72

Ozanimod (RPC1063) Reduces the Plasma Biomarker Neurofilament Light Chain in Preclinical Rodent Models of Multiple Sclerosis

Kristen R. Taylor Meadows, Fiona L. Scott, Christiane Villescaz, Bryan Clemons, Carlos Lopez, Samantha Sawa-Ballweber, Kevin Dines, Gregory J. Opiteck

Receptos, a wholly owned subsidiary of Celgene Corporation, San Diego, California, United States.

INTRODUCTION

- Ozanimod (RPC1063) selectively targets sphingosine 1-phosphate ($S1P_{R1}$) and $S1P_{R5}$ and has shown therapeutic benefit in clinical trials of relapsing multiple sclerosis (RMS) and ulcerative colitis[1,2]
- Ozanimod is an oral, once-daily immunomodulator that selectively targets $S1P_{R1}$ and $S1P_{R5}$ to mitigate autoimmune responses, while potentially maintaining a level of immune surveillance and allowing rapid recovery of lymphocytes following treatment cessation[3,4]
- In addition, ozanimod has been shown preclinically to penetrate the central nervous system (CNS), where its effects on $S1P_{R1}$ and $S1P_{R5}$ expressed on specific cells may confer neuroprotective benefits[4–9]
- Neurofilament light chain (NfL) is released into the cerebrospinal fluid and serum/plasma following neuronal injury and degeneration[10]
- It may serve as a biomarker for monitoring neurological damage in RMS[11,12]

Figure 1. Ozanimod Decreases Plasma NfL in the EAE Mouse Model of MS

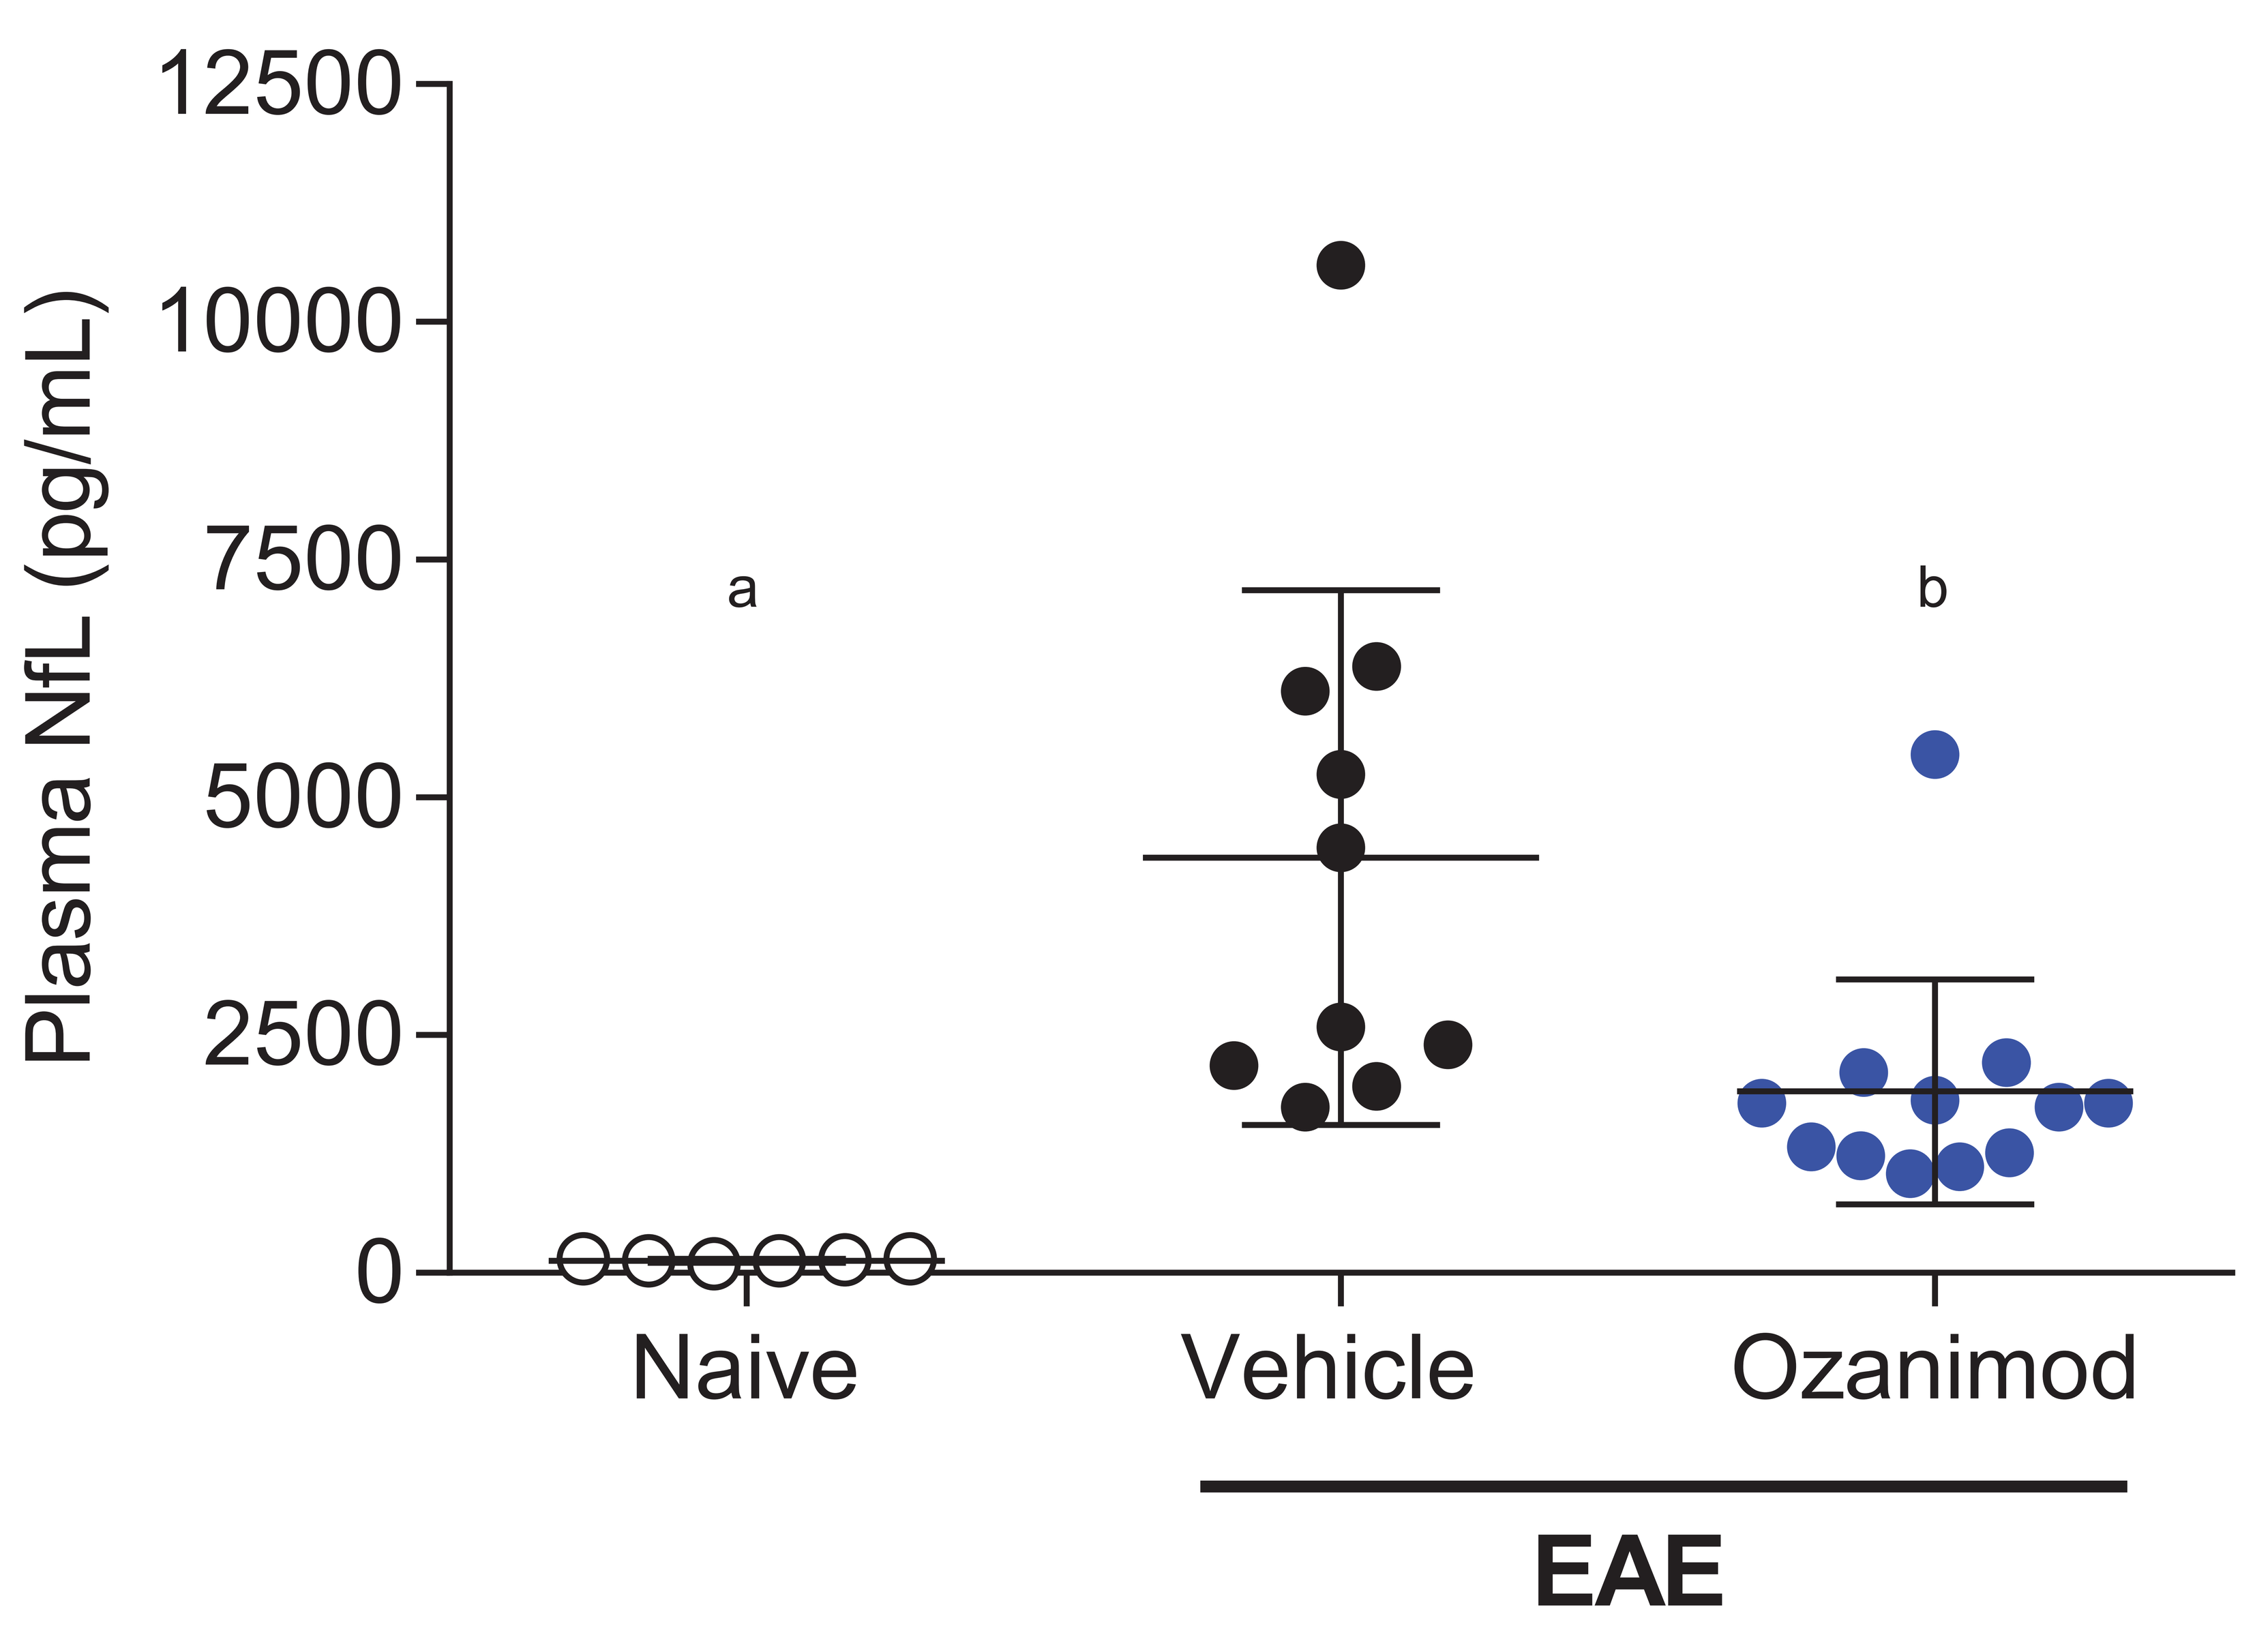
Plasma NfL (pg/mL)
12500
10000
7500
5000
2500
0
a
b
Naive
Vehicle
Ozanimod
EAE

EAE, experimental autoimmune encephalomyelitis; MS, multiple sclerosis; NfL, neurofilament light chain.

$^{a}P<0.001$, $^{b}P<0.01$ versus vehicle-treated animals.

RESULTS

- Plasma NfL was significantly elevated in EAE (Figure 1) and cuprizone-treated mice (Figure 2) over naive controls
- Elevated plasma NfL levels correlated with spinal cord inflammation and demyelination in EAE and with neuronal breaks in the corpus callosum of mice treated with cuprizone
- Ozanimod treatment in EAE significantly reduced plasma NfL levels correlating with a significant reduction in clinical scores and spinal cord inflammation and demyelination
- Mice treated with RP-101074 in the cuprizone model had significantly reduced plasma NfL and neuronal breaks, supportive of a neuroprotective effect

DISCLOSURES

All authors: Shareholder, Celgene Corporation.

ACKNOWLEDGMENTS

Support for third-party editorial assistance for this poster was provided by Jamie Weaver, PhD, of CodonMedical, an Ashfield Company, part of UDG Healthcare plc, and was funded by Celgene Corporation.

DISCUSSION

- Plasma NfL was significantly elevated in EAE mice and in the cuprizone-treated mice compared with naive, non-diseased animals
- Elevated plasma NfL correlated with clinical scores and spinal cord inflammation and demyelination in EAE, as well as with neuronal breaks in the cuprizone model
- In both of these preclinical studies, plasma NfL served as a biomarker indicative of CNS injury, inflammation, and demyelination
- These observed effects indicate a potential positive and direct CNS effect of ozanimod

METHODS

- The concentration of NfL was measured in the plasma of experimental autoimmune encephalomyelitis (EAE) and cuprizone-induced demyelination mouse models
- Blood was isolated from mice via terminal cardiac puncture, and plasma NfL was measured at Quanterix (Lexington, MA) using the Simoa technology platform
- In EAE, mice were therapeutically dosed with ozanimod for 14 days, and then spinal cords were assessed at termination for inflammation by hematoxylin and eosin staining and demyelination by Luxol Fast Blue at termination
- In the cuprizone model, mice were treated with an ozanimod surrogate (RP-101074) for 6 weeks concurrently with cuprizone, and corpus callosum sections at termination were stained with the anti-neurofilament H mouse monoclonal antibody (SMI-32) to evaluate neuronal breaks

Figure 2. The Ozanimod Surrogate RP-101074 Reduces Plasma NfL in the Cuprizone-induced Demyelination Mouse Model

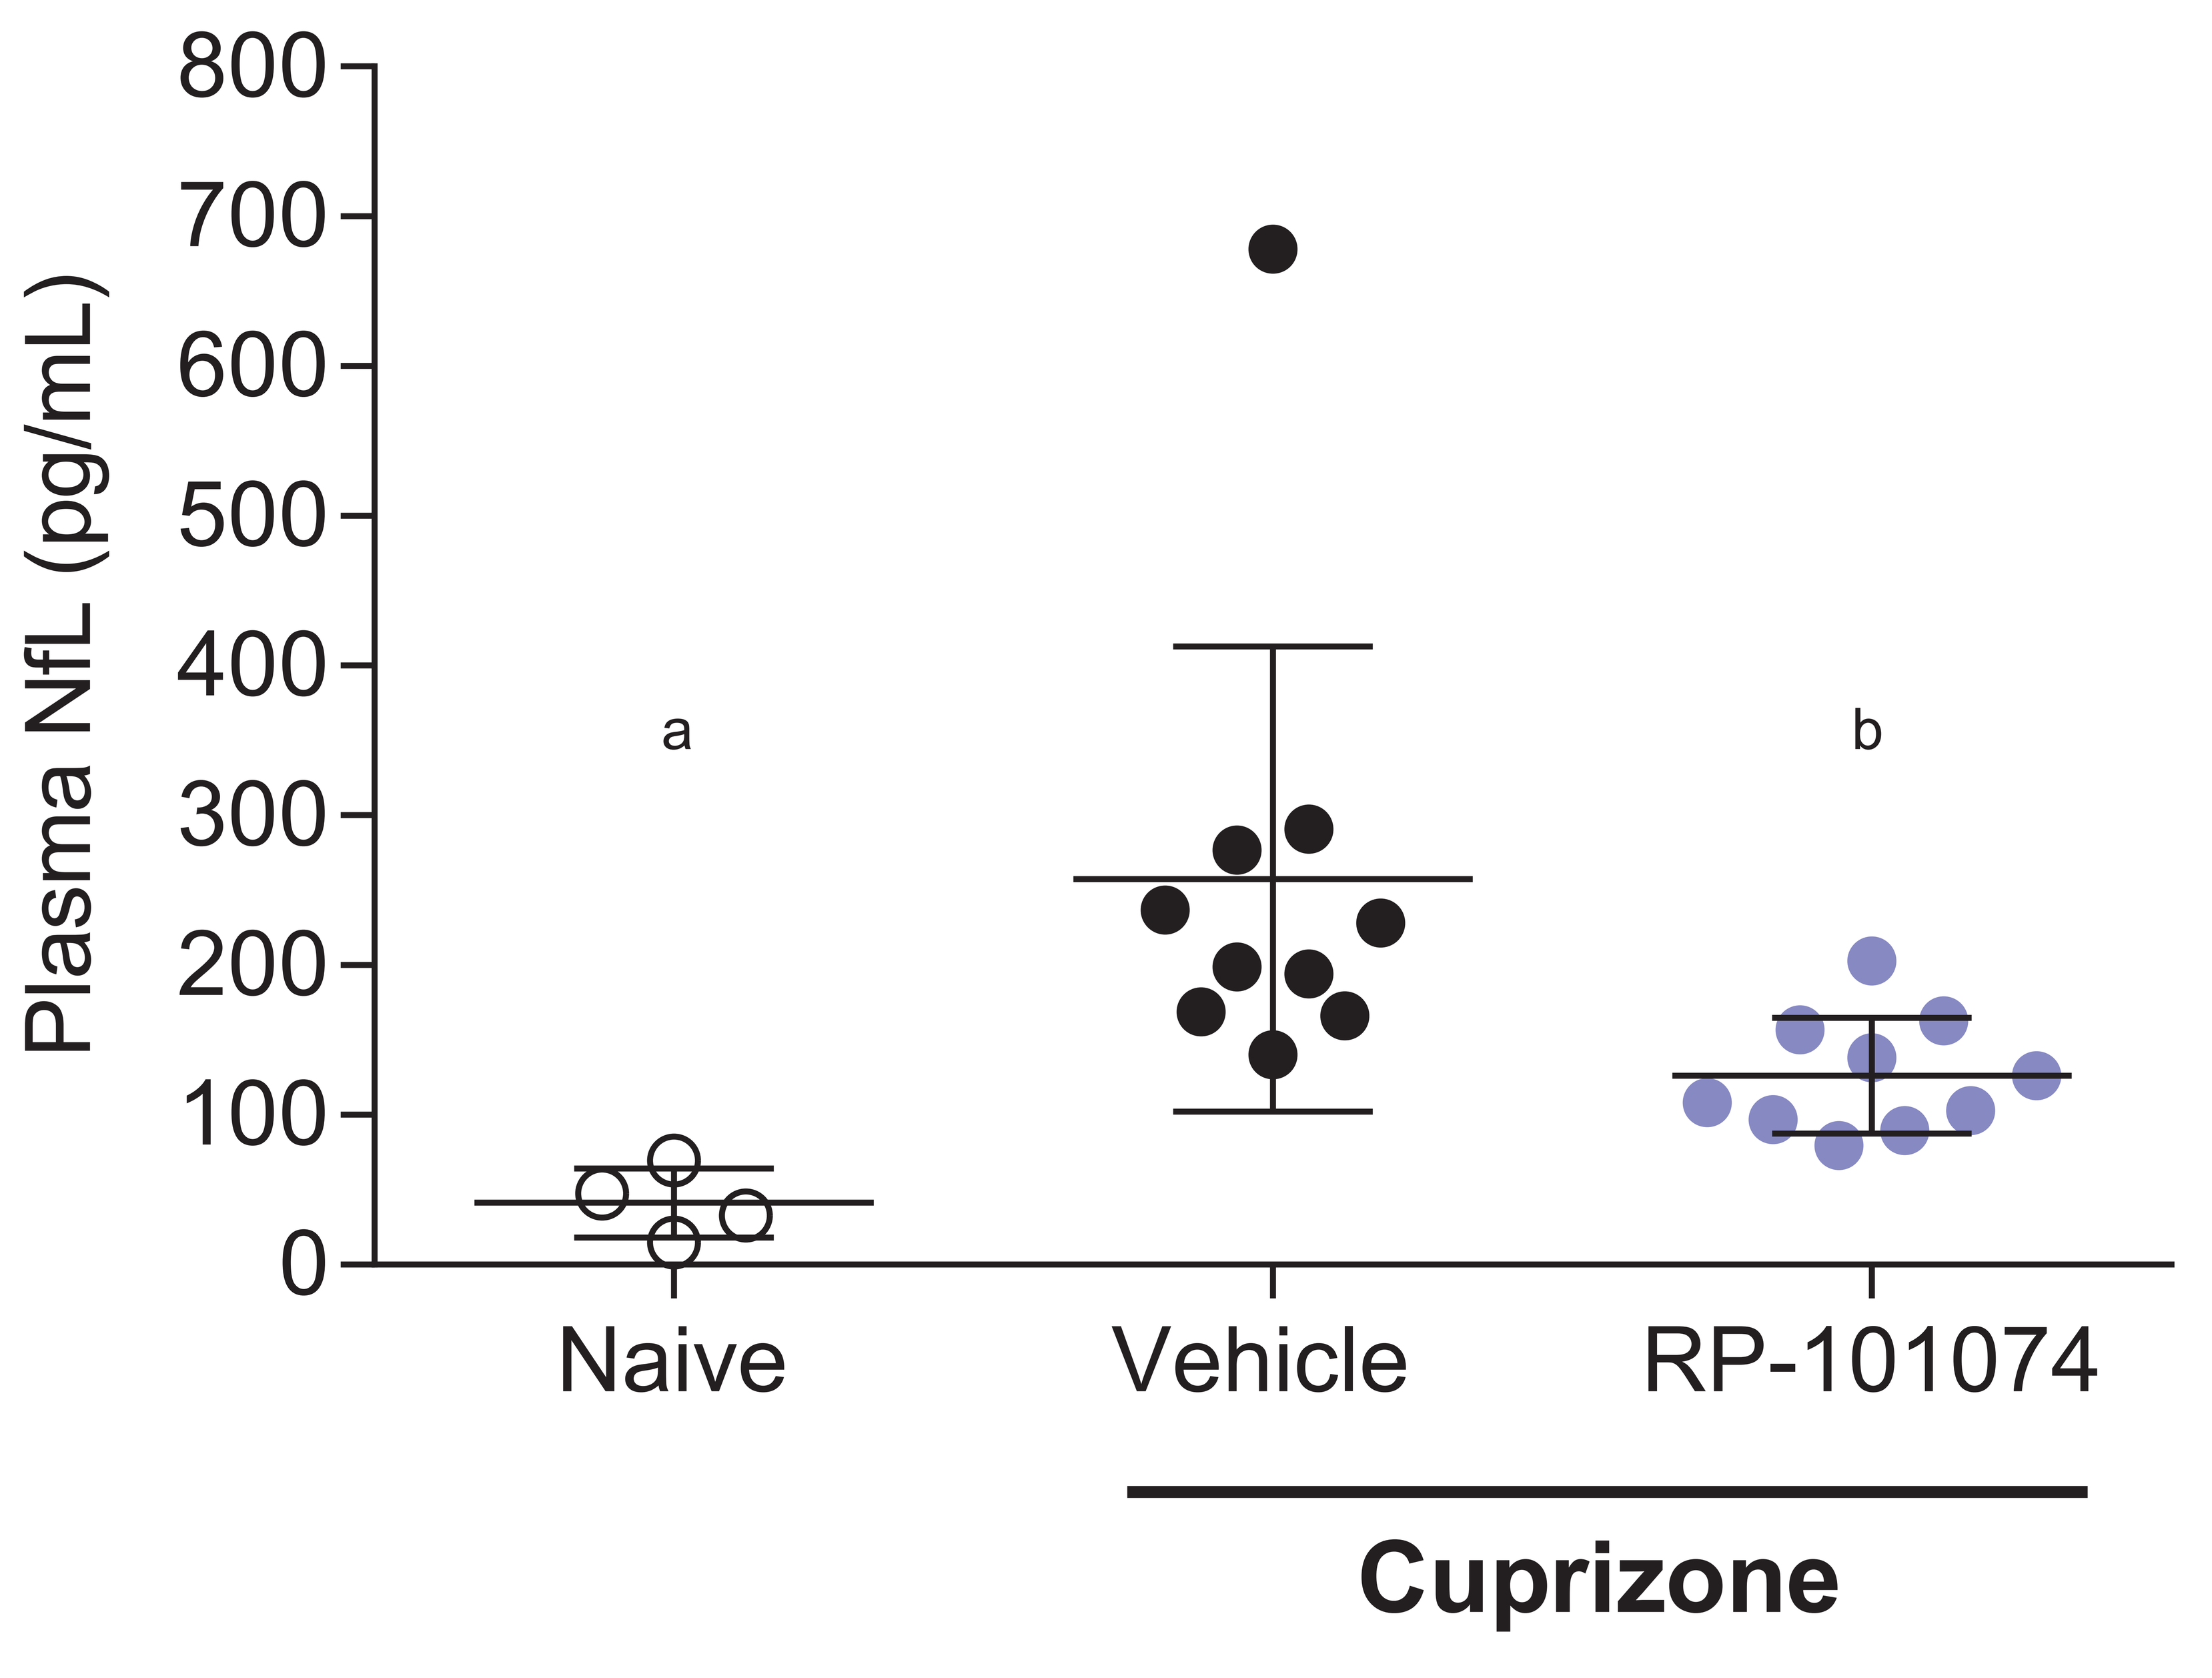
Plasma NfL (pg/mL)
800
700
600
500
400
300
200
100
0
a
b
Naive
Vehicle
RP-101074
Cuprizone

NfL, neurofilament light chain.

$^{a}P<0.01$, $^{b}P<0.05$, versus vehicle-treated animals.

LITERATURE

1. Cohen JA, et al. *Lancet Neurol*. 2016;15:373–81.
2. Sandborn WJ, et al. *N Engl J Med*. 2016;374:1754–62.
3. Tran JQ, et al. *J Clin Pharmacol*. 2017. [Epub ahead of print]
4. Scott FL, et al. *Br J Pharmacol*. 2016;173:1178–92.
5. Osinde M, et al. *Neuropharmacology*. 2007;52:1210–8.
6. Mullershausen F, et al. *J Neurochem*. 2007;102:1151–61.
7. Jaillard C, et al. *J Neurosci*. 2005;25:1459–69.
8. Miron VE, et al. *Ann Neurol*. 2008;63:61–71.
9. Miron VE, et al. *Am J Pathol*. 2008;173:1143–52.
10. Lu C-H, et al. *Neurology*. 2015; 84:2247–57.
11. Kuhle J, et al. *Neurology*. 2015; 84:1639–43.
12. Piehl F, et al. *Mult Scler*. 2017. [Epub ahead of print]

Copies of this poster obtained through the QR code are for personal use only and may not be reproduced without permission from the author of this poster.

Kristen Taylor Meadows
krtaylor@celgene.com

Celgene

Presented at the European Charcot Foundation 25th Annual Meeting; November 30–December 2, 2017; Baveno, Italy