluntary, multinational, prospective, observational, exposure-registration study operating in the following countries:
 - Australia, Austria, Belgium, Czech Republic, Denmark, Finland, France, Germany, Greece, Ireland, Italy, the Netherlands, Norway, Spain, Sweden, Switzerland, and the United Kingdom (**Figure 1**)

Figure 1. Map of Countries Participating in the International Teriflunomide Pregnancy Exposure Registry[15]

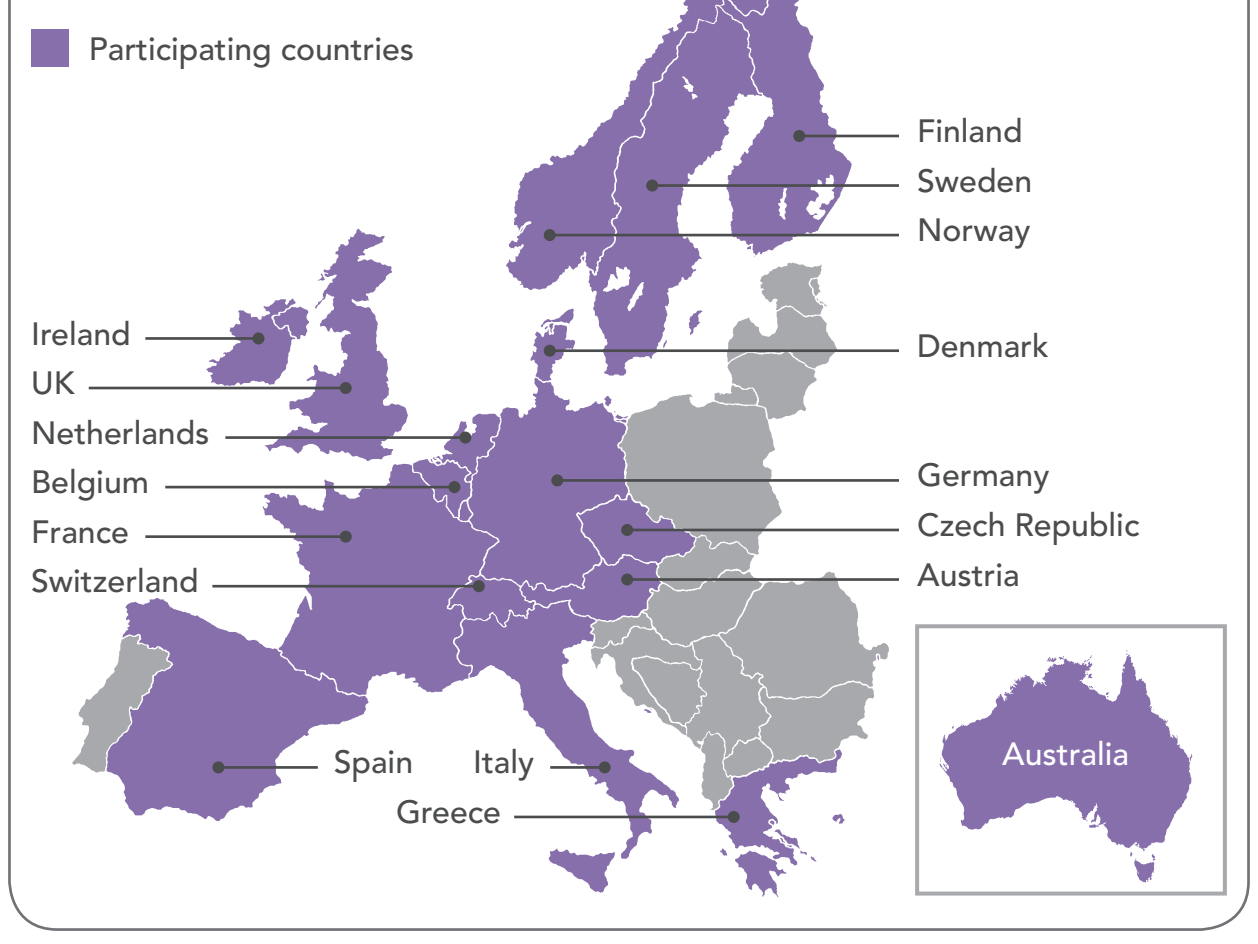
Participating countries
Finland
Sweden
Norway
Denmark
Ireland
UK
Netherlands
Belgium
France
Switzerland
Germany
Czech Republic
Austria
Spain
Italy
Greece
Australia

- National Coordinators liaise with healthcare professionals (HCPs) to collect information on teriflunomide-exposed pregnancies and oversee enrollment in the registry
- To maximize recruitment, an open-enrollment approach is employed, with participation available to all eligible women through reporting HCPs, such as obstetricians, neurologists, and general practitioners
- The registry design and inclusion/exclusion criteria are shown in **Figure 2** and **Table 2**, respectively

Figure 2. Registry Design[1]

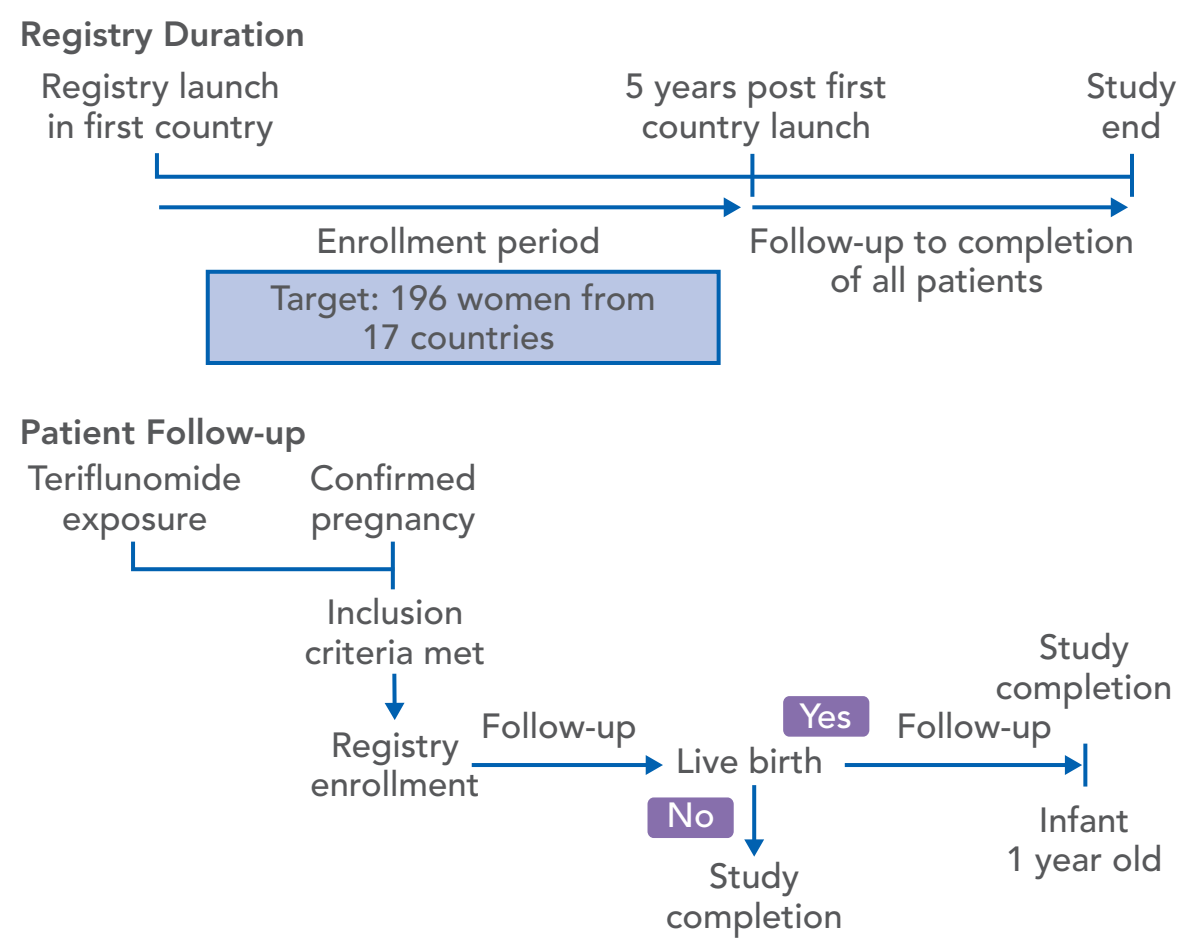
Registry Duration
Registry launch in first country
5 years post first country launch
Study end
Enrollment period
Target: 196 women from 17 countries
Follow-up to completion of all patients
Patient Follow-up
Teriflunomide exposure
Confirmed pregnancy
Inclusion criteria met
Registry enrollment
Follow-up
Live birth
Yes
No
Follow-up
Study completion
Infant 1 year old
Study completion

Table 2. Inclusion and Exclusion Criteria[1,a]

Inclusion criteria	**Pregnant women with MS who:** • Have teriflunomide exposure (any dose, any duration, any time) after Day 1 of last menstrual period until pregnancy end • Receive healthcare in the participating countries (shown above) • Provide written informed consent • Authorize release of medical information for self and live-born infant(s) • Are not participating in a teriflunomide clinical trial at time of pregnancy exposure
Exclusion criteria	**Teriflunomide-exposed pregnant women with MS who:** • Do not receive healthcare in a country where a registry is operational • Were participating in a clinical trial investigating teriflunomide at time of pregnancy exposure

[a]If a National Coordinator determines that a patient is not eligible, all completed forms will be sent to the Registry Coordinating Center for tracking purposes. These cases will not be entered into the registry database.

Patient Enrollment

- Once a new teriflunomide-exposed pregnancy is reported, HCPs contact the patient to confirm they meet inclusion criteria and obtain their consent for enrollment in the registry
- To enroll a patient, HCPs can contact the National Coordinating Center in their respective country

Outcomes

- Pregnancy outcomes in addition to infant characteristics during the first year of life are being collected (**Table 3**)

Table 3. International Teriflunomide Pregnancy Registry Information Collation[1]

Maternal information	• Demographics • Current pregnancy information (LMP, EDD, age at conception) • Obstetric history, including history of birth defects • Family history of birth defects (maternal/paternal) • Concomitant medications and other exposures • Teriflunomide and accelerated elimination procedure (agent dosage, duration, results), pregnancy attribution • Concurrent acute or chronic medical conditions during pregnancy, including MS (history and status) • Prenatal tests (type, gestational age, results)
Pregnancy outcome	• Live birth • Spontaneous abortion (<20 weeks of gestation) • Fetal death (≥20 weeks of gestation) • Induced abortion without evidence of birth defects • Termination of pregnancy for fetal anomaly after prenatal diagnosis • Ectopic pregnancy • Molar pregnancy • Neonatal (28 days after live birth) or maternal (during pregnancy or at time of delivery) death
Birth defects	• Birth defects will be classified according to EUROCAT[2] and MACDP[3] conventions, and reviewed by the Registry's birth defect evaluator
Infant characteristics	• Infant characteristics, including prematurity and serious adverse outcomes, observed during first year of life

EDD, estimated date of delivery; EUROCAT, European Surveillance of Congenital Anomalies; LMP, first day of last menstrual period; MACDP, Metropolitan Atlanta Congenital Defects Program.

Statistical Analysis

- The registry aims to enroll 196 pregnant women, projected to result in 104 live births; this sample size is estimated to provide an 80% power to detect a 3.95-fold increase in risk ratio of birth defects associated with teriflunomide exposure vs EUROCAT[2]
- Analyses will be based on prospective cases of women with teriflunomide exposure during pregnancy prior to the knowledge, or perceived knowledge, of pregnancy outcome (ie, structural defect or genetic abnormality noted on a prenatal test), and will be conducted in 3 populations:
 - **Primary analysis population:** Eligible pregnant women with available pregnancy outcomes and birth-defect status of any live-born infant(s) available at birth or 1-year follow-up. Used for evaluation of primary objective and rate of birth defect (secondary objective)
 - **Pregnant women population:** Eligible pregnant women with pregnancy outcomes available. Used for evaluation of secondary objectives related to pregnancy outcomes
 - **Live infant population:** All live-born infants from the pregnant women population. Used for evaluation of secondary objectives related to live births
- Retrospective cases are not included in the calculation of birth defect rates (but will be reviewed carefully by the Registry and summarized separately in interim and final reports)
- Teriflunomide pregnancy exposure data will be classified by gestational week and trimester

RESULTS

- Patient enrollment commenced in early 2015 and is planned to continue until December 2019
- Interim data with a cutoff date of April 26, 2017, have been collected from the registry
 - Fourteen patients have been recruited from 7 countries: 4 each from France and Spain; 2 from Germany; and 1 each from Australia, Denmark, Greece, and Italy
 - Outcomes are available for 7 pregnancies:
 - Six healthy babies have been born, with no abnormalities reported to date
 - One patient in Spain had an elective termination that was not motivated by the result of a prenatal test or by any suspicion of a potential birth defect

References

1. Hellwig et al. Poster P1159, EAN 2015.
2. European Surveillance of Congenital Anomalies. Guide 1.3 and reference documents, 2013. http://www.eurocat-network.eu/. Accessed Aug 2017.
3. Centers for Disease Control and Prevention. MACDP 6-Digit Code Defect List, 2007. https://www.cdc.gov/ncbddd/birthdefects/documents/macdpcode0807.pdf. Accessed Aug 2017.
4. O'Connor et al. *Neurology*. 2006;66:894.
5. O'Connor et al. *N Engl J Med*. 2011;365:1293.
6. Confavreux et al. *Lancet Neurol*. 2014;13:247.
7. Miller et al. *Lancet Neurol*. 2014;13:977.
8. AUBAGIO® (teriflunomide) SmPC. Sanofi-Aventis Groupe, 2017.
9. Davenport et al. Poster IP002, DGN 2017.
10. ARAVA® (leflunomide) PI. Food and Drug Administration, 2015.
11. Chambers et al. *Arthritis Rheum*. 2010;62:1494.
12. Cassina et al. *Arthritis Rheum*. 2012;64:2085.
13. Vukusic et al. Oral Presentation 205, ECTRIMS-ACTRIMS, 2017.
14. Kieseier et al. *Neurol Ther*. 2014;3:133.
15. Lebrun-Frénay et al. Poster EP1732, ECTRIMS-ACTRIMS 2017.

Acknowledgments and Disclosures

This poster was reviewed by Alex Lublin, PhD, and Jonathan Valenzano, PharmD, of Sanofi. Editorial support for this poster was provided by Louise Gildea, of Fishawack Communications, and was funded by Sanofi.

CLF: Consulting fees, honoraria, or scientific committee support (Biogen, Genzyme, MedDay, Merck Serono, Novartis, Roche, Teva). **DR:** Consulting fees (Bayer Schering, Biogen, MedDay, Merck Serono, Novartis, Roche, Sanofi, Teva Neuroscience); research support (Biogen Idec, Genzyme, GW Pharma, Merck Serono, Mitsubishi, Novartis, Teva Neuroscience). **MB:** Employee of Sanofi. **SJ** and **PT:** Employees of Sanofi, with ownership interest. **AG:** Consultancy fees (Almirall, Biogen Idec, Genzyme, Merck Serono, Mylan); travel grants or grants as speaker (Merck Serono, Novartis, Sanofi/Genzyme, Teva Neuroscience).

Data included in this poster were presented at EAN 2015 and ECTRIMS-ACTRIMS 2017.

Teriflunomide is approved in many countries, including the US and the European Union, for the treatment of relapsing multiple sclerosis or relapsing-remitting multiple sclerosis. This material may contain information that is outside of the approved labeling in some countries.

Presented at the 25th Annual Meeting of the European Charcot Foundation, November 30–December 2, 2017, Baveno, Italy

Funding provided by Sanofi