Abstract No. 63

Rapid reduction of lesion accumulation in specific white matter tracts as assessed by lesion mapping in patients with clinically isolated syndrome

Antonio Giorgio[1], Marco Battaglini[1], Giordano Gentile[1], Maria Laura Stromillo[1], Andrea Visconti[2], Nicola De Stefano[1]

[1]Department of Medicine, Surgery and Neuroscience, University of Siena, Siena, Italy; [2]Medical Affairs Department, Merck Serono Italy

INTRODUCTION AND PURPOSE

- Administration of interferon (IFN) beta-1a in patients with relapsing remitting (RR) MS reduces brain lesion accumulation over time, as assessed by magnetic resonance imaging (MRI). Our study aims to identify treatment-specific spatio-temporal areas using lesion probability mapping (LPM).

METHODS

- We performed a post-hoc analysis of MRI data in RRMS patients from the IMPROVE study, a randomized (2:1) clinical study (ClinicalTrials.gov identifier NCT00441103) comparing patients treated with IFN beta-1a 44 mcg given subcutaneously three times per week (n=120) versus placebo (n=60).

Analysis of white matter (WM) lesion spatial distribution

- We used MRI images acquired at weeks 4, 8, 12 and 16 to create LPMs of the cumulative combined unique active (CUA) lesions in each patient group.
- At each time point, differences in lesion spatial distribution between treated and placebo groups were assessed along several white matter (WM) tracts by using pre-defined anatomic WM atlases.

Analysis of WM lesion frequency

- Differences in lesion frequency were assessed with a voxelwise comparison, using FMRIB software Library (FSL) randomisation, between treated and placebo groups within the general linear model framework and using nonparametric permutations ($p<0.05$, cluster-corrected).

RESULTS

WM lesion distribution analysis

- The treated group presented 50% reduction in CUA lesions compared to the Placebo group, these differences were observed as early as week 4 and were sustained through week 16 (Treated group: 41 cm^3 at week 4; 95 cm^3 at week 16; mean: 24 cm^3/month; Placebo group 62 cm^3 at week 4; 196 cm3 at week 16; mean: 48 cm^3/month) (**Fig. 1**).

Fig. 1. Follow-up LPM (week 4-16) of the spatial distribution of cumulative CUA lesions in the Placebo and Treated groups. The intensity corresponds to the probability, for that voxel, that a new lesion occurs.

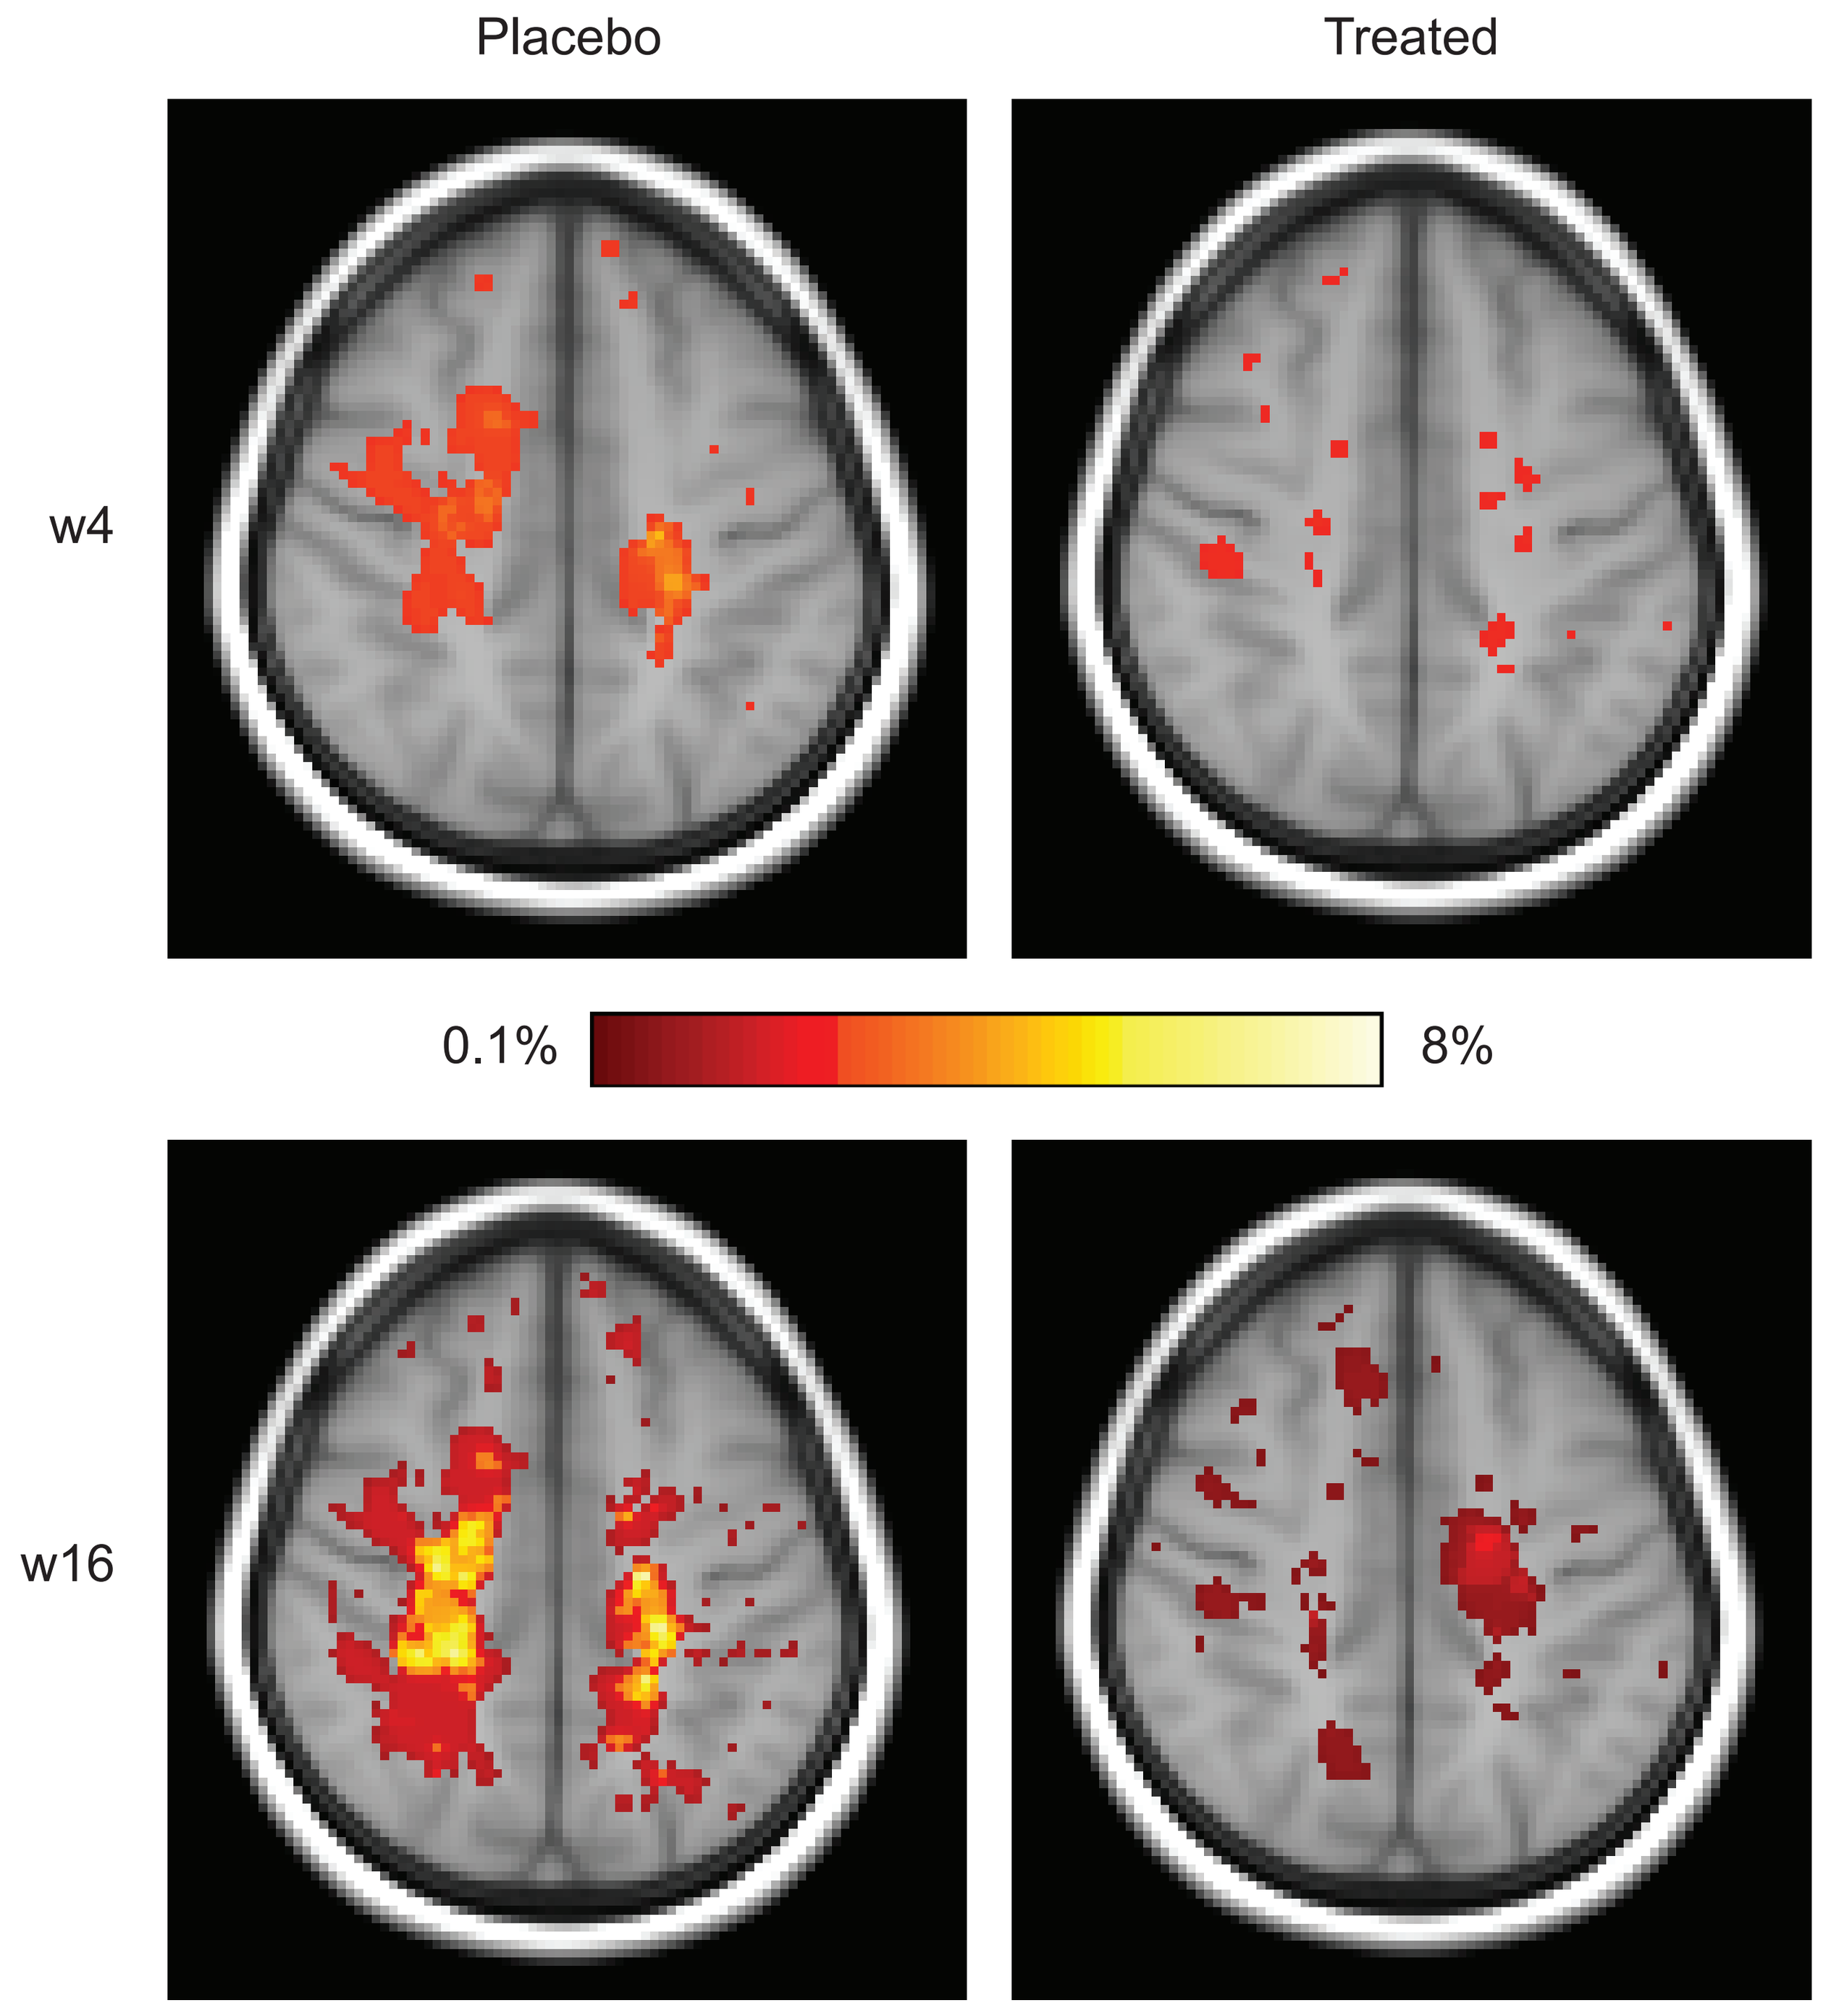
Placebo
Treated
w4
0.1%
8%
w16

- Similar results were obtained with the WM tract analysis, with a significant reduction of lesion accumulation in the treated group in the order of 50% in the corticospinal tract (CST), 52% in the left anterior thalamic radiation (ATR) and 65% in the superior longitudinal fascicle (SLF) (**Fig. 2**).

WM lesion frequency analysis

- On voxelwise analysis, LPM of the Treated group showed lower frequency of CUA lesions than the Placebo group since week 4. This became particularly pronounced at week 16 in the left CST ($p<0.005$), ATR ($p<0.005$) and SLF ($p<0.02$) (**Fig. 3**).

Fig. 2. Follow-up LPM (week 4-16) of the spatial distribution of cumulative CUA lesions at the level of WM tracts in the Placebo and Treated groups. The intensity corresponds to the probability, for that voxel, that a new lesion occurs.

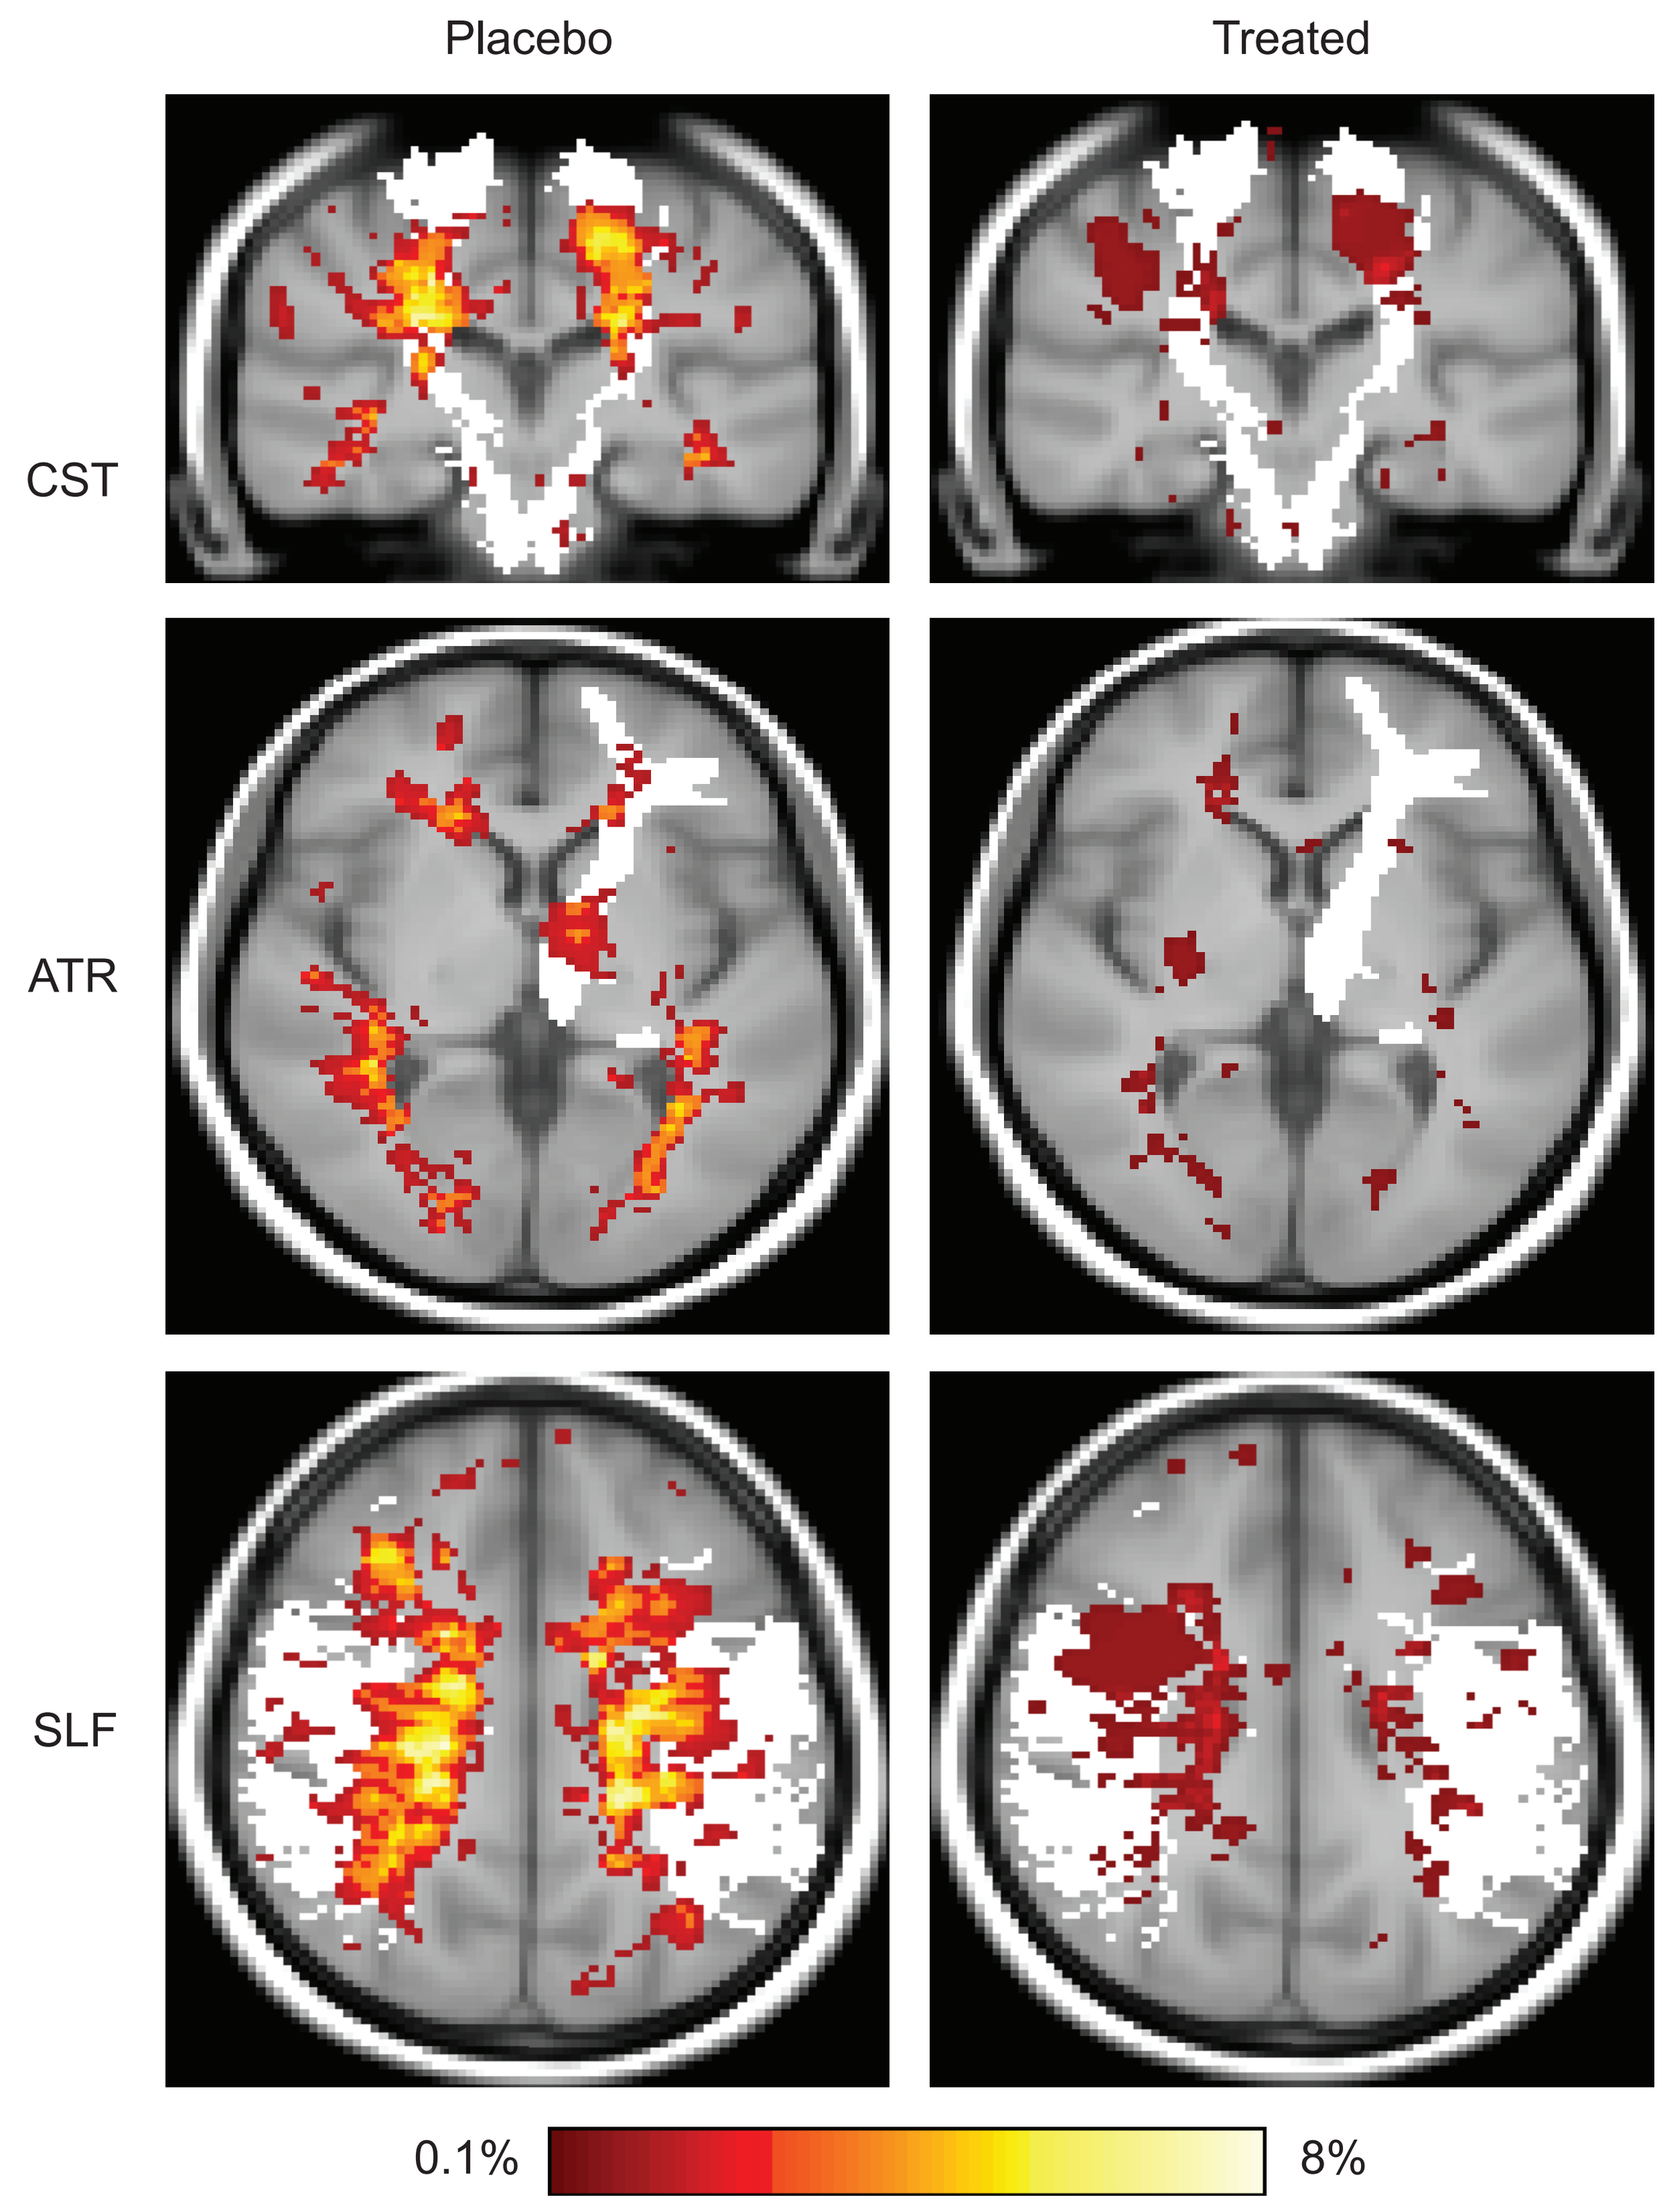
Placebo
Treated
CST
ATR
SLF
0.1%
8%

Fig. 3. LPM showing clusters of significant lower frequency of CUA lesion occurrence in Treated with respect to Placebo at week 16. At each voxel, the color intensity corresponds to the p-value.

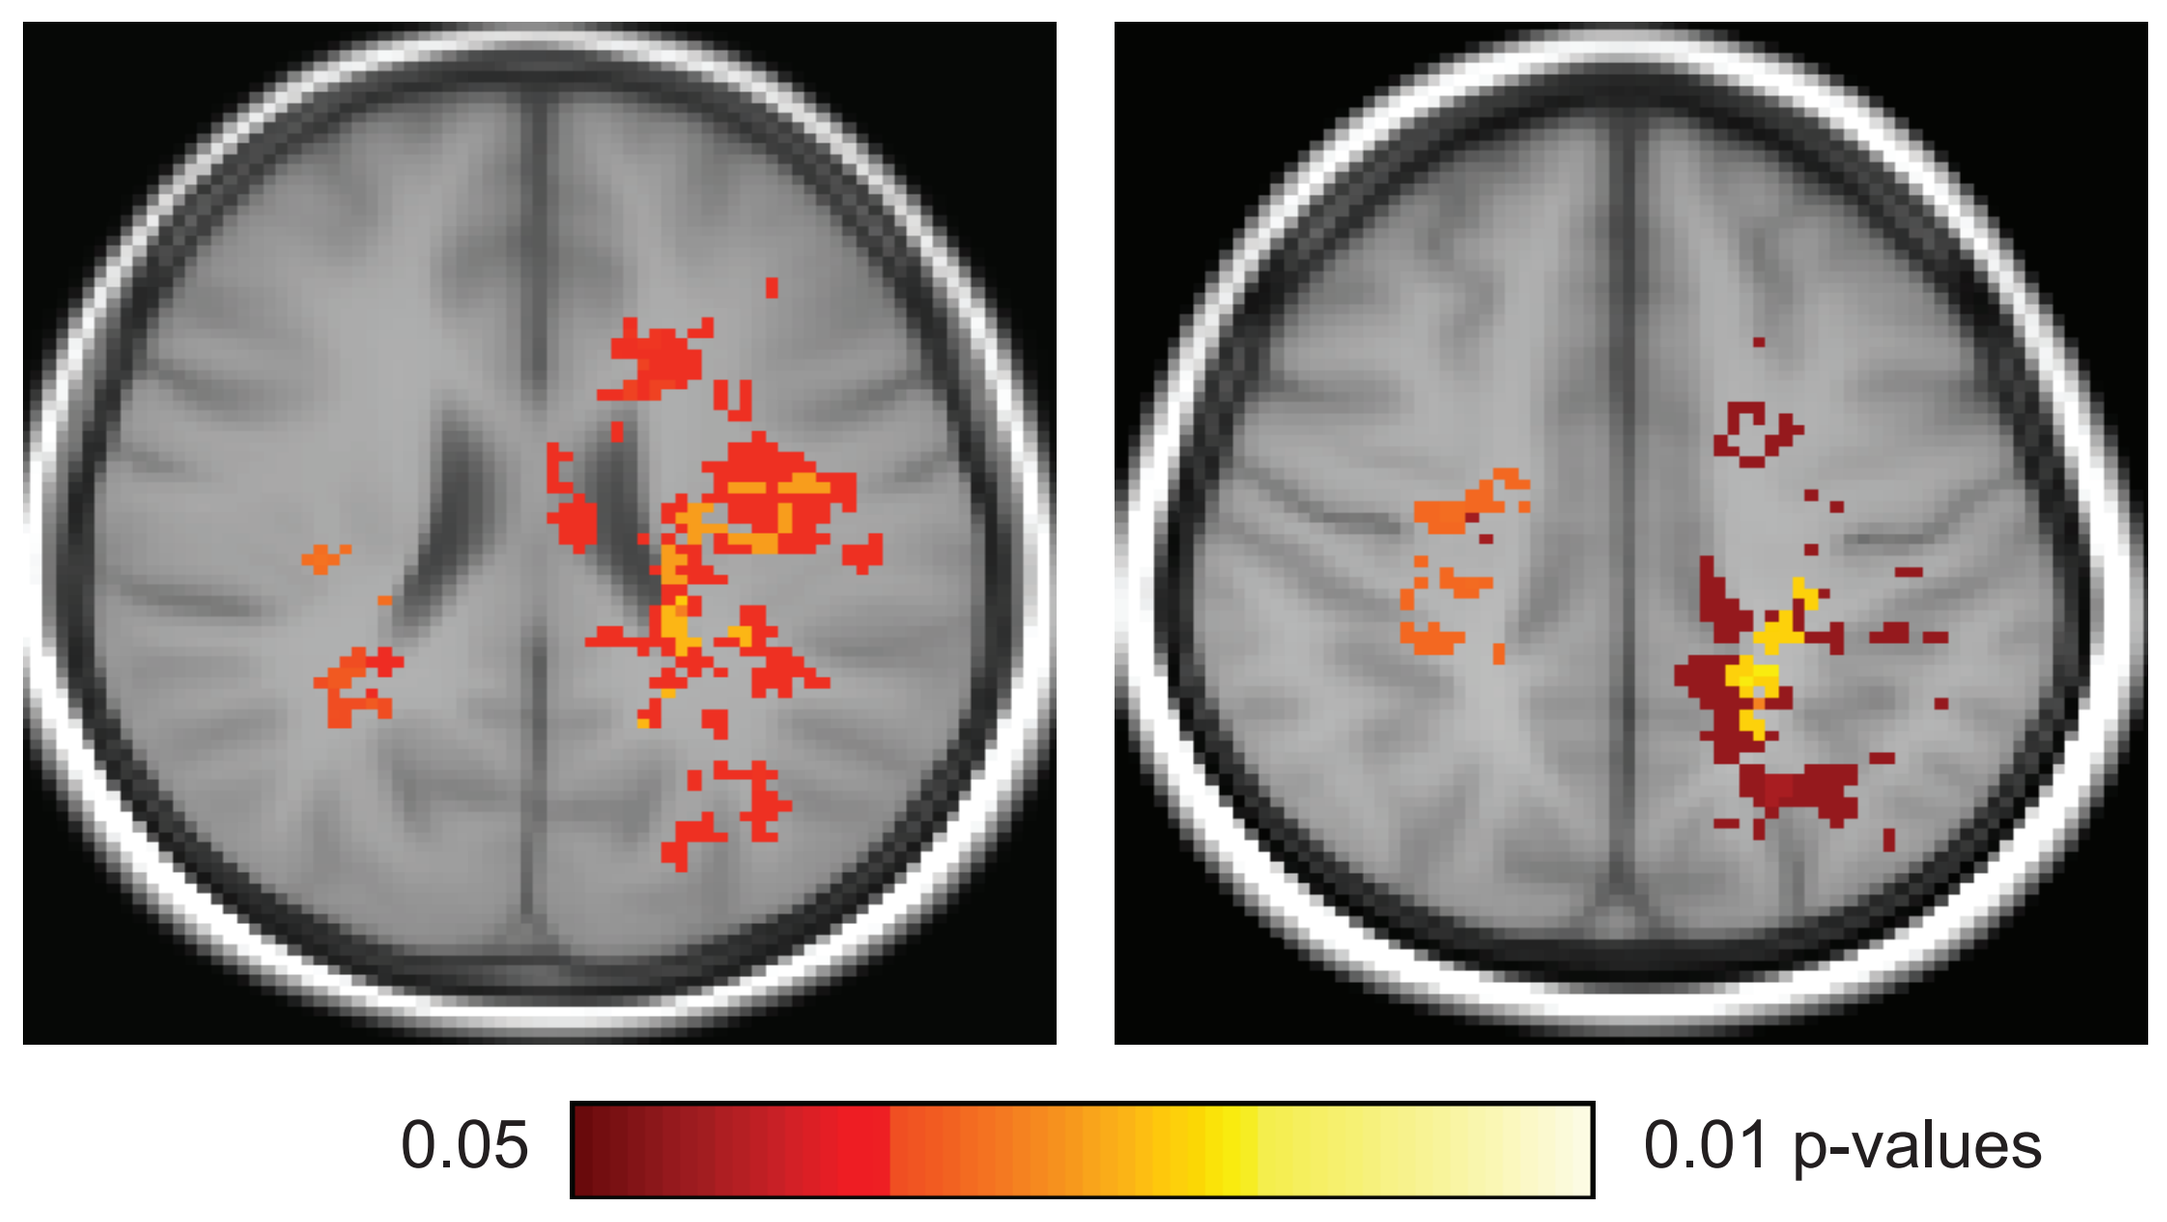
0.05
0.01 p-values

DISCLOSURES

A. Giorgio, M. Battaglini, G. Gentile, M. L. Stromillo and A. Visconti have nothing to disclose. N De Stefano has received honoraria from Biogen-Idec, Genzyme, Merck Serono, Novartis, Roche and Teva for consulting services, speaking and travel support. He serves on advisory boards for Merck Serono, Novartis, Biogen-Idec, Roche, and Genzyme, he has received research grant support from the Italian MS Society

ACKNOWLEDGEMENTS

This study was sponsored by Merck Serono S.p.A., Rome, Italy, an affiliate of Merck KGaA, Darmstadt, Germany.

CONCLUSIONS

- **Treatment with IFN beta-1a, in comparison to placebo, rapidly reduces lesion accumulation in RRMS patients in WM tracts, reaching after 16 weeks the highest local differences in clinically eloquent WM areas such as CST, SLF and ATR.**

Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors.

GET POSTER PDF

Presented at European Charcot Foundation (ECF) 2018; 15-17 November; Baveno, Italy.