75

Effect of Ozanimod (RPC1063) on Action Potential Parameters in Adult Human Purkinje Fibers

Najah Abi-Gerges[1], Paul E. Miller[1], Andre Ghetti[1], Fiona L. Scott[2], Kristen R. Taylor Meadows[2], Bryan Clemons[2], Paul Frohna[2], Guy Page[1], Gregory J. Opiteck[2]

[1]AnaBios Corporation, San Diego, California, United States; [2]Receptos, a wholly owned subsidiary of Celgene Corporation, San Diego, California, United States

INTRODUCTION

- Isolated human Purkinje fibers (PFs) in combination with electrophysiology are a unique tool that can be used to understand subtle differences between drugs on cardiac conduction
- Fingolimod, a drug used to treat multiple sclerosis, has been associated with second degree or higher atrioventricular (AV) block[1]
- Ozanimod has not been shown to be associated with comparable cardiac abnormalities[2,3]
- Ozanimod (RPC1063) is an oral, once-daily immunomodulator selectively targeting sphingosine 1-phosphate (S1P) receptor 1 ($S1P_{R1}$) and 5 ($S1P_{R5}$) that has shown therapeutic benefit in clinical trials of relapsing multiple sclerosis and ulcerative colitis[4,5]
- A key difference between fingolimod and ozanimod is the former's constitutive signaling through $S1P_{R3}$
- To determine the causality of $S1P_{R3}$ agonism on higher-order human cardiac conduction and, therefore, determine whether the higher selectivity of ozanimod explains its improved clinical profile

METHODS

- At pacing rates of 1 and 2 Hz (mimicking normal and elevated heart rates, respectively), human PFs from female hearts were used to evaluate the effects of ozanimod and fingolimod on action potential duration (APD) at 30%, 50%, and 90% repolarization (APD30, APD50, and APD90, respectively) and on recognized pro-arrhythmia predictors (triangulation [APD90–APD30], short-term variability [STV] of APD90, and incidence of early afterdepolarizations [EAD]) (Figure 1)
- Vehicle control (DMSO 0.3%) and flecainide (10 µM) were used to determine the stability and responsiveness, respectively, of the PFs[6]

Figure 1. Experimental Design: A) Schematic Image of the Human Heart; B) Isolated PFs; C) PFs in the Experimental Apparatus; D) PF Action Potential; E) Experimental Design Used

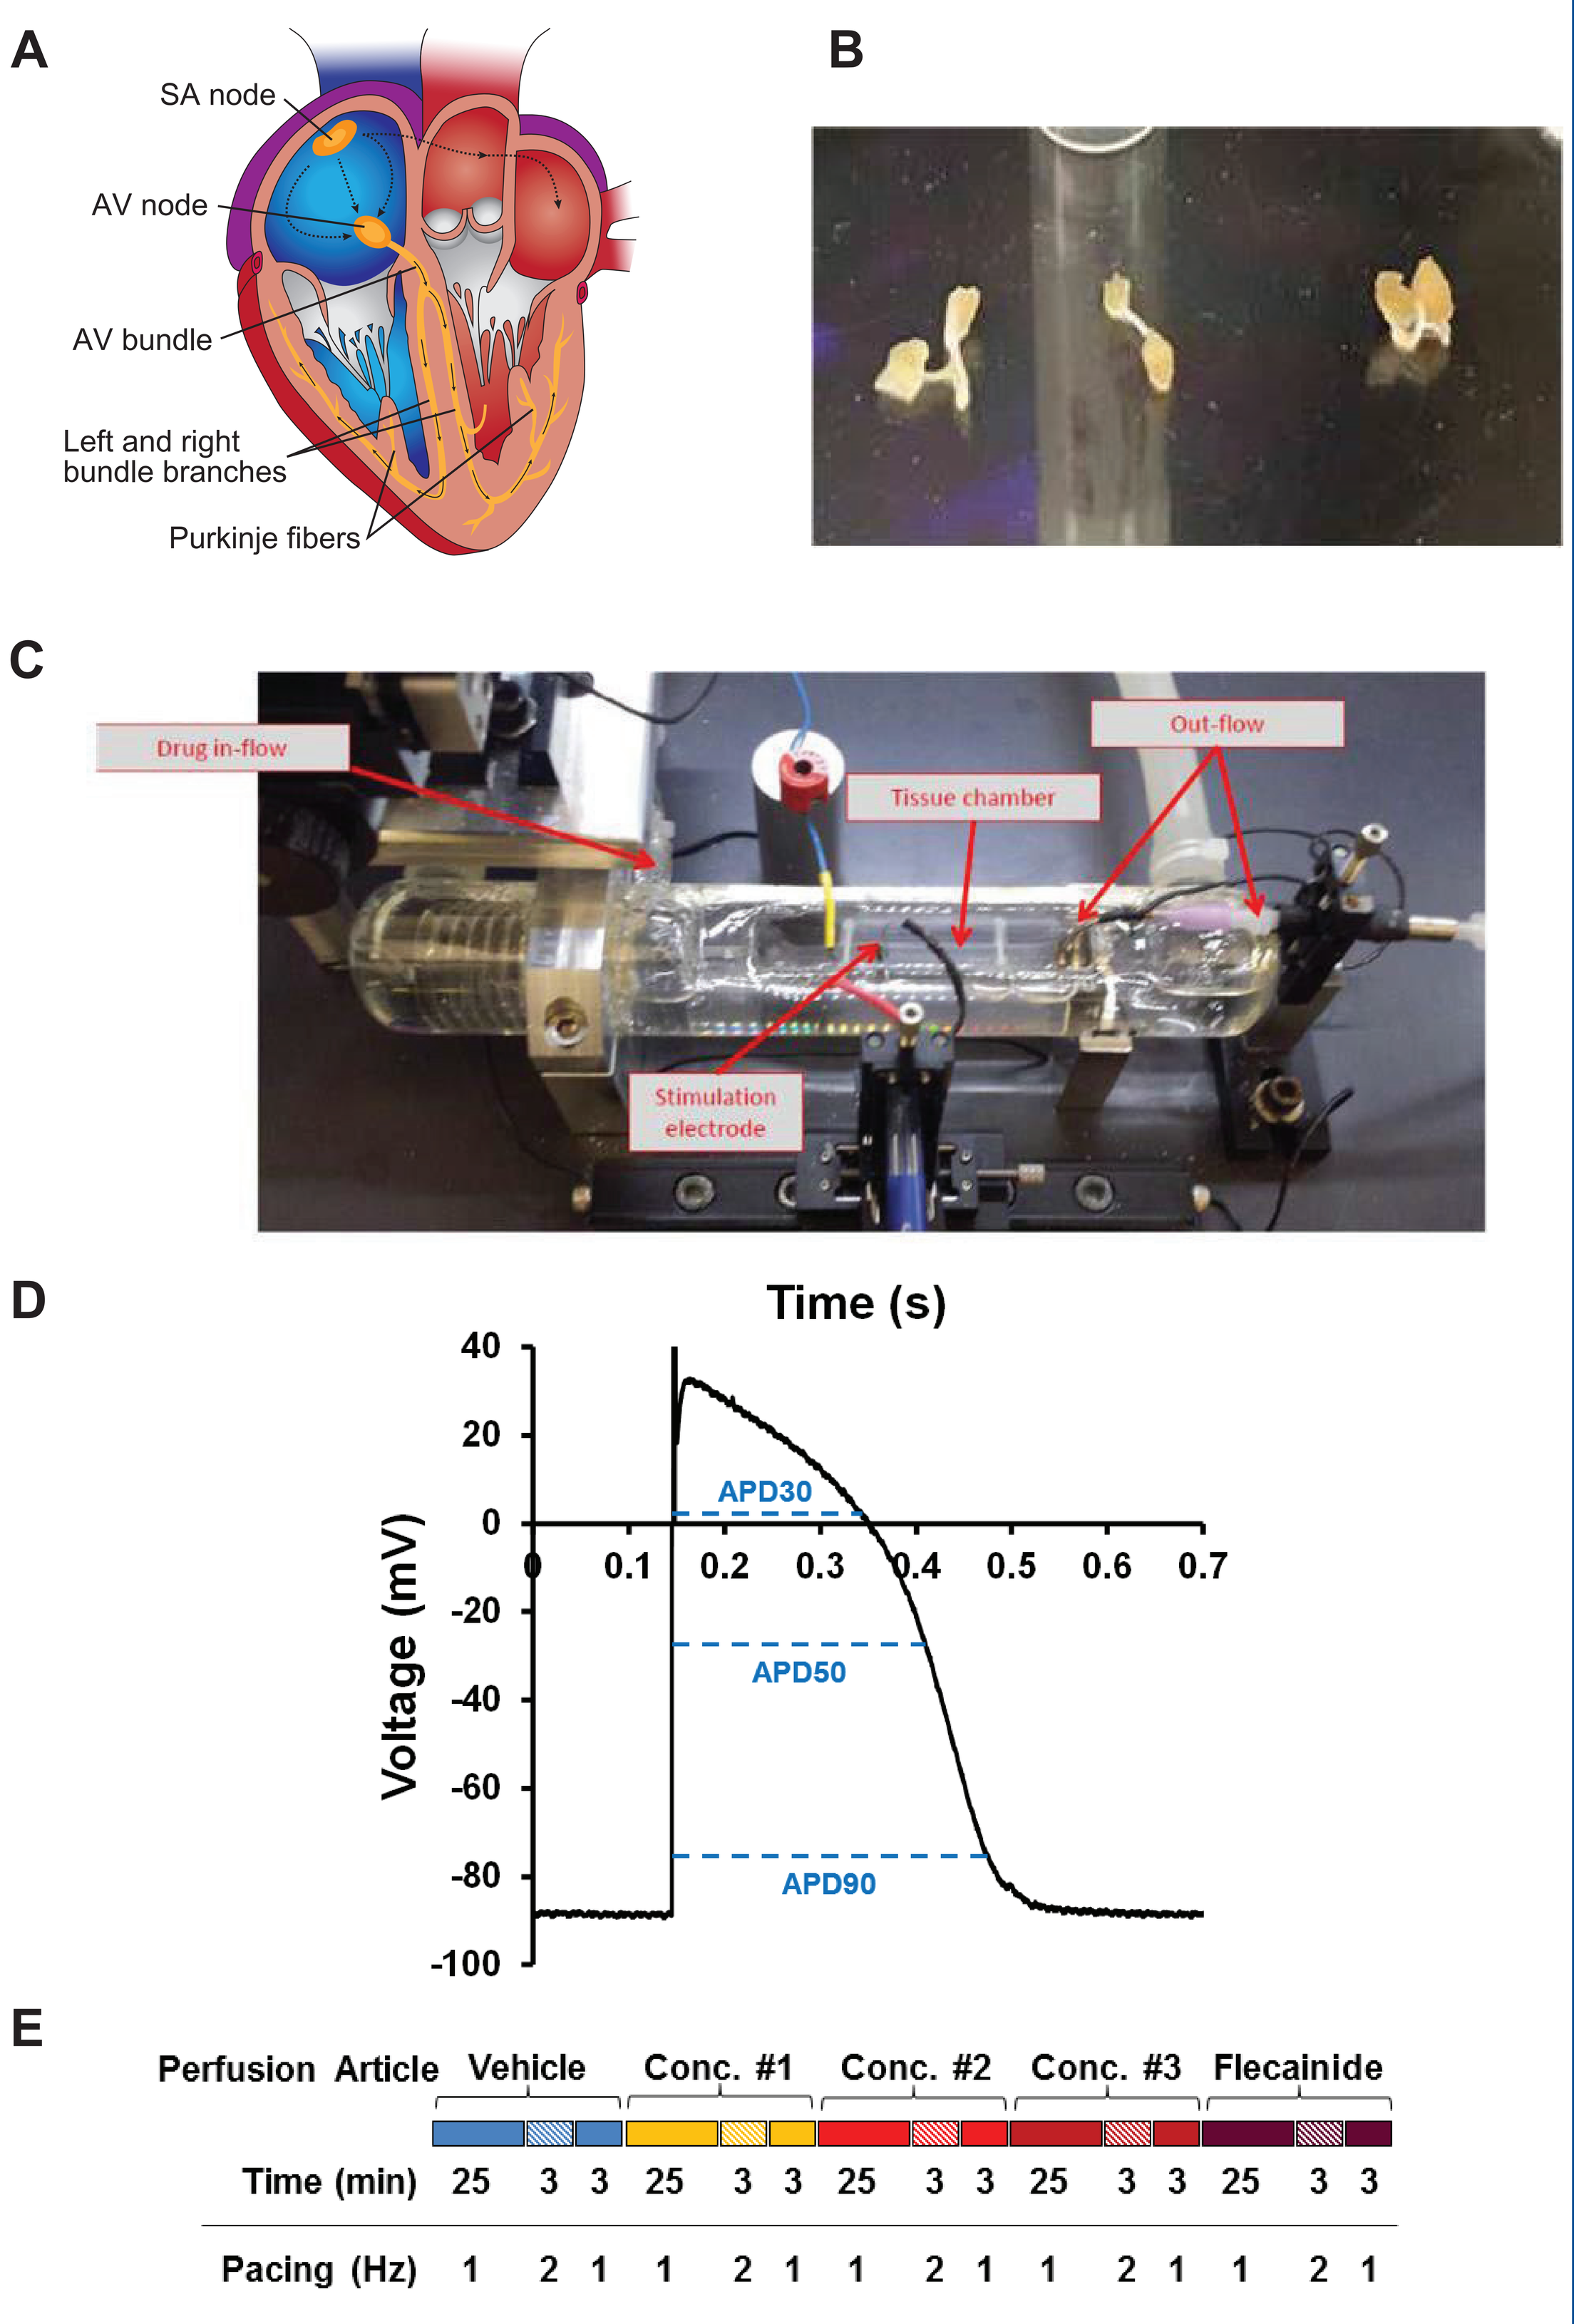
A
SA node
AV node
AV bundle
Left and right bundle branches
Purkinje fibers
B
C
Drug in-flow
Tissue chamber
Out-flow
Stimulation electrode
D
Time (s)
Voltage (mV)
APD30
APD50
APD90
E
Perfusion Article Vehicle Conc. #1 Conc. #2 Conc. #3 Flecainide
Time (min) 25 3 3 25 3 3 25 3 3 25 3 3 25 3 3
Pacing (Hz) 1 2 1 1 2 1 1 2 1 1 2 1 1 2 1

APD, action potential duration; AV, atrioventricular; conc, concentration; min, minutes; PF, Purkinje fibers; s, seconds; SA, sinoatrial.

RESULTS

Ozanimod showed no significant effects on APD or pro-arrhythmia markers

- Ozanimod, up to a physiological concentration of 150 nM, had no significant effects on APD and did not increase the manifestation of pro-arrhythmia markers or induce beat escape (BE); the electrical stimulus does not trigger an action potential after full repolarization (Figures 2, 3, and 4)

Fingolimod showed effects on pro-arrhythmia markers

- Although fingolimod, up to a physiological concentration of 500 nM, also had no significant effects on APD (Figure 7), triangulation, or EADs, it did elicit an increase in STV (APD90) (Figure 6)
- Moreover, 50 nM fingolimod showed BE, which was more pronounced at 500 nM (Figures 5 and 6)
- Further, PAT-1229, an $S1P_{R3}$ antagonist, was able to reverse fingolimod-induced BE
- Vehicle controls showed no change in APD or any manifestation of pro-arrhythmic markers or BE (data not shown)

DISCUSSION

- The model recapitulated the clinical observations that fingolimod treatment causes dose-dependent cardiac conduction abnormalities at concentrations similar to those reported in the peripheral blood and which appear to be the result of $S1P_{R3}$ agonism
- The data agreed with the clinical observation that ozanimod has no effect on the measured conduction parameters, which appear to be the result of ozanimod's higher selectivity
- Future work on cardiac conduction may involve the un-phosphorylated form of fingolimod and the active metabolites of ozanimod and may be extended to the effect of S1P agonists on heart rhythm, through the use of isolated and perfused human sinoatrial node

RESULTS

The positive control, flecainide, showed changes in APD, triangulation, and BE

- In contrast, flecainide decreased APD30 and APD50, increased triangulation and STV (APD90), with no induction of EADs, but elicited BE (Figures 2 and 5)

Figure 2. Ozanimod Showed No BE

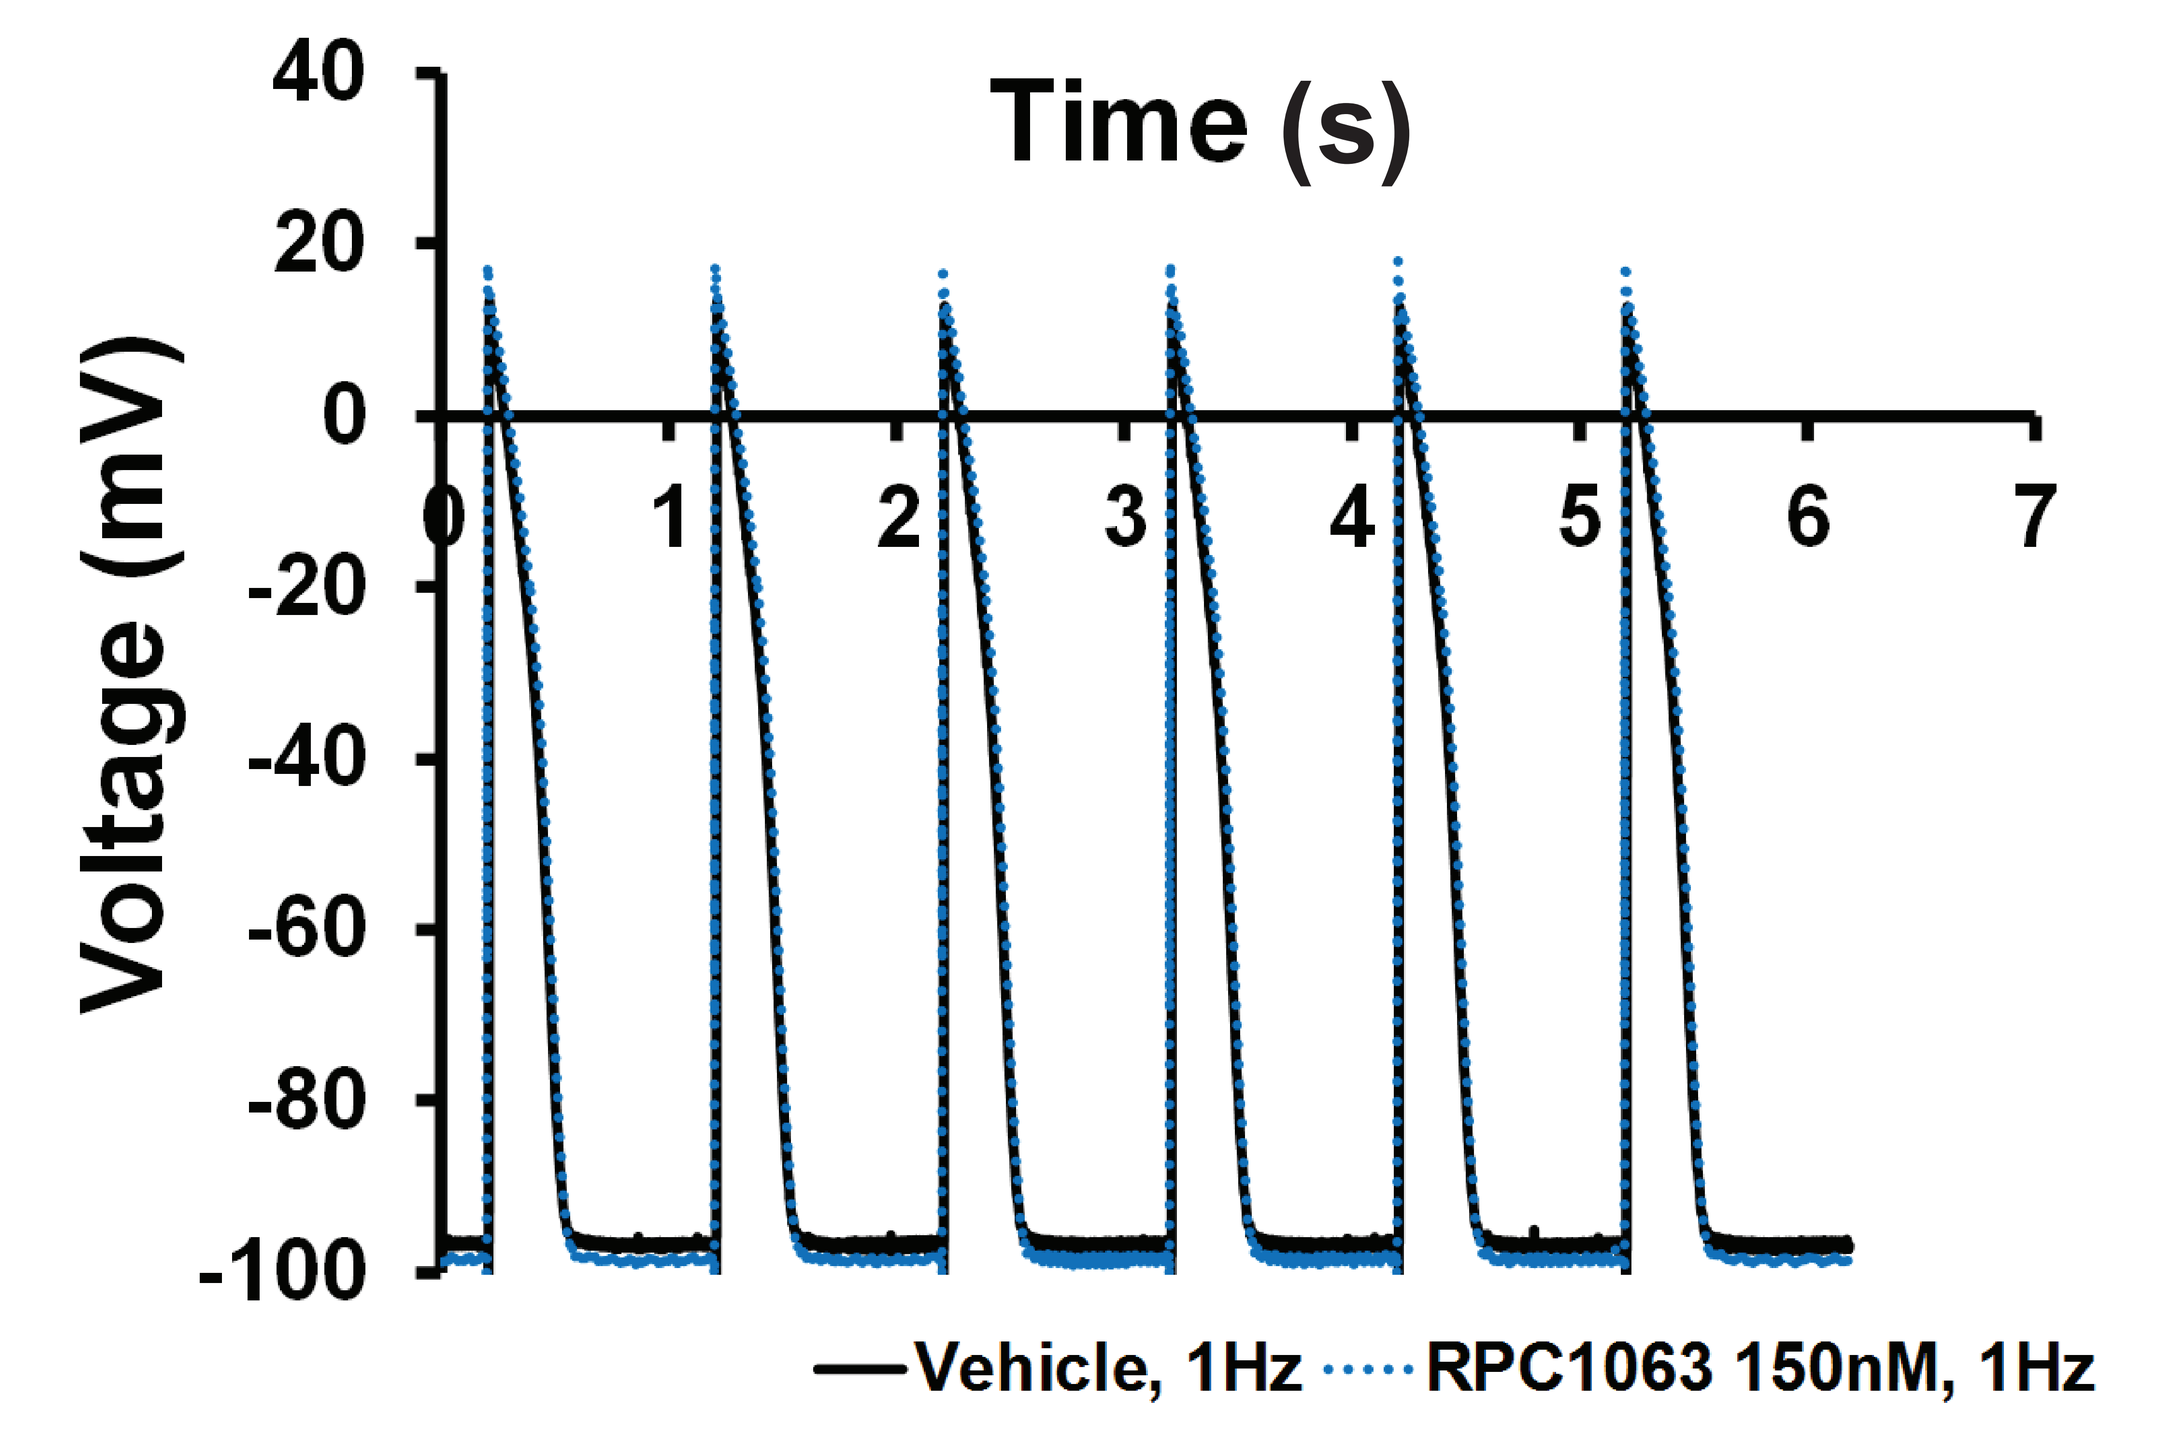
Time (s)
Voltage (mV)
Vehicle, 1Hz
RPC1063 150nM, 1Hz

BE, beat escape.

Figure 5. BE Was Present With Fingolimod

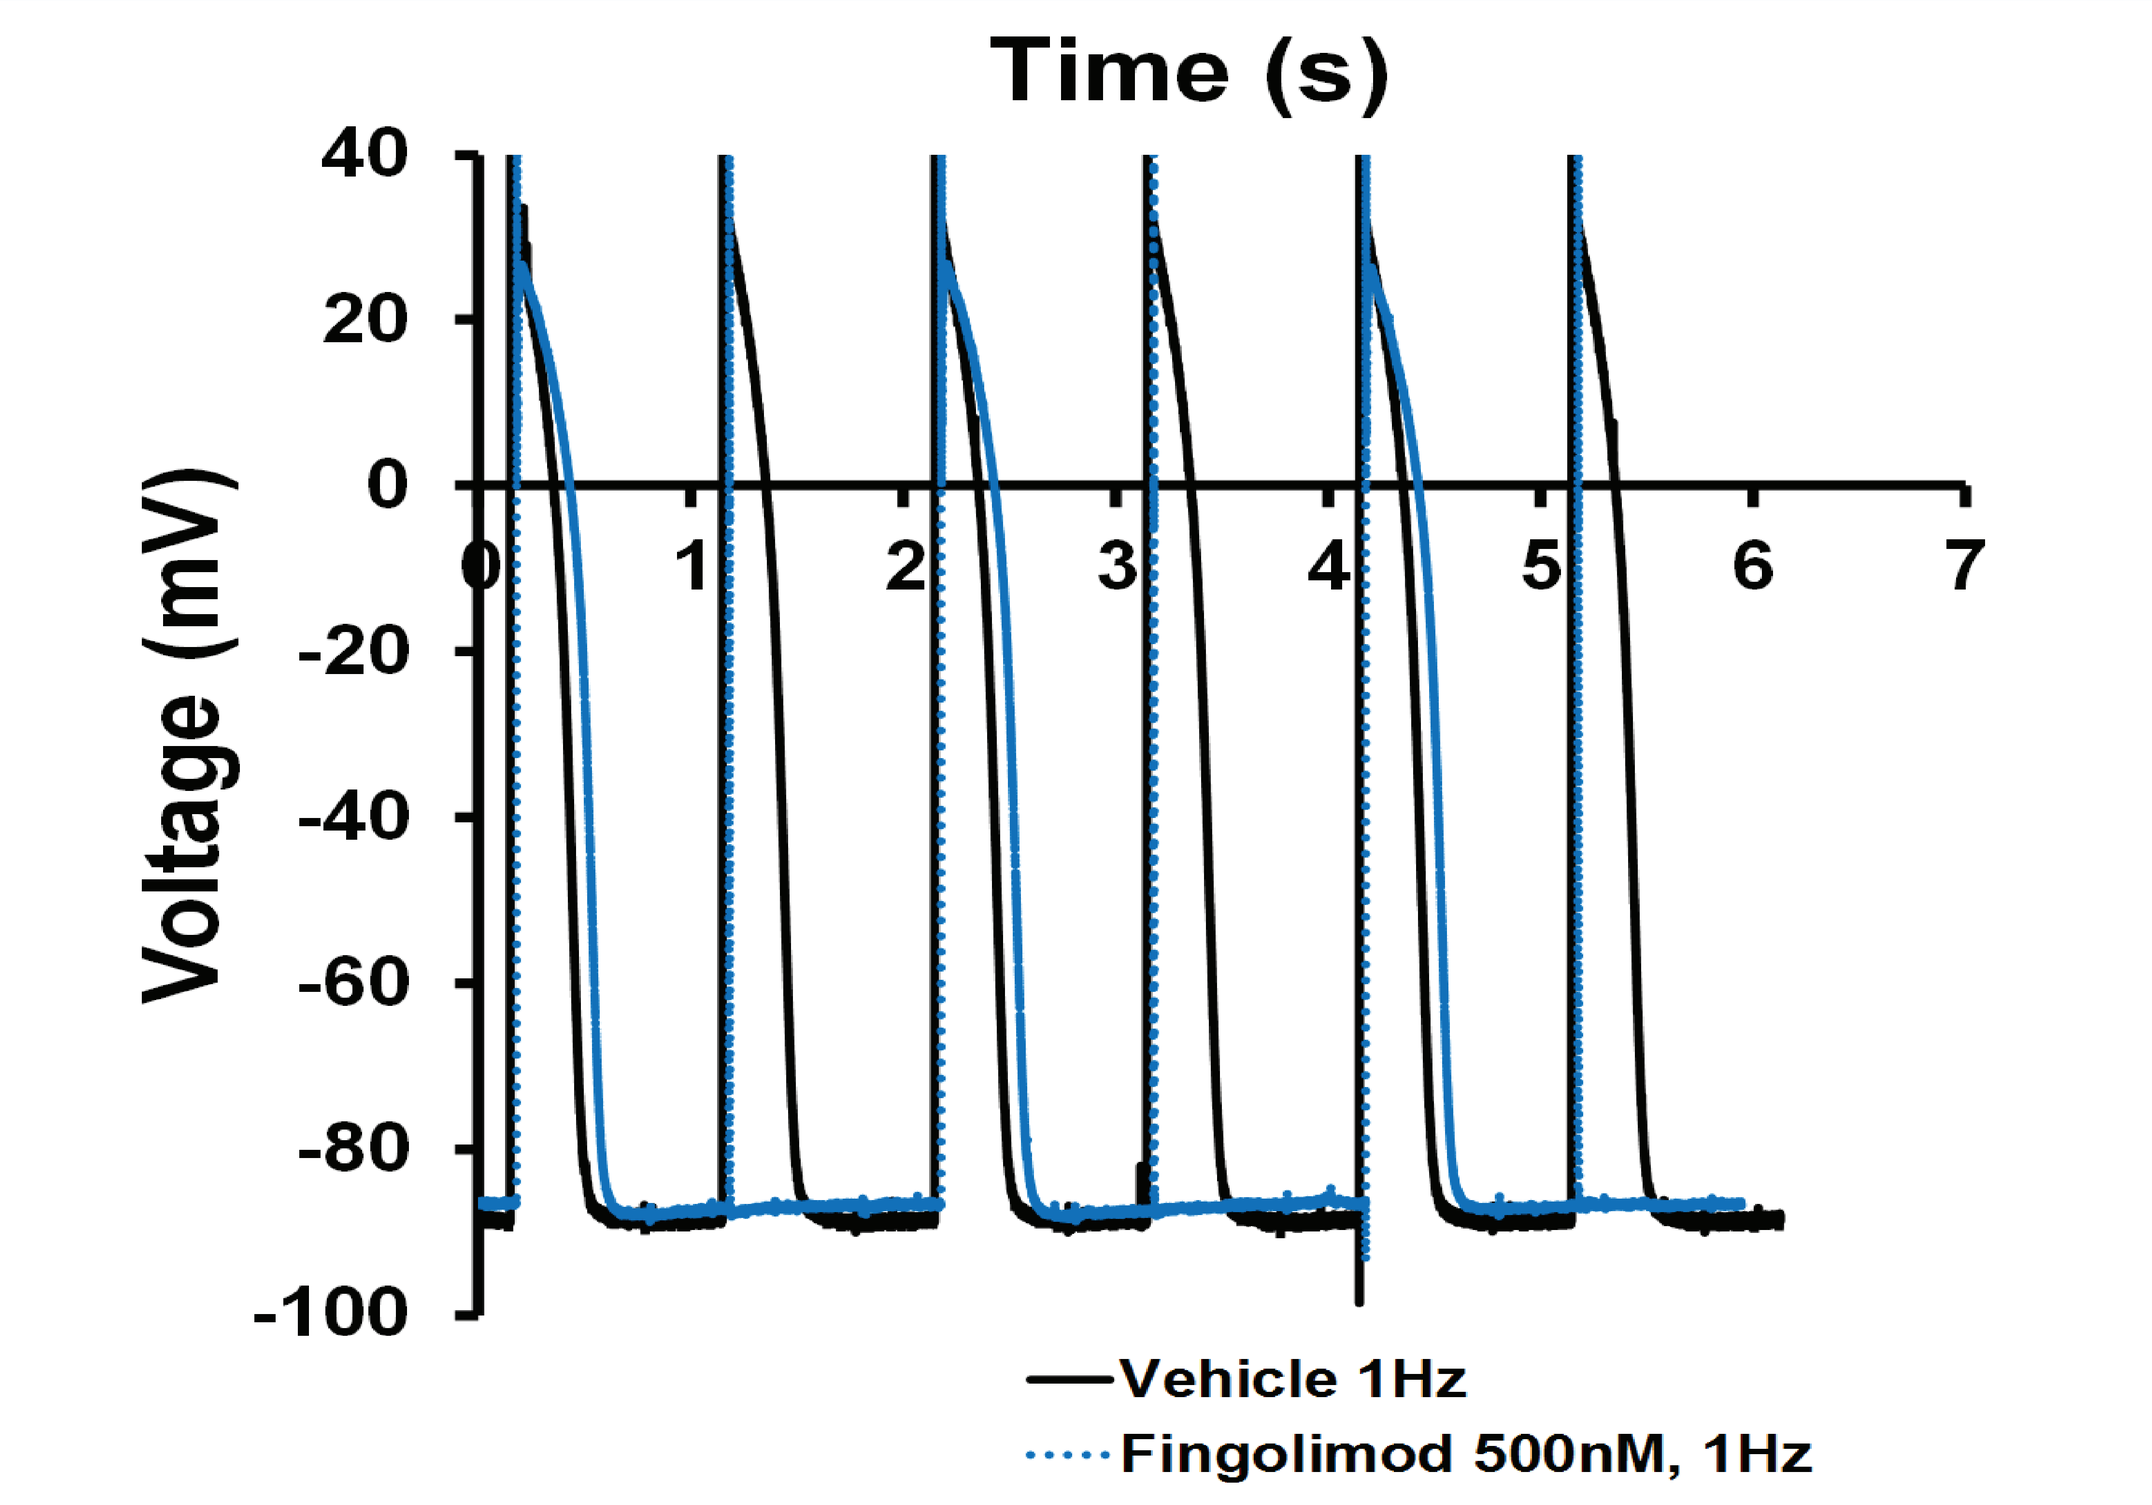
Time (s)
Voltage (mV)
Vehicle 1Hz
Fingolimod 500nM, 1Hz

BE, beat escape.

Figure 3. Ozanimod Pro-Arrhythmia and BE Assessment

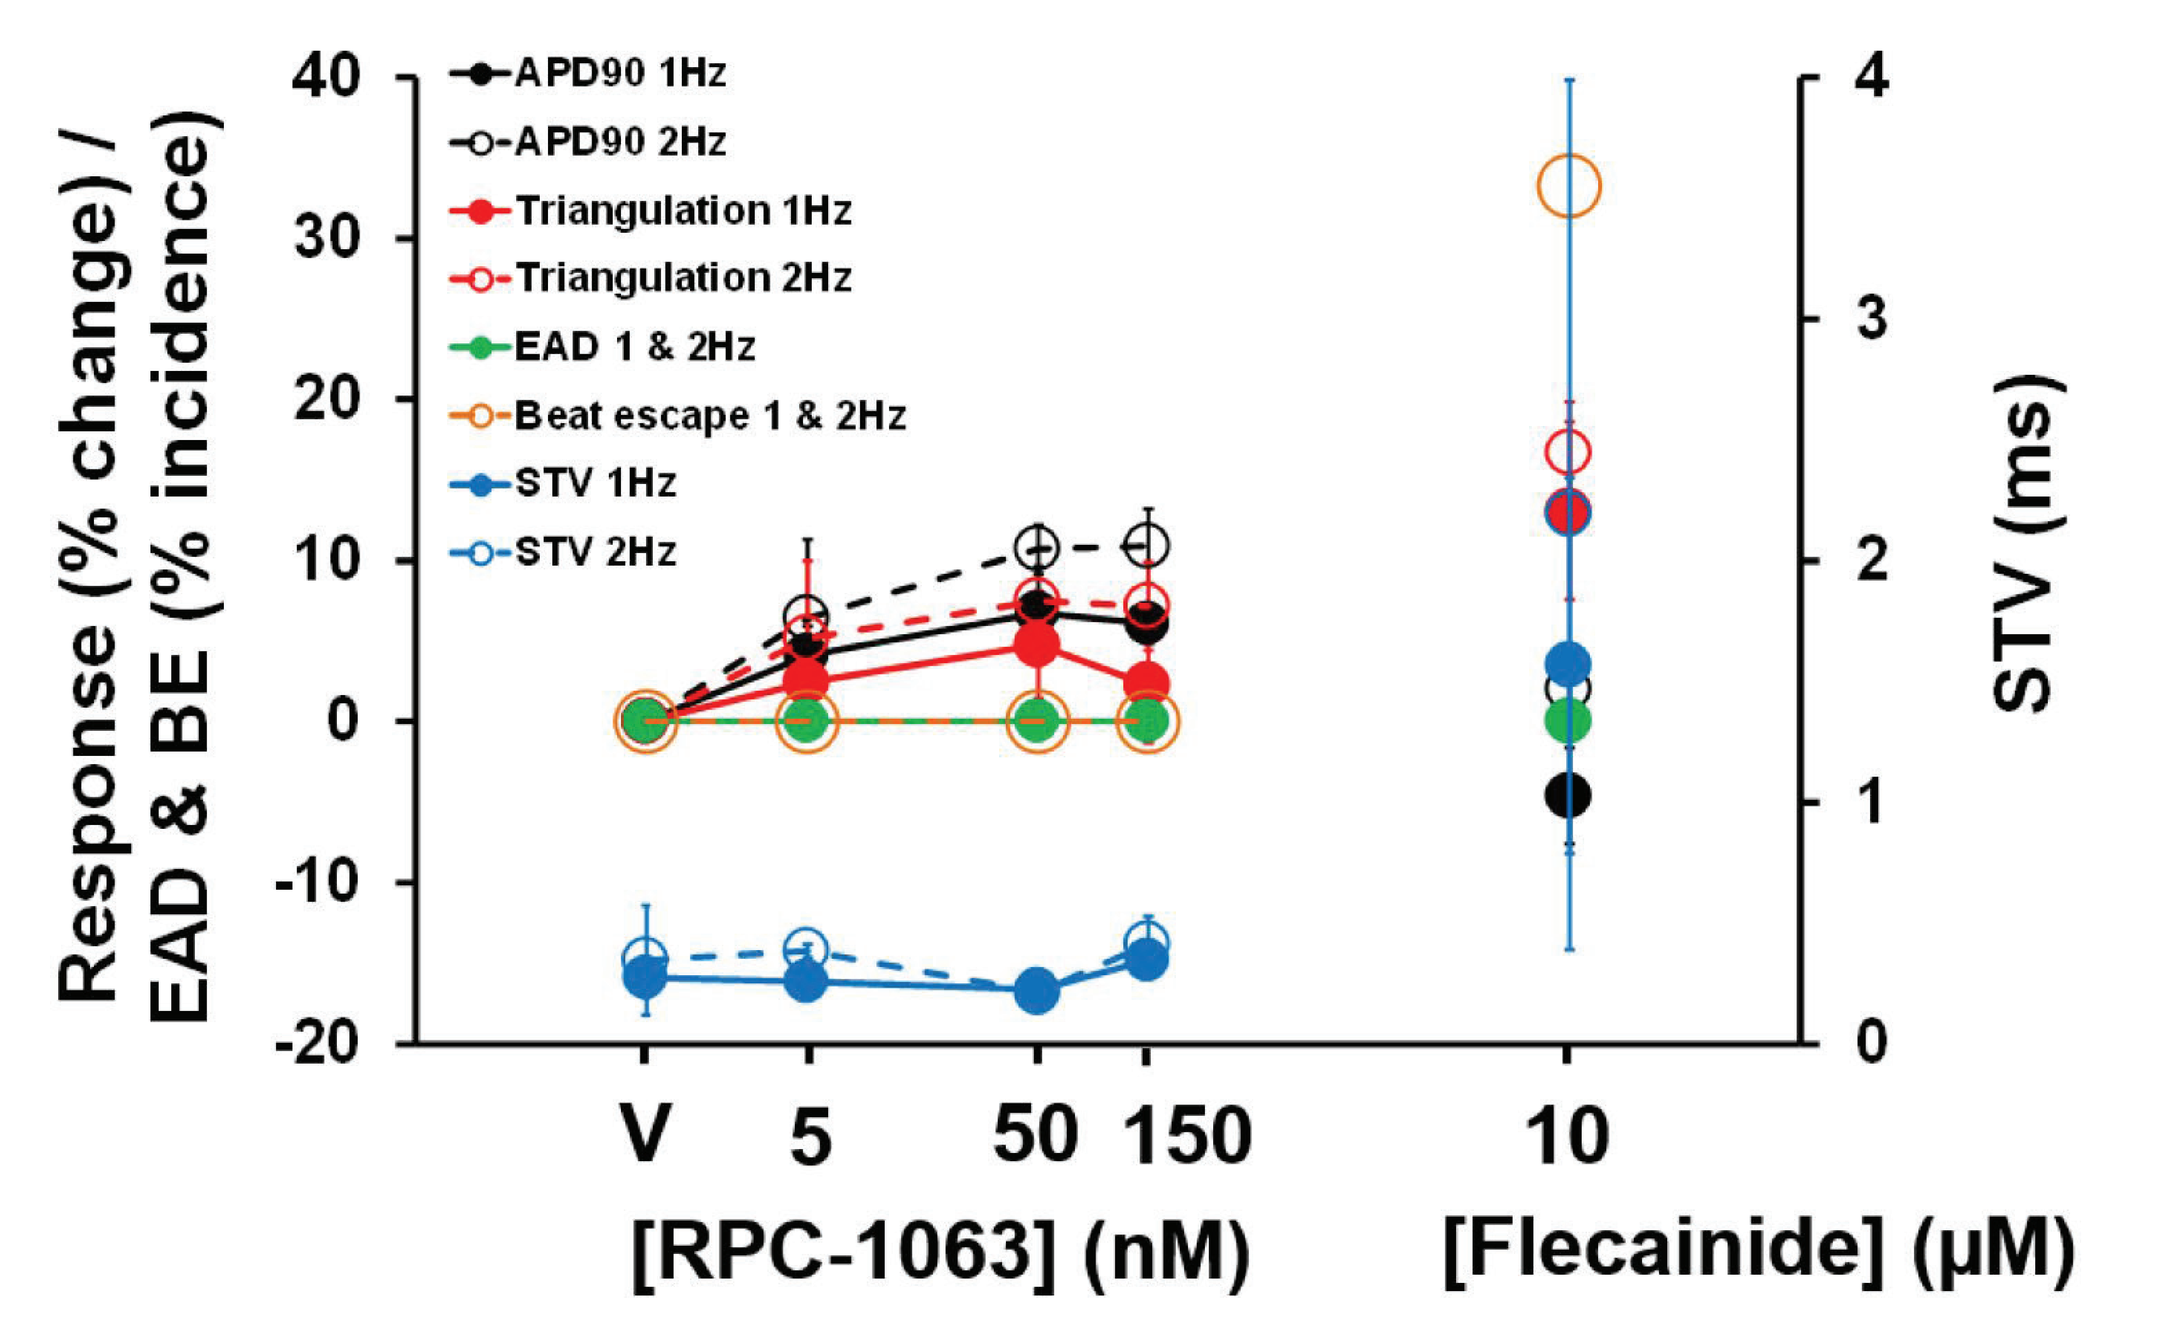
APD90 1Hz
APD90 2Hz
Triangulation 1Hz
Triangulation 2Hz
EAD 1 & 2Hz
Beat escape 1 & 2Hz
STV 1Hz
STV 2Hz
Response (% change) / EAD & BE (% incidence)
STV (ms)
V 5 50 150
10
[RPC-1063] (nM)
[Flecainide] (µM)

APD, action potential duration; BE, beat escape, EAD, early afterdepolarizations; STV, short-term variability; V, vehicle.

Figure 6. Fingolimod Pro-Arrhythmia and BE Assessment

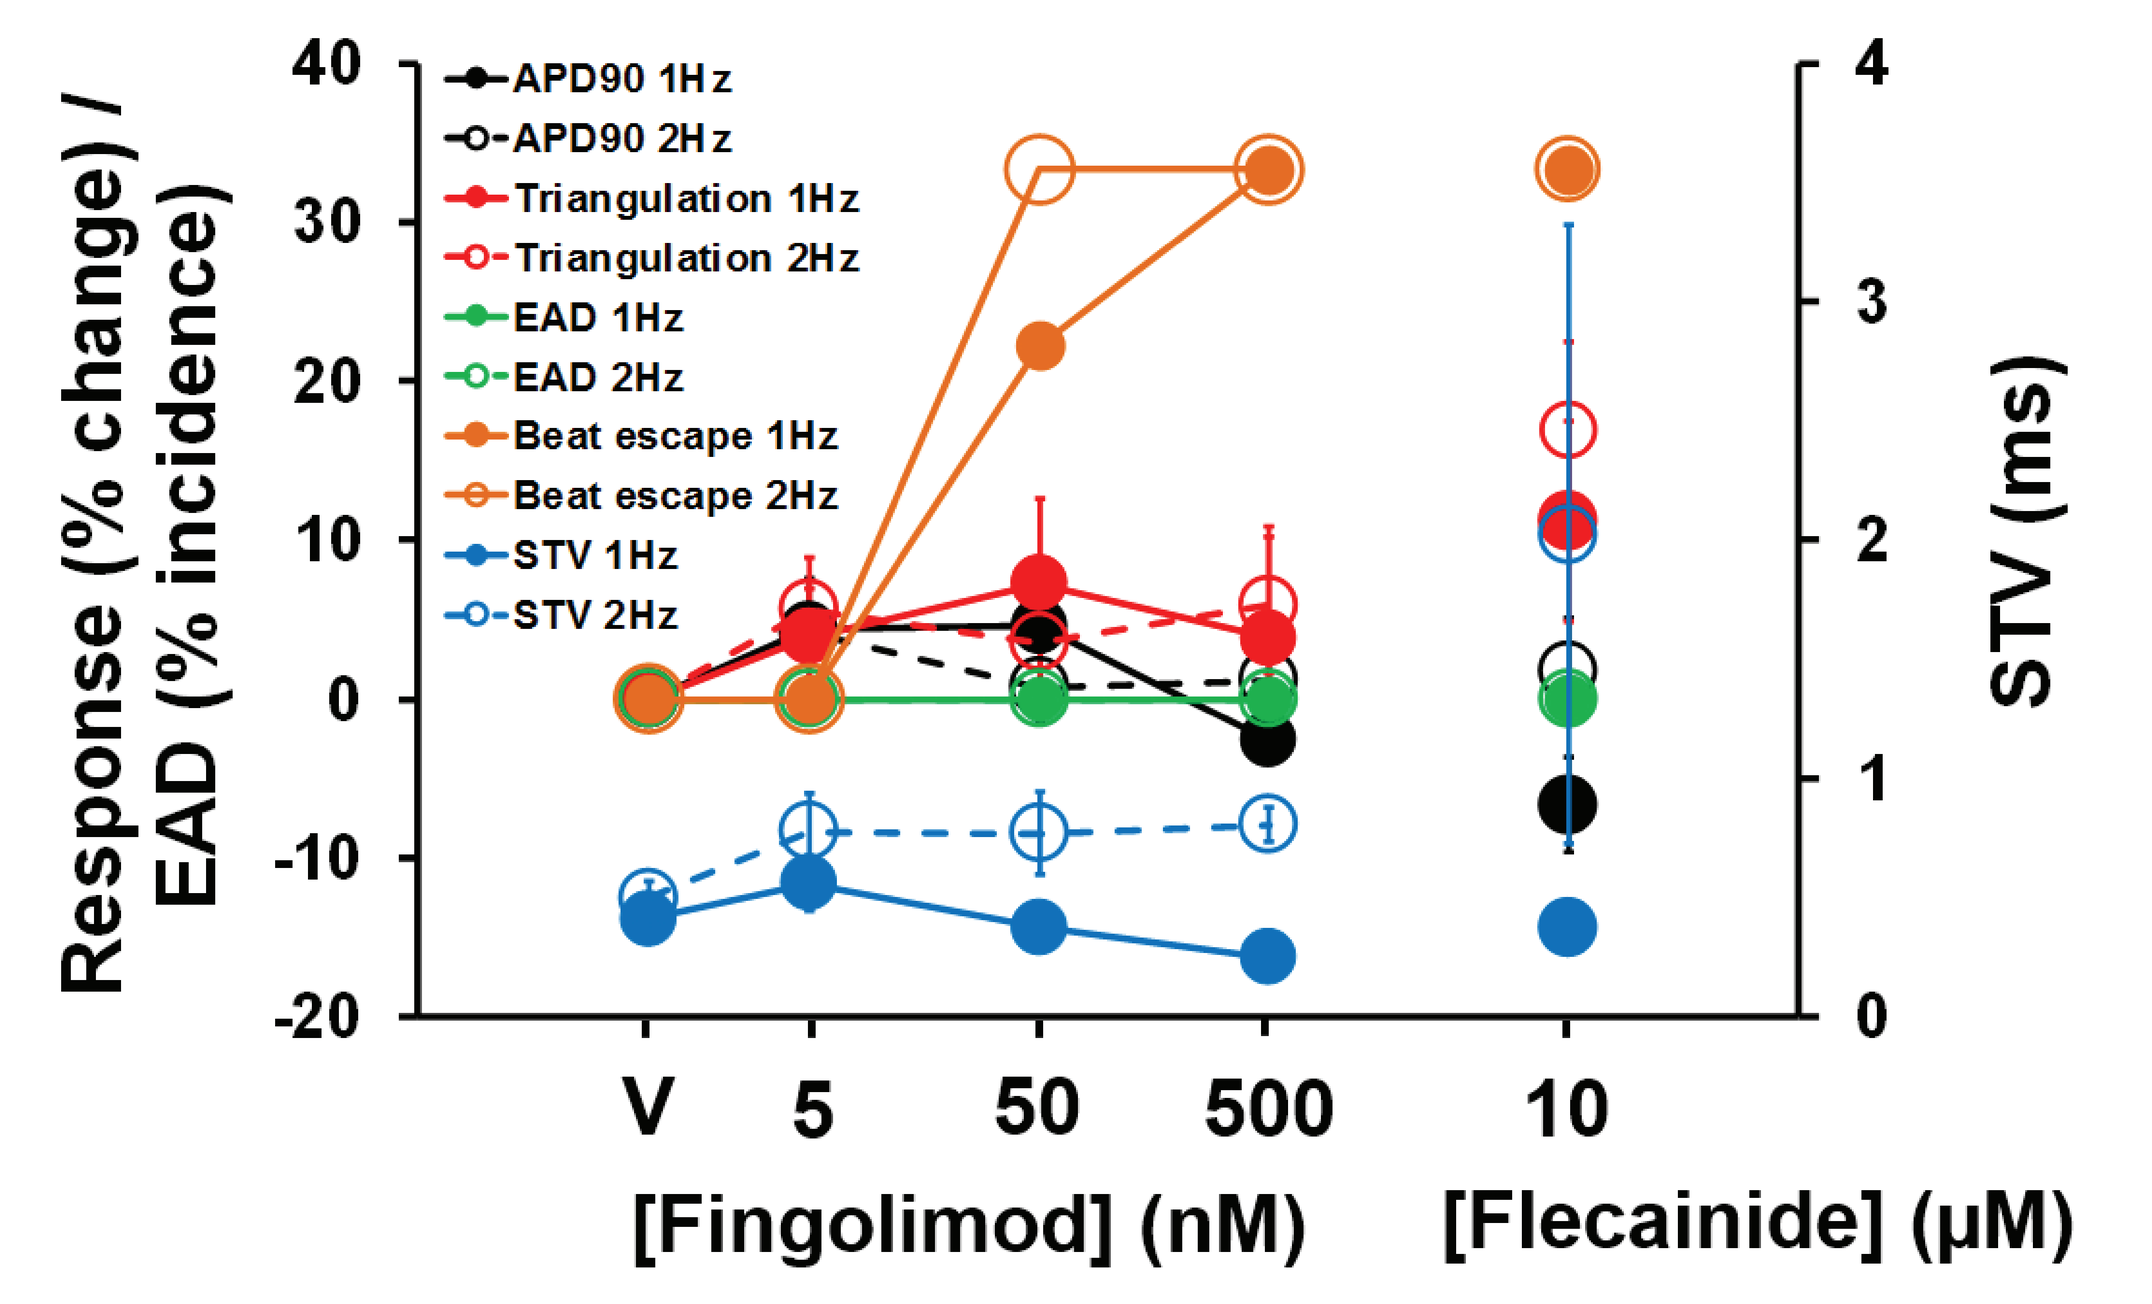
APD90 1Hz
APD90 2Hz
Triangulation 1Hz
Triangulation 2Hz
EAD 1Hz
EAD 2Hz
Beat escape 1Hz
Beat escape 2Hz
STV 1Hz
STV 2Hz
Response (% change) / EAD (% incidence)
STV (ms)
V 5 50 500
10
[Fingolimod] (nM)
[Flecainide] (µM)

APD, action potential duration; BE, beat escape, EAD, early afterdepolarizations; STV, short-term variability; V, vehicle.

Figure 4. Ozanimod Showed No Significant Effects on APD

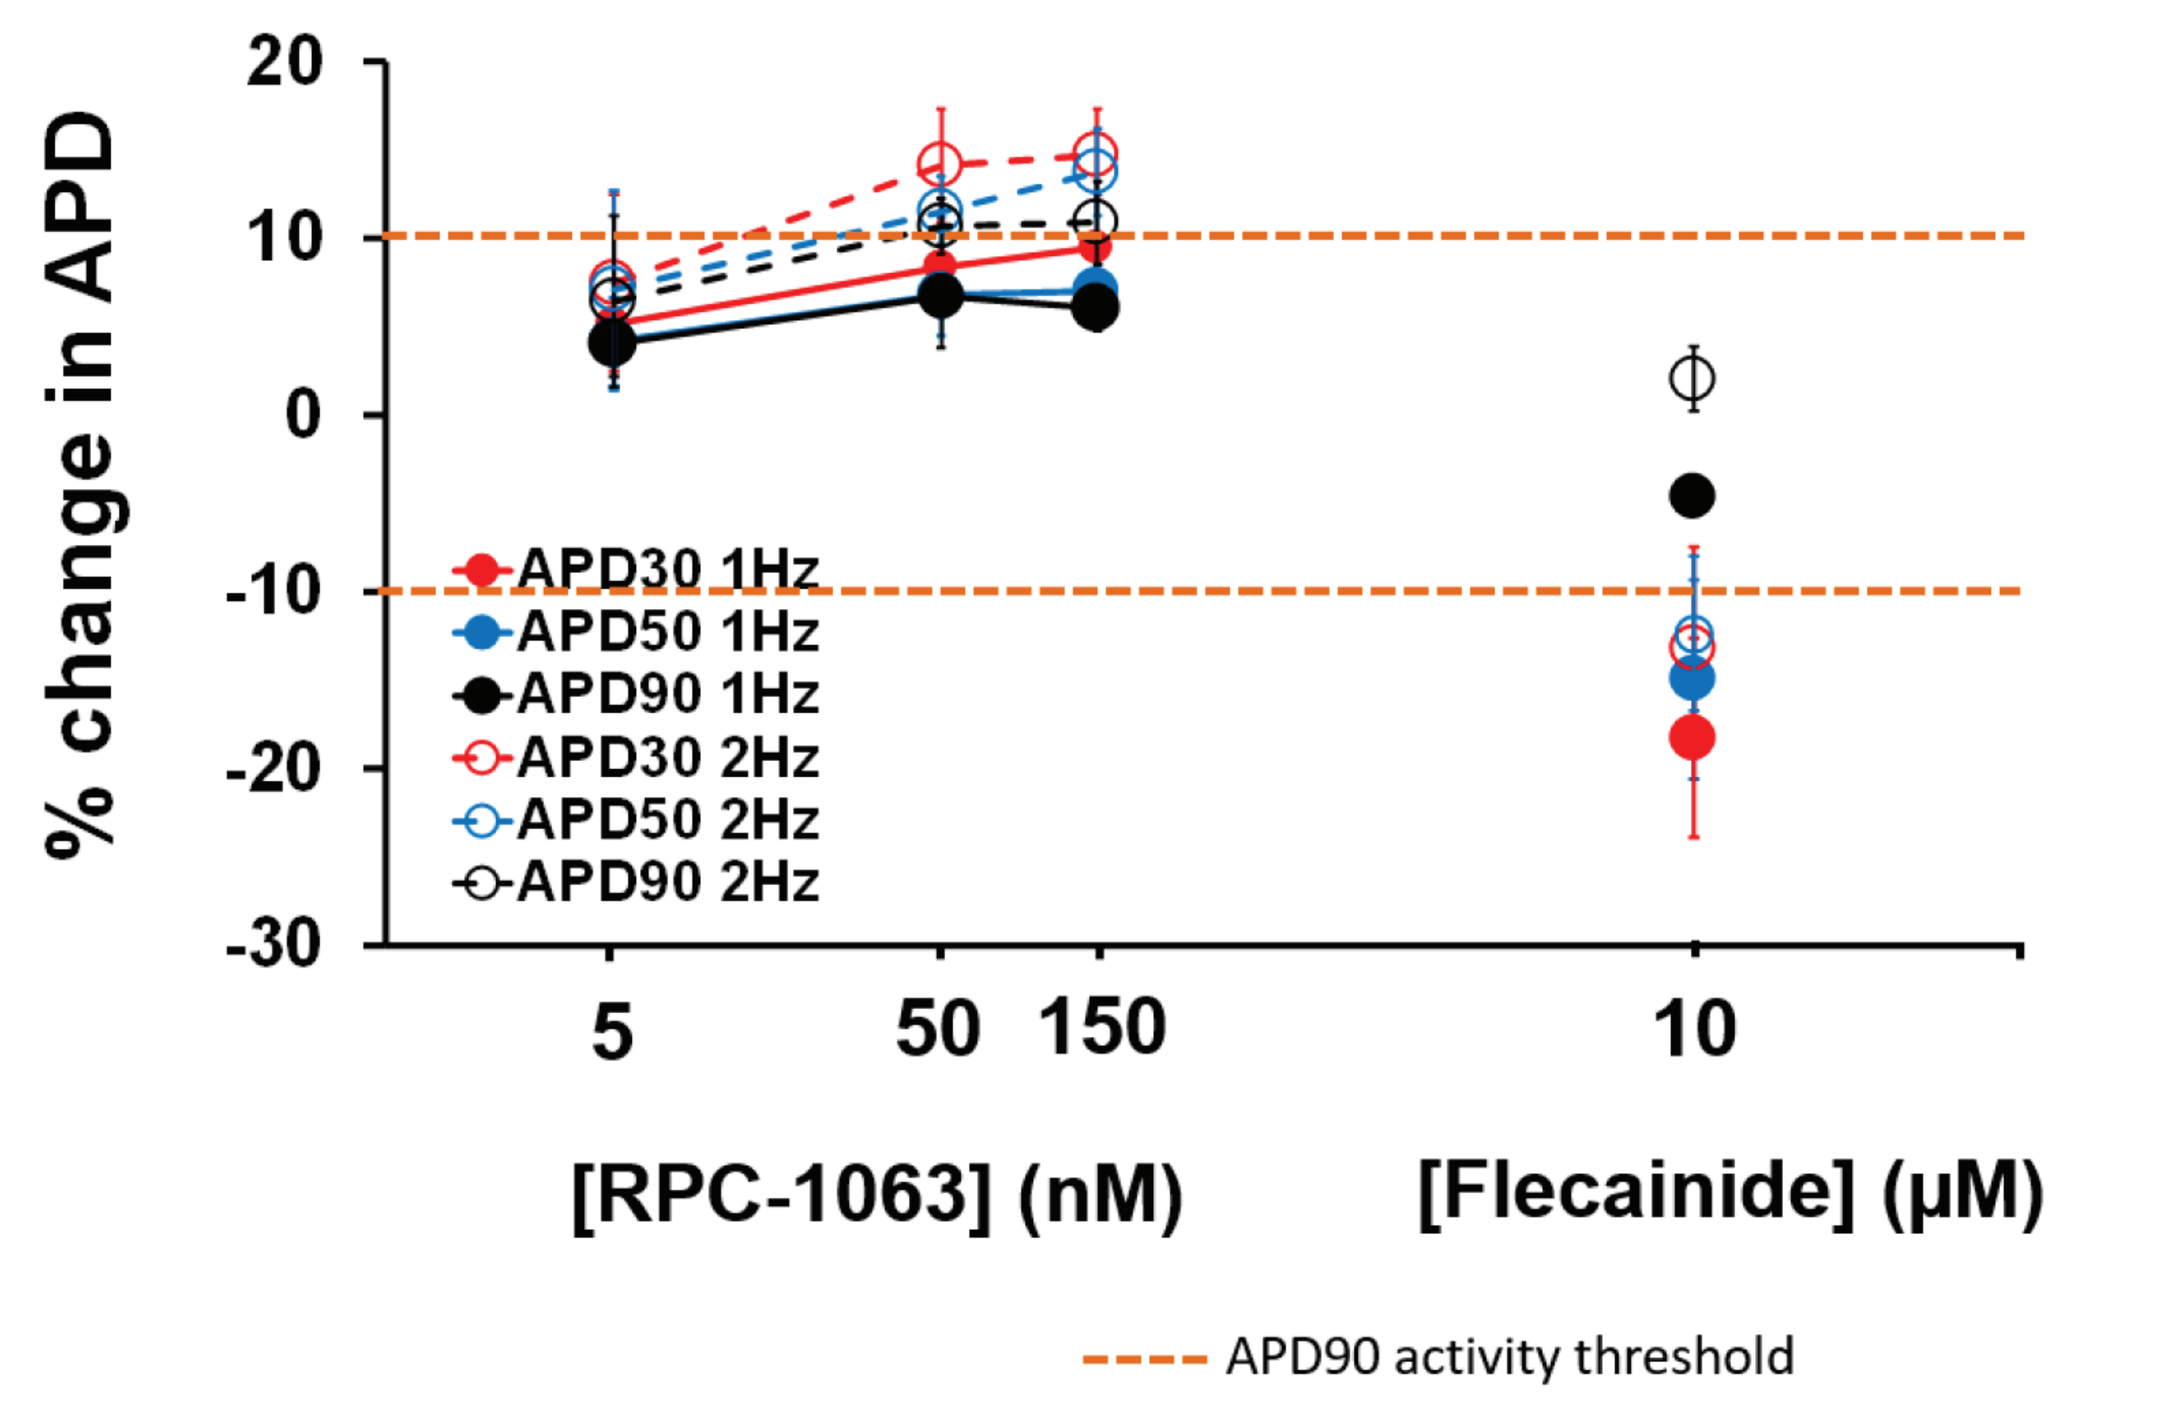
% change in APD
APD30 1Hz
APD50 1Hz
APD90 1Hz
APD30 2Hz
APD50 2Hz
APD90 2Hz
5 50 150
10
[RPC-1063] (nM)
[Flecainide] (µM)
APD90 activity threshold

APD, action potential duration.

Figure 7. Fingolimod Showed No Significant Effects on APD

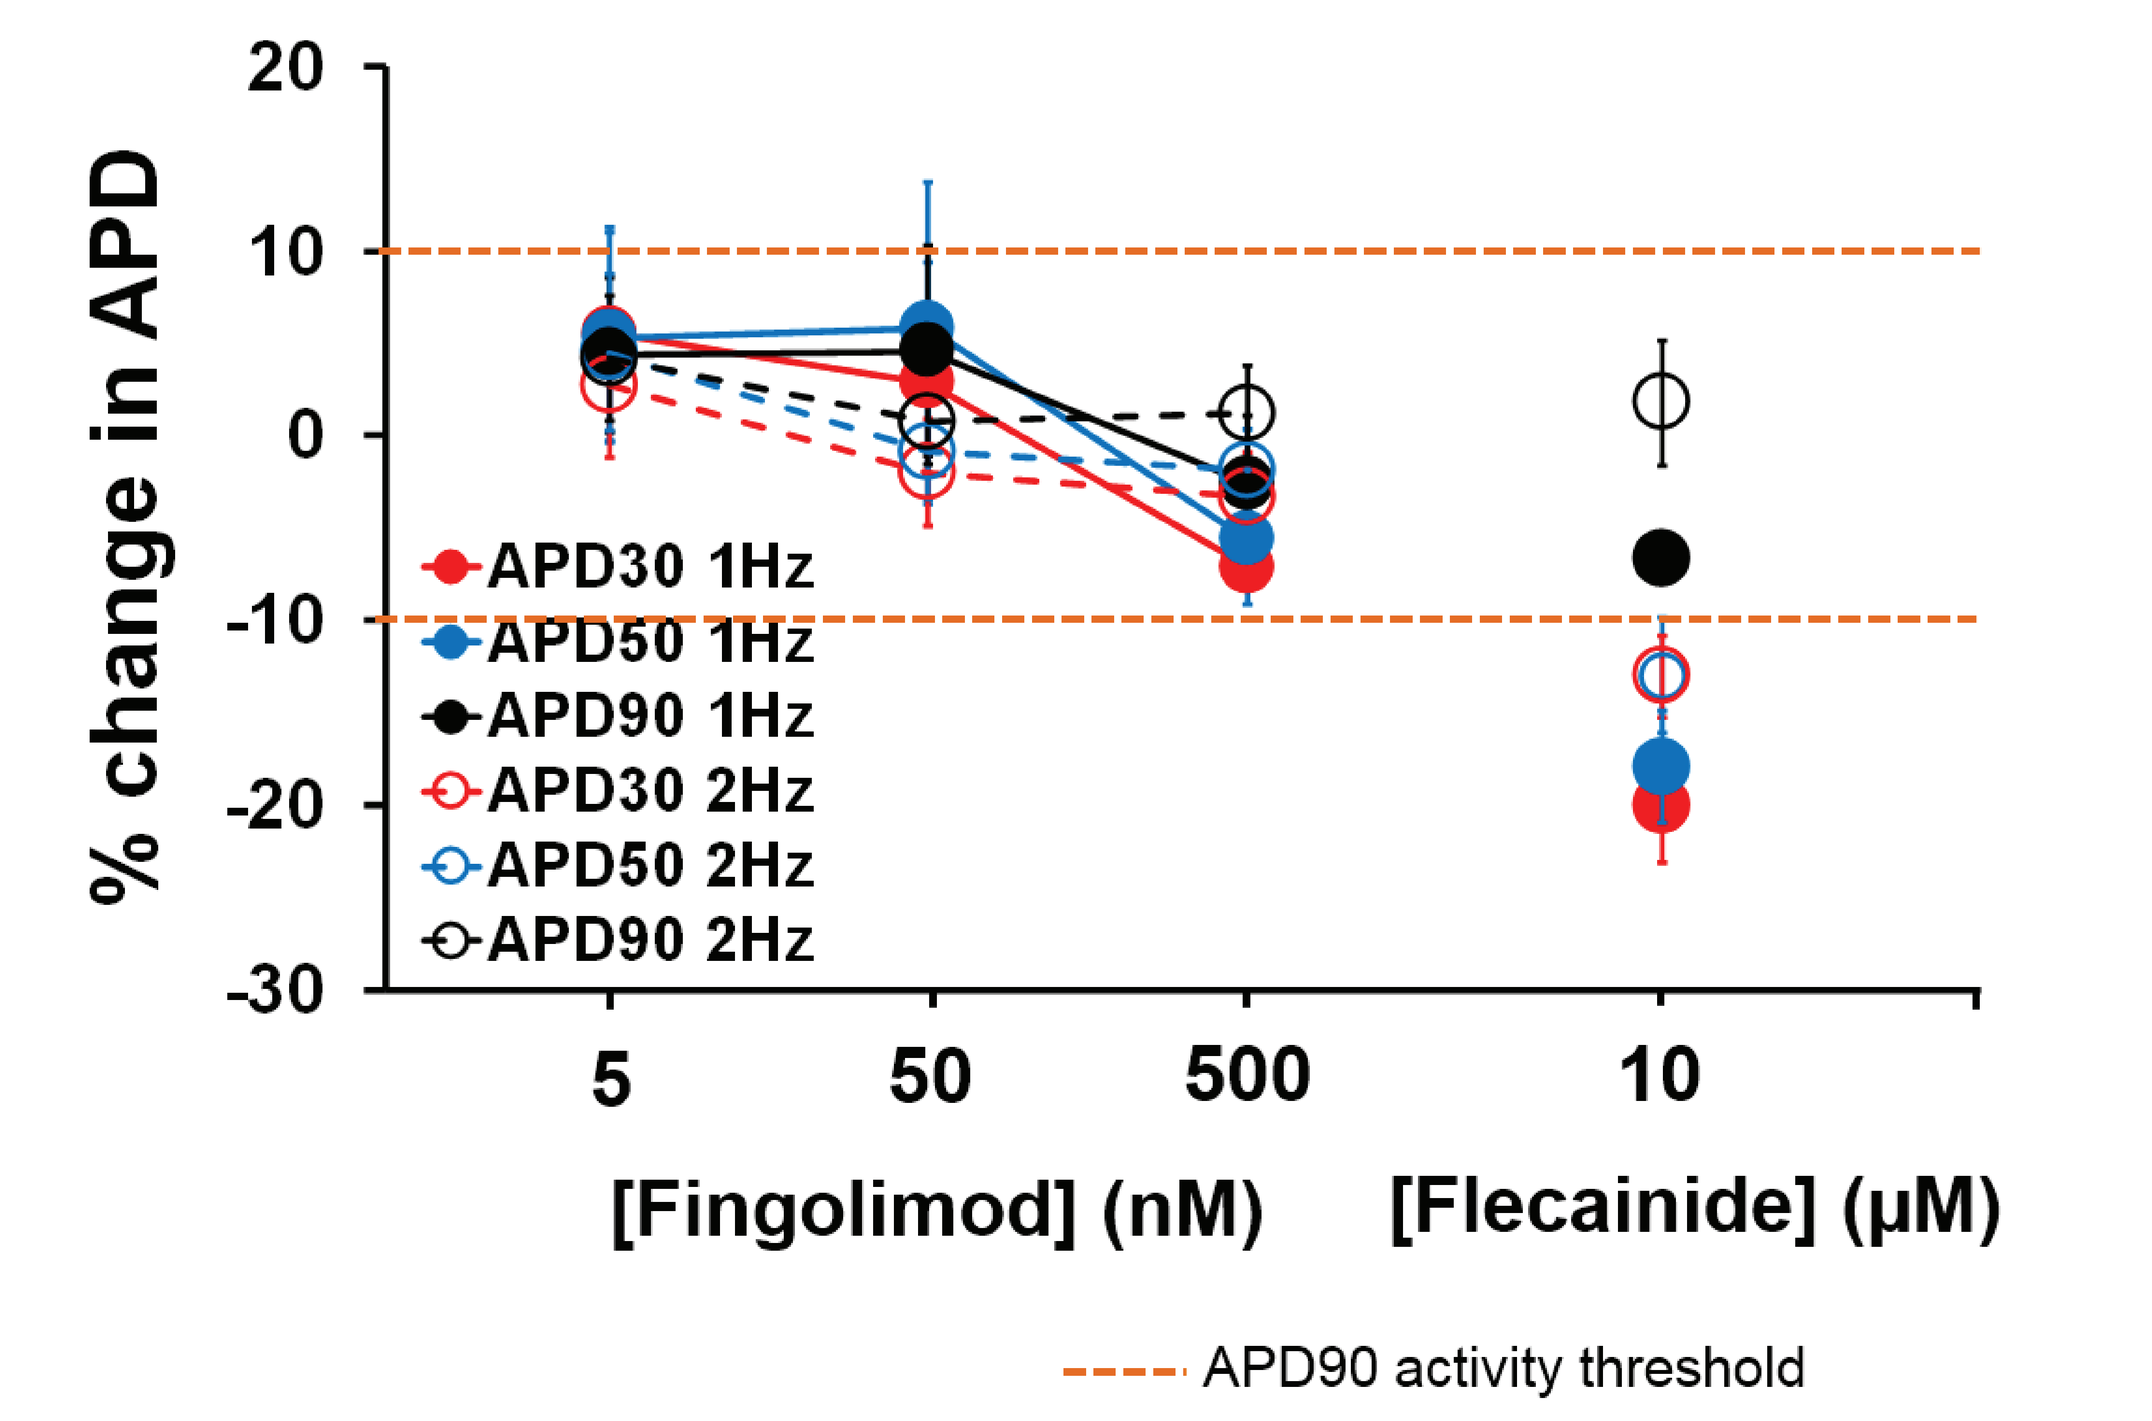
% change in APD
APD30 1Hz
APD50 1Hz
APD90 1Hz
APD30 2Hz
APD50 2Hz
APD90 2Hz
5 50 500
10
[Fingolimod] (nM)
[Flecainide] (µM)
APD90 activity threshold

APD, action potential duration.

LITERATURE

1. Gilenya® (fingolimod) capsules, for oral use [Prescribing Information]. East Hanover, NJ: Novartis Pharmaceuticals Corporation; 2016.
2. Comi G, et al. ECTRIMS 2017 Oral Presentation #232.
3. Cohen JA, et al. ECTRIMS 2017 Oral Presentation #280.
4. Cohen JA, et al. *Lancet Neurol*. 2016;15:373–81.
5. Sandborn WJ, et al. *N Engl J Med*. 2016;374:1754–62.
6. Page G, et al. *J Pharmacol Toxicol Methods*. 2016;81:183–95.

DISCLOSURES

N Abi-Gerges, PE Miller, A Ghetti, G Page: Financial support for research, Celgene Corporation.

FL Scott, KR Taylor Meadows, B Clemons, P Frohna, GJ Opiteck: Shareholder, Celgene Corporation.

ACKNOWLEDGMENTS

We thank Phachareeya Ratchada, Ashley Alamillo, Katrina Sweat, Yannick Miron, and Ky Truong (AnaBios) for technical assistance. Support for third-party editorial assistance for this poster was provided by Jamie Weaver, PhD, of CodonMedical, an Ashfield Company, part of UDG Healthcare plc, and was funded by Celgene.

Copies of this poster obtained through the QR code are for personal use only and may not be reproduced without permission from the author of this poster.

Najah Abi-Gerges
najah.abigerges@anabios.com

Celgene AnaBios Early Human Insights

Presented at the European Charcot Foundation 25th Annual Meeting; November 30–December 2, 2017; Baveno, Italy